3072

Preliminary Interim Results of the First-in-Human, Dose-Finding PROCLAIM-CX-072 Trial of the PD-L1 Probody Therapeutic CX-072 in Combination With Ipilimumab in Patients With Advanced Solid Tumors

Rachel E. Sanborn,[1] C. Willemien Menke-van der Houven van Oordt,[2] Karen A. Autio,[3] Hendrik-Tobias Arkenau,[4] Javier Garcia-Corbacho,[5] Patricia LoRusso,[6] Ruth Plummer,[7] Nataliya Uboha,[8] Omid Hamid,[9] Jerzy Wydmanski,[10] Vanessa Huels,[11] Beiyao Zheng,[11] Matthias Will,[11] Rachel Humphrey,[11] Fiona Thistlethwaite,[12] Elisabeth de Vries,[13] Anthony El-Khoueiry[14]

[1]Earle A. Chiles Research Institute, Providence Cancer Institute, Portland, OR; [2]VUMC Cancer Centre Amsterdam, Amsterdam, Netherlands; [3]Memorial Sloan Kettering Cancer Center, New York, NY; [4]Sarah Cannon Research Institute UK Limited, London, United Kingdom; [5]Hospital Clinic Barcelona, Barcelona, Spain; [6]Yale University School of Medicine–Yale Cancer Center, New Haven, CT; [7]Northern Centre for Cancer Care, Newcastle, United Kingdom; [8]University of Wisconsin–Carbone Cancer Center, Madison, WI; [9]The Angeles Clinic and Research Institute, Los Angeles, CA; [10]NZOZ Vegamed, Katowice, Poland; [11]CytomX Therapeutics, Inc., South San Francisco, CA; [12]The Christie NHS Foundation Trust and University of Manchester, Manchester, United Kingdom; [13]Universitair Medisch Centrum Groningen, Groningen, Netherlands; [14]USC Norris Comprehensive Cancer Center, Los Angeles, CA

BACKGROUND

- Programmed cell death ligand 1 (PD-L1) is expressed on many cancer and immune cells; by binding the receptor programmed cell death 1 (PD-1), a negative regulator of T-lymphocyte activation, it can block cancer immune detection[1,2]
- Antibodies targeting the PD-1 pathway have demonstrated deep and durable cancer treatment responses when administered as monotherapy and even greater efficacy in combination regimens[3]
- Immune-related adverse events (AEs), including interstitial pneumonitis, colitis, and transaminitis, are known toxicities of PD-1/PD-L1 axis-blocking antibodies. These toxicities are considerably higher when dual checkpoint inhibition is implemented[4-7]
- Probody™ therapeutics are fully recombinant antibody prodrugs that are designed to remain relatively inactive, both systemically and in healthy tissue, thereby avoiding binding to target antigen in healthy tissue. Probody therapeutics are designed to be activated specifically in the tumor microenvironment by tumor resident proteases[8,9]
- CX-072 is a Probody therapeutic directed against PD-L1 for the treatment of patients with cancer
- Coinhibition of the 2 immune checkpoints, PD-1 and CTLA-4, has led to increased efficacy at the expense of increased immune-related toxicities; up to 55% of patients receiving combination therapy experience grade 3 or 4 treatment-related AEs (TRAEs)[10,11]
- The PROCLAIM-CX-072 (**PRO**body **CL**inical **A**ssessment **I**n **M**an) study is evaluating tolerability and preliminary antitumor activity of multiple doses and expansion of CX-072 as monotherapy (Part A [Poster 3071], A2, and D) or as combination therapy with ipilimumab (Part B1 and B2) or vemurafenib (Part C) in patients with advanced, unresectable solid tumors or lymphoma (ClinicalTrials.gov identifier, NCT03013491)

OBJECTIVES AND END POINTS

- The primary objectives of Part B1 of the study are to assess the safety and tolerability and to determine the maximum tolerated dose (MTD) and the dose-limiting toxicity (DLT) of CX-072 when administered in a concomitant combination schedule with ipilimumab
- Secondary objectives are to obtain preliminary evidence of anticancer activity in patients treated with CX-072 combined with ipilimumab using
 - Response rate (Response Evaluation Criteria in Solid Tumors [RECIST] v 1.1)
 - Time to response and duration of response
 - Progression-free survival

METHODS

Study Design

- This is a first-in-human, open-label, dose-escalation, multicenter, phase 1/2a study of CX-072 designed to evaluate the safety and to determine the MTD and/or maximum achieved dose of CX-072 in combination therapy with ipilimumab (Part B1)
- Patients are ≥18 years of age with Eastern Cooperative Oncology Group performance status 0-1
- To be included in Part B1 (n ≤ 42 patients), the following are required
 - To have any metastatic or advanced unresectable solid tumor or lymphoma (excluding thymic epithelial tumor, thymoma, and thymic carcinoma) (measurable and nonmeasurable disease)
 - To be naive to immunotherapy, including to PD-1/PD-L1 and CTLA-4 inhibitor therapy, and to have a tumor type not approved for immune checkpoint inhibitors
- CX-072 (0.3, 1.0, 3.0, and 10 mg/kg) in combination with ipilimumab (3.0 mg/kg or 10 mg/kg for the highest CX-072 dose level) is administered intravenously every 21 days for 4 cycles, followed by CX-072 monotherapy every 14 days
- Initiation of cohort enrollment required successful completion of the CX-072 monotherapy dose level evaluated in Part A (Poster 3071)

