P4-01-15

Results of a phase 2 study of praluzatamab ravtansine (CX-2009) in patients with advanced breast cancer (ABC)

San Antonio Breast Cancer Symposium®
December 6-10, 2022

Kathy Miller,[1] Sara M. Tolaney,[2] Leisha Emens,[3] Sung-Bae Kim,[4] Erika Hamilton,[5] Cristina Saura,[6] Lucia Sanz,[6] Valentina Boni,[7] Filipa Lynce,[2] Juan M. Cejalvo,[8] Jennifer A. Crozier,[9] Shirley Wang,[10] Shannon Tsai,[10] Hojjat Bazzazi,[10] Hirdesh Uppal,[10] Alison Hannah,[10] Sara Hurvitz[11]

[1]Indiana University Melvin and Bren Simon Comprehensive Cancer Center, Indianapolis, IN, USA; [2]Dana-Farber Cancer Institute, Harvard Medical School, Boston, MA, USA; [3]University of Pittsburgh Medical Center, Hillman Cancer Center, Pittsburgh, PA, USA; [4]Asan Medical Center, University of Ulsan College of Medicine, Seoul, Korea; [5]Sarah Cannon Research Institute/Tennessee Oncology, Nashville, TN, USA; [6]Vall d´Hebron Institute of Oncology, Barcelona, Spain; [7]NEXT Madrid, Universitary Hospital Quironsalud Madrid, Spain; [8]Hospital Clinico Universitario of Valencia, Valencia, Spain; [9]Baptist MD Anderson Cancer Center, Jacksonville, FL, USA; [10]CytomX Therapeutics, Inc., South San Francisco, CA, USA; [11]University of California, Los Angeles/Jonsson Comprehensive Cancer Center, Los Angeles, CA, USA

BACKGROUND

- Praluzatamab ravtansine (CX-2009) is a Probody® drug conjugate consisting of a humanized anti-CD166 monoclonal antibody, a peptide masking the antigen-binding site, a protease cleavable linker, and a DM4 payload conjugated to the antibody (**Figure 1**)[1]
- Upregulated tumor protease activity, a hallmark of cancer, cleaves the substrate linker and releases the masking peptide, which allows CX-2009 to bind to CD166[2]
- CD166 (activated leukocyte cell adhesion molecule, ALCAM) is a transmembrane protein that functions as a junctional adhesion molecule and facilitates cell migration, differentiation, and hematopoiesis. It is widely expressed on dividing, normal, and malignant cells[3]
- CD166 is highly expressed in hormone receptor positive (HR+) breast cancer and variably expressed in TNBC (**Figure 2**)

Figure 1. CX-2009, a Probody drug conjugate targeting CD166