Assessments

- Imaging for tumor response assessment is performed every 8 weeks for the first 12 months, then every 12 weeks thereafter
- After the last dose of study medication, patients will be evaluated every 3 months for disease progression and overall survival until study withdrawal or death
- Archival tissue or fresh biopsy samples are provided at baseline
- Serial blood samples for pharmacokinetic (PK) analysis are collected to characterize the PK profile of CX-072 in combination with ipilimumab
- Participating patients provide serial blood samples for measurement of exploratory biomarkers of immune modulation
- Data snapshot for the analysis was taken on April 20, 2018

INTERIM RESULTS

Patients

- Patients included in the study to date (N = 16) received the following doses of CX-072 (mg/kg) + ipilimumab 3.0 mg/kg
 - 0.3, n = 6; 1.0, n = 3; 3.0, n = 3; 10.0, n = 4
- Baseline characteristics are presented in **Table 1**
- At the time of analysis, 4 patients (25%) were still receiving treatment
 - 12 patients discontinued treatment because of disease progression (n = 8), symptomatic deterioration (n = 3), or death (n = 1)

Table 1. Baseline Characteristics

	All Patients N = 16
Median age (range), years	60 (28-70)
Sex, n (%)	
Male	8 (50.0)
Female	8 (50.0)
Race, n (%)	
White	13 (81.3)
Asian	1 (6.3)
Not reported/unknown	2 (12.5)
ECOG performance status, n (%)	
0	6 (37.5)
1	10 (62.5)
No. of previous cancer treatments, median (range)	3 (1-12)
Cancer types, n (%)	
Pancreatic carcinoma	2 (12.5)
Other[a]	14 (87.5)

ECOG, Eastern Cooperative Oncology Group.
[a]One patient each had anal squamous cell carcinoma, breast (ER+) carcinoma, cervix carcinoma, colon carcinoma, gastric cancer, glioblastoma, osteosarcoma, salivary gland carcinoma, cancer of unknown primary, small cell lung cancer, small cell neuroendocrine prostate cancer, testicular carcinoma, triple-negative breast cancer, and head and neck squamous cell carcinoma.

Duration of Treatment

- Mean (range) durations of treatment are reported in **Table 2**

Table 2. Duration of CX-072 + Ipilimumab Treatment

CX-072 (mg/kg) + Ipilimumab 3.0 mg/kg Dose	0.3 n = 6	1.0 n = 3	3.0 n = 3	10.0 n = 4	All Patients N = 16
Time on treatment, mean (range), months	3.0 (1-10)	4.6 (3-6)	3.4 (1-4)	1.8 (1-3)	3.1 (1-10)

Safety

- All combination cohorts through CX-072 10.0 mg/kg (dose selected for monotherapy cohort expansion) with ipilimumab 3.0 mg/kg are now enrolled; the MTD has not been reached
- Treatment-emergent AEs (TEAEs) are summarized in **Table 3**
 - One patient with head and neck squamous cell carcinoma who received CX-072 0.3 mg/kg + ipilimumab 3.0 mg/kg experienced a DLT (grade 3 dyspnea)
- 5 (31.3%) patients* experienced grade 3 TRAEs
 - CX-072 0.3 mg/kg + ipilimumab 3.0 mg/kg: 2 patients (colitis and dyspnea/pneumonitis)
 - CX-072 1.0 mg/kg + ipilimumab 3.0 mg/kg: 1 patient with both headache and hyponatremia
 - CX-072 10.0 mg/kg + ipilimumab 3.0 mg/kg: 1 patient with both amylase and lipase increase (grade 4)
- There were 4 deaths (all from disease progression) within 30 days of last study drug administration
- The most common TEAEs occurring in ≥3 patients are reported in **Table 4**

*A grade 3 TRAE in 1 patient was designated as nontreatment related postdata snapshot.

Table 3. Safety Summary, Patients Experiencing Event, n (%)

CX-072 (mg/kg) + Ipilimumab 3.0 mg/kg Dose	0.3 + 3.0 n = 6	1.0 + 3.0 n = 3	3.0 + 3.0 n = 3	10.0 + 3.0 n = 4	All Patients N = 16
Any TEAE	6 (100.0)	3 (100.0)	3 (100.0)	4 (100.0)	16 (100.0)
Grade ≥3	4 (66.7)	3 (100.0)	0	3 (75.0)	10 (62.5)
SAE	3 (50.0)	3 (100.0)	0	2 (50.0)	8 (50.0)
TEAE related to any study drug					
Grade ≥3	2 (33.3)	2 (66.7)	0	1 (25.0)	5 (31.3)
SAE	2 (33.3)	3 (100.0)	0	0	5 (31.3)

SAE, serious adverse event; TEAE, treatment-emergent adverse event.
Adverse events were coded using MedDRA version 19.1.

Table 4. Most Common TEAEs Occurring in ≥3 Patients Experiencing Event, n (%)

CX-072 (mg/kg) + Ipilimumab 3.0 mg/kg Dose	0.3 n = 6		1.0 n = 3		3.0 n = 3		10.0 n = 4		All Patients N = 16	
Grade	1-2	3-4	1-2	3-4	1-2	3-4	1-2	3-4	1-2	3-4
Nausea	4 (66.7)	0	3 (100.0)	0	0	0	0	0	7 (43.8)	0
Fatigue	3 (50.0)	0	2 (66.7)	0	0	0	1 (25.0)	0	6 (37.5)	0
Decreased appetite	2 (33.3)	0	1 (33.3)	0	1 (33.3)	0	1 (25.0)	0	5 (31.3)	0
Pruritus	2 (33.3)	0	0	0	1 (33.3)	0	2 (50.0)	0	5 (31.3)	0
Rash	1 (16.7)	0	1 (33.3)	0	2 (66.7)	0	0	0	4 (25.0)	0
Abdominal pain	1 (16.7)	1 (16.7)	0	0	0	0	1 (25.0)	0	2 (12.5)	1 (6.3)
Asthenia	1 (16.7)	0	0	0	2 (66.7)	0	0	0	3 (18.8)	0
Cough	2 (33.3)	0	0	0	0	0	1 (25.0)	0	3 (18.8)	0
Pyrexia	1 (16.7)	0	1 (33.3)	0	0	0	1 (25.0)	0	3(18.8)	0
Vomiting	2 (33.3)	0	1 (33.3)	0	0	0	0	0	3 (18.8)	0

TEAE, treatment-emergent adverse event.
Patients were grouped according to the most severe grade experienced for a particular adverse event. Adverse events were coded using MedDRA version 19.1.

Tumor Response

- Among evaluable patients (n = 12), best tumor response (**Table 5**) was
 - Complete response (n = 1): anal cell squamous cell carcinoma, CX-072 0.3 mg/kg + ipilimumab 3.0 mg/kg; PD-L1 negative, microsatellite-stable, intermediate tumor mutational burden, HPV-positive
 - Partial response (n = 2): testicular cancer and unknown primary (likely small bowel)
- Target lesions decreased from baseline in 3 of 10 (30%) patients with measurable disease at baseline (**Figure 1A**)
- Percentage change in tumor burden over time is presented in **Figure 1B**

Table 5. Best Tumor Response in Evaluable Patients[a] per RECIST v1.1, n (%)

CX-072 (mg/kg) + Ipilimumab (3.0 mg/kg) Dose, %	0.3 n = 5	1.0 n = 3	3.0 n = 2	10.0 n = 2	All Evaluable Patients N = 12
Objective response rate[b]	1 (20.0)	1 (33.3)	1 (50.0)	0	3 (25.0)
Complete response	1 (20.0)	0	0	0	1 (8.3)
Partial response	0	1 (33.3)	1 (50.0)	0	2 (16.7)
Stable disease	0	1 (33.3)	0	0	1 (8.3)
Progressive disease	4 (80.0)	1 (33.3)	1 (50.0)	2 (100.0)	8 (66.7)

RECIST, Response Evaluation Criteria in Solid Tumors.
[a]Evaluable patients are those with an adequate disease assessment at baseline and ≥1 postbaseline tumor assessment.
[b]Includes patients with unconfirmed response.

Figure 1. (A) Best percentage change from baseline in target lesions and (B) spider plots.[a]

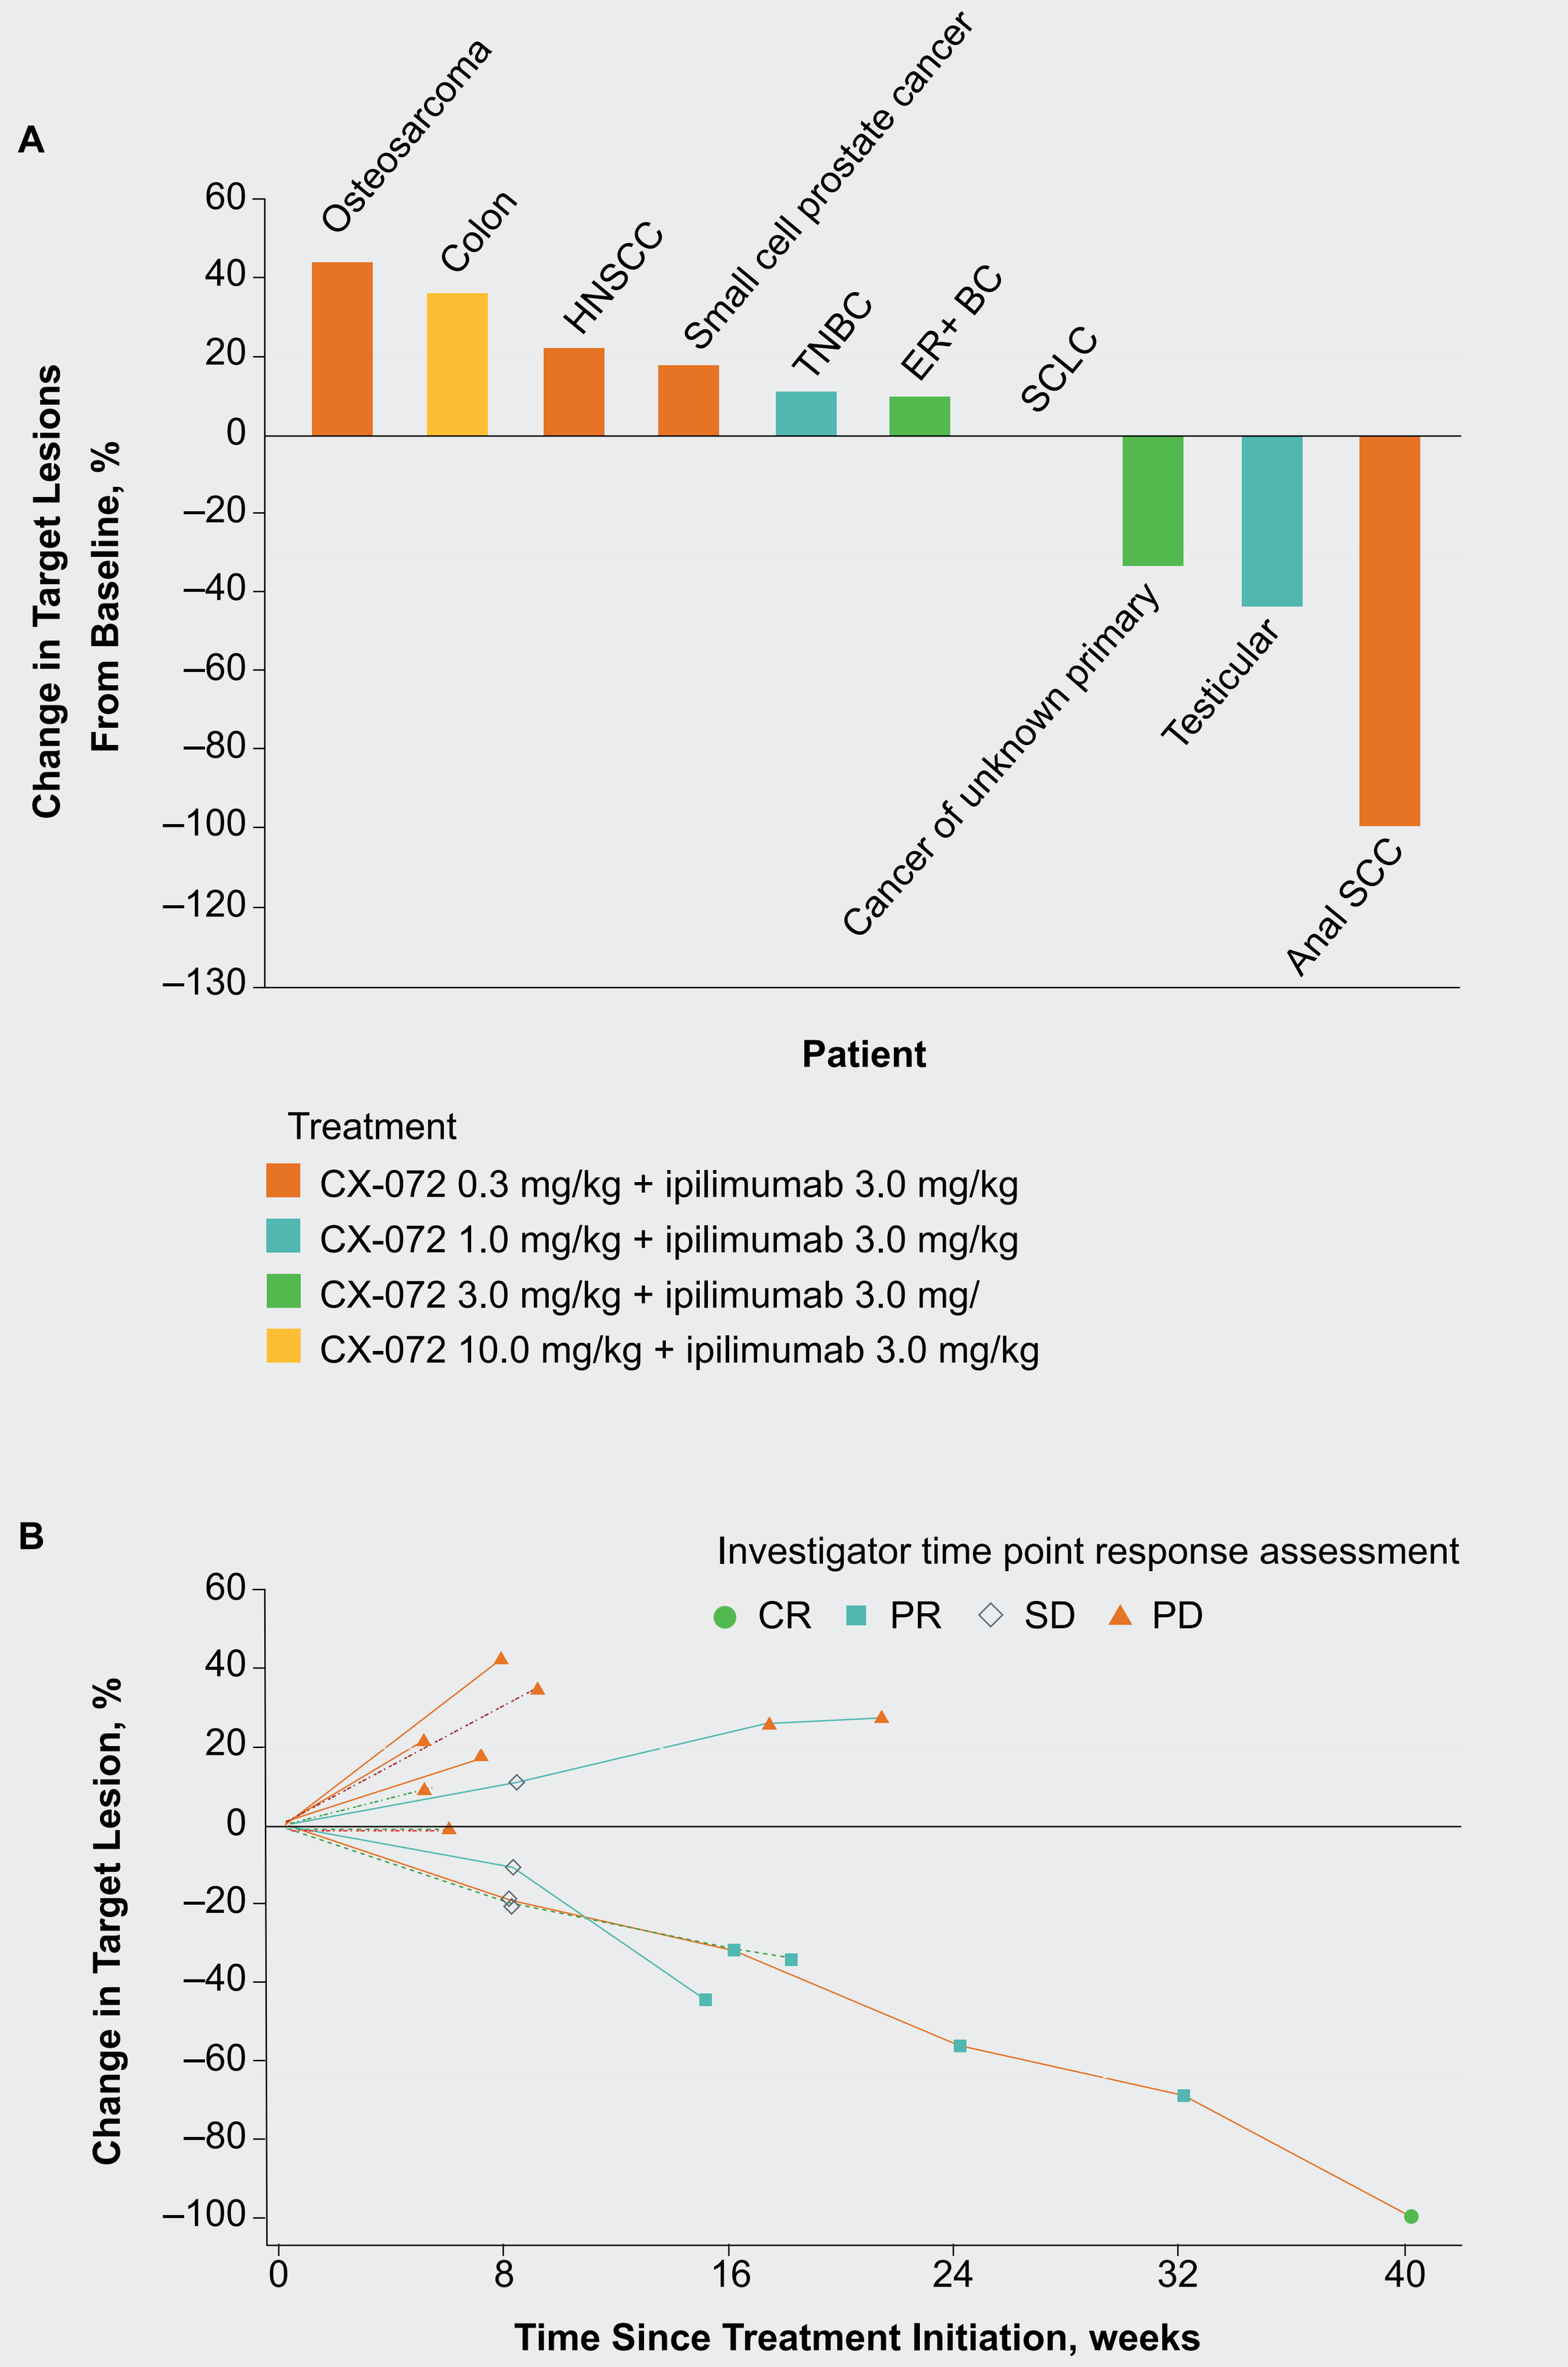
A
Change in Target Lesions From Baseline, %
60
40
20
0
−20
−40
−60
−80
−100
−120
−130
Osteosarcoma
Colon
HNSCC
Small cell prostate cancer
TNBC
ER+ BC
SCLC
Cancer of unknown primary
Testicular
Anal SCC
Patient
Treatment
CX-072 0.3 mg/kg + ipilimumab 3.0 mg/kg
CX-072 1.0 mg/kg + ipilimumab 3.0 mg/kg
CX-072 3.0 mg/kg + ipilimumab 3.0 mg/
CX-072 10.0 mg/kg + ipilimumab 3.0 mg/kg
B
Investigator time point response assessment
CR
PR
SD
PD
Change in Target Lesion, %
60
40
20
0
−20
−40
−60
−80
−100
0
8
16
24
32
40
Time Since Treatment Initiation, weeks

CR, complete response; ER+ BC, estrogen receptor–positive breast cancer; HNSCC, head and neck squamous cell carcinoma; PD, progressive disease; PR, partial response; RECIST, Response Evaluation Criteria in Solid Tumors; SCC, squamous cell carcinoma; SCLC, small cell lung cancer; SD, stable disease; TNBC, triple-negative breast cancer.
[a]As evaluated per RECIST v1.1. Plots include data from evaluable patients with measurable disease at baseline.

Sample Case Studies

- Patient A has anal squamous cell carcinoma with intermediate tumor mutational burden (9 mutations/ megabase) (microsatellite-stable, HPV-positive, and PD-L1 status unknown). Patient was treated with CX-072 0.3 mg/kg + ipilimumab 3 mg/kg and had confirmed complete response postdata snapshot at follow-up staging (**Figure 2A**)
- Patient B has cancer of unknown primary and negative PD-L1 status. Patient was treated with CX-072 3 mg/kg + ipilimumab 3 mg/kg and had an unconfirmed partial response at follow-up staging (**Figure 2B**)

Figure 2. (A) Patient A: anal squamous cell carcinoma (CX-072 3 mg/kg). (B) Patient B: cancer of unknown primary (CX-072 10 mg/kg).

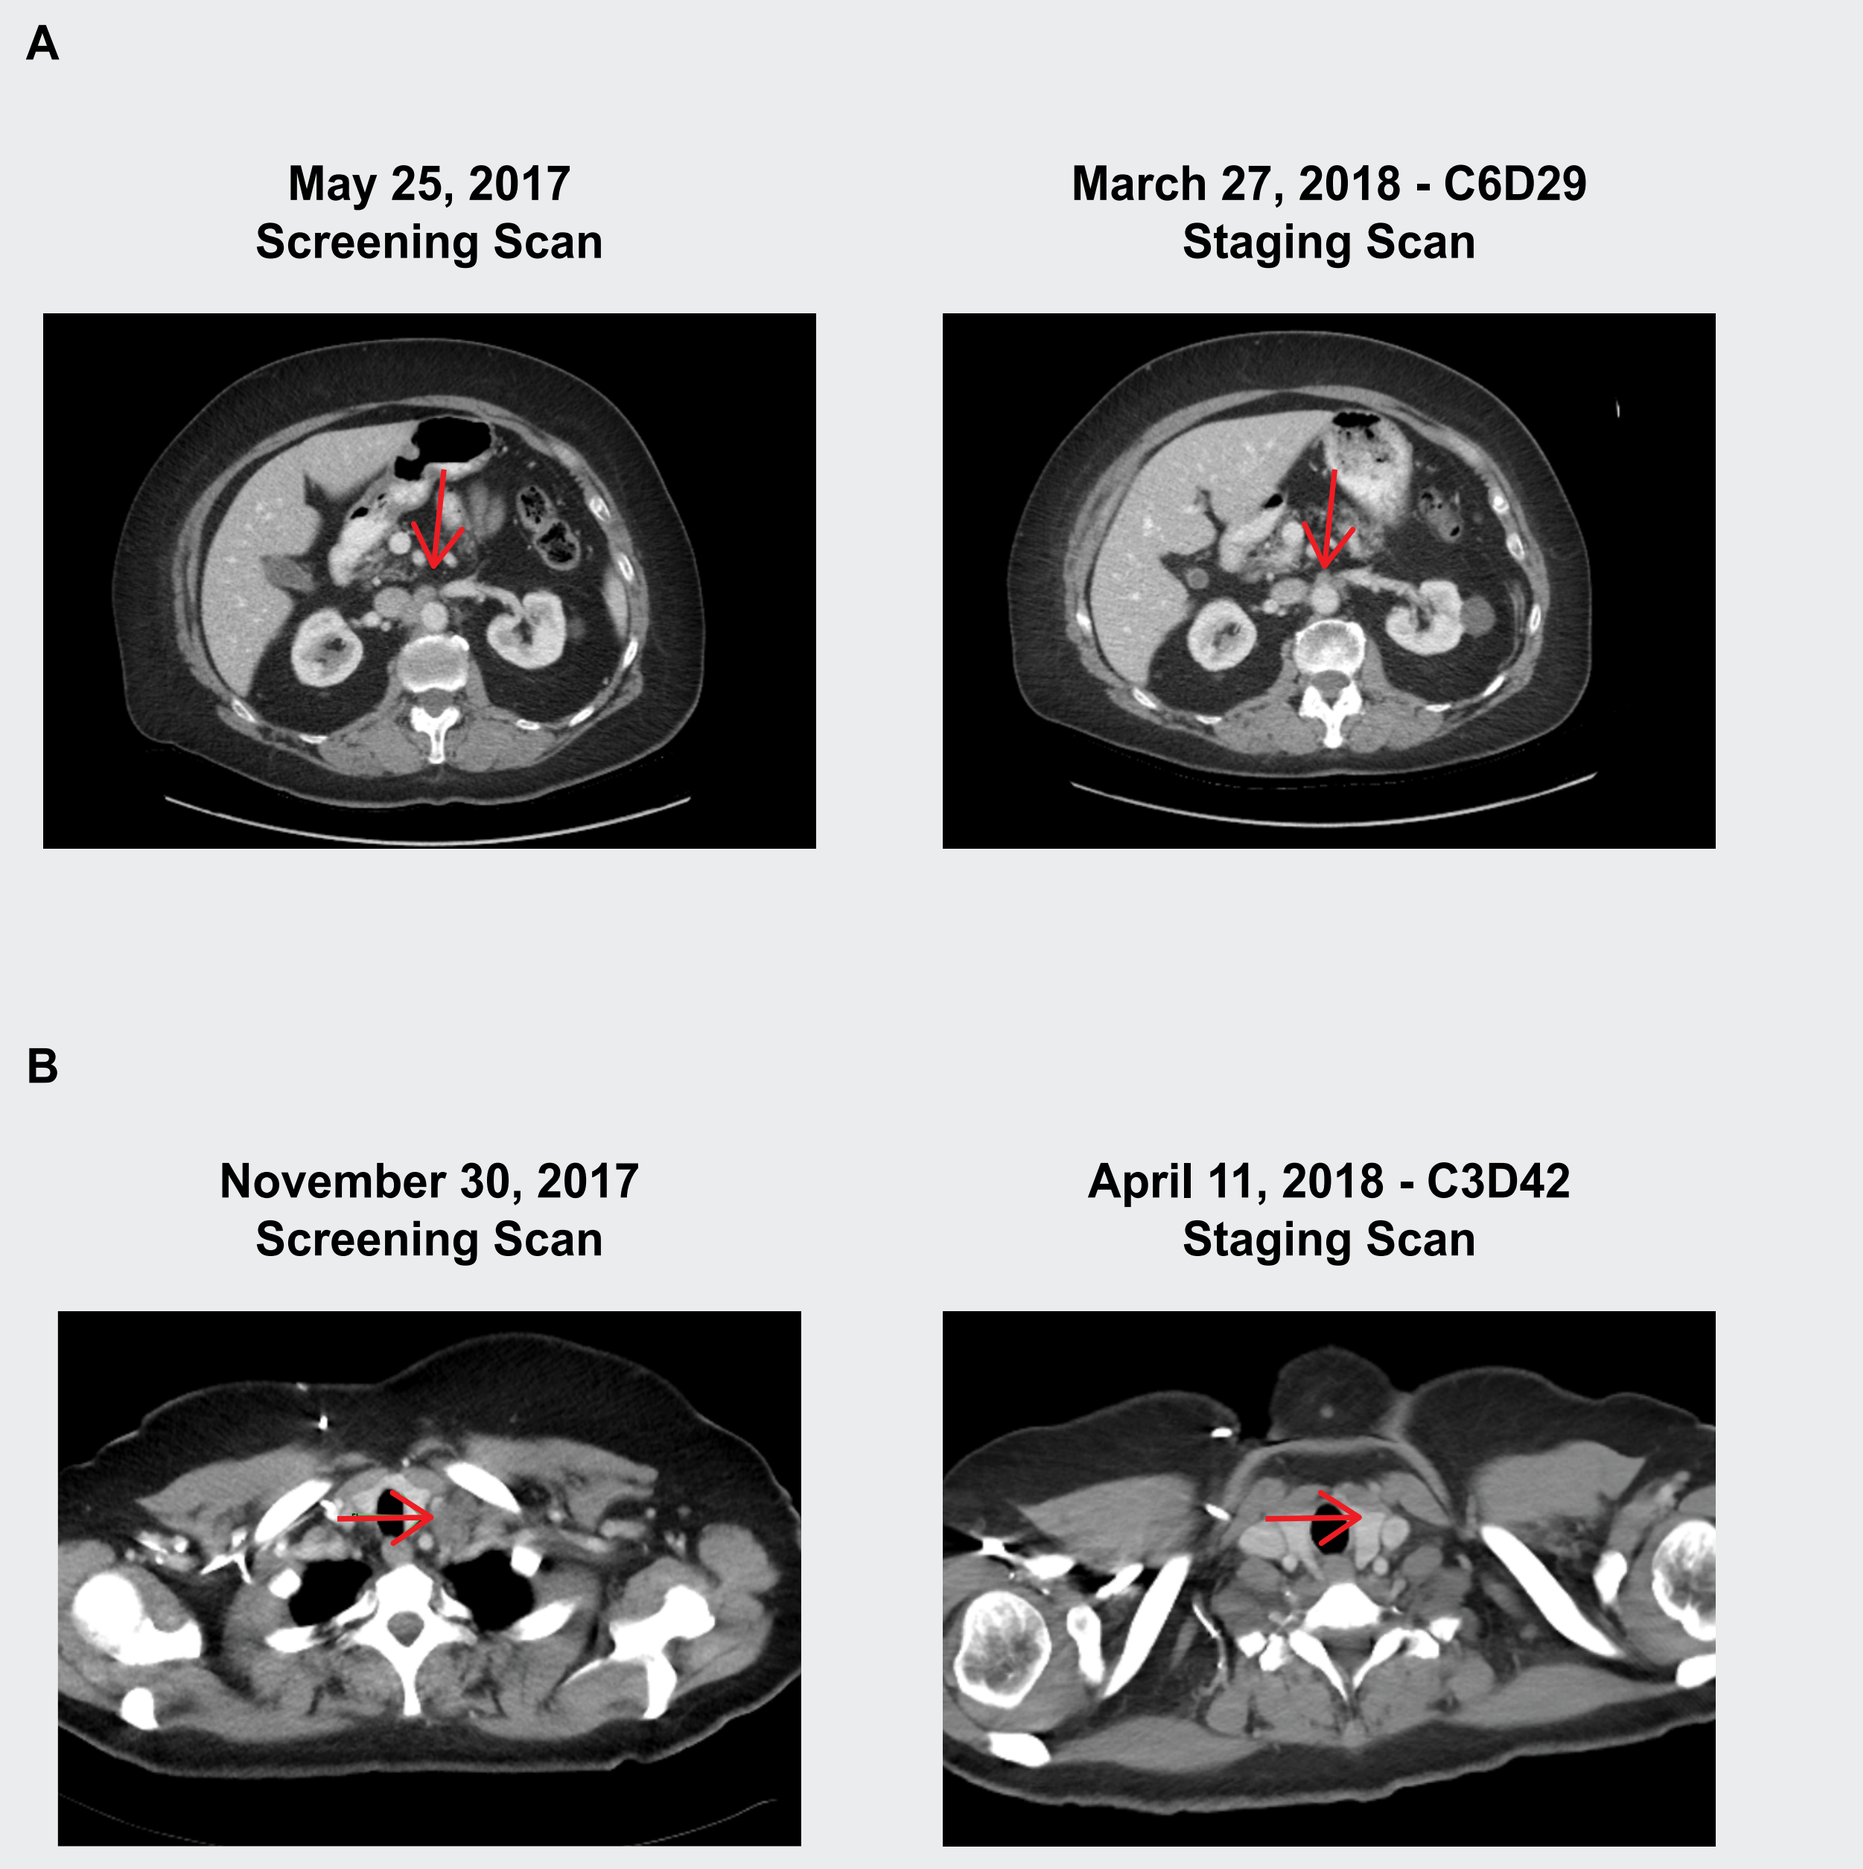
A
May 25, 2017
Screening Scan
March 27, 2018 - C6D29
Staging Scan
B
November 30, 2017
Screening Scan
April 11, 2018 - C3D42
Staging Scan

INTERIM CONCLUSIONS

- Early safety observations in this dose-escalation study of the combination of the anti-PD-L1 Probody CX-072 and ipilimumab 3 mg/kg indicate a TRAE rate trending below the level reported for other PD-1 pathway inhibitors in combination with ipilimumab
- No new safety signals were observed with the combination of the anti–PD-L1 Probody CX-072 and ipilimumab 3 mg/kg
- Preliminary efficacy results showed 1 complete response and 2 partial responses (3/12; 25%) and warrant further investigation
- Cohort expansion of the combination of CX-072 + ipilimumab and further dose-escalation steps with ipilimumab 10 mg/kg are pending

REFERENCES

1. Herbst RS et al. *Nature.* 2014;515:563-567.
2. Yu LY et al. *Int J Environ Res Public Health.* 2017;14.
3. Iwai Y et al. *J Biomed Sci.* 2017;24:26.
4. Wolchok JD et al. *N Engl J Med.* 2013;369:122-133.
5. Larkin J et al. *N Engl J Med.* 2015;373:23-34.
6. Ahn MJ et al. *Expert Opin Drug Saf.* 2017;16:465-469.
7. Hwu P et al. *Ann Oncol.* 2016;27:379-400.
8. Desnoyers LR et al. *Sci Transl Med.* 2013;5:207ra144.
9. Polu KR et al. *Expert Opin Biol Ther.* 2014;14:1049-1053.
10. Larkin J et al. *N Engl J Med.* 2015;373:23-34.
11. Wolchok JD et al. *N Engl J Med.* 2017;377:1345-1356.

ACKNOWLEDGMENTS

The authors thank the participating patients and their families and all staff at participating sites. This study is sponsored by CytomX Therapeutics, Inc. Medical writing assistance was provided by ApotheCom (San Francisco, CA, USA) and was funded by CytomX Therapeutics, Inc.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

PROBODY is a trademark of CytomX Therapeutics, Inc. All other brands and trademarks referenced herein are the property of their respective owners.

http://bit.ly/2LN2nTY

View Other Publications

Presented at the American Society of Clinical Oncology (ASCO) Annual Meeting; June 1–5, 2018; Chicago, Illinois

© 2018 CytomX Therapeutics, Inc.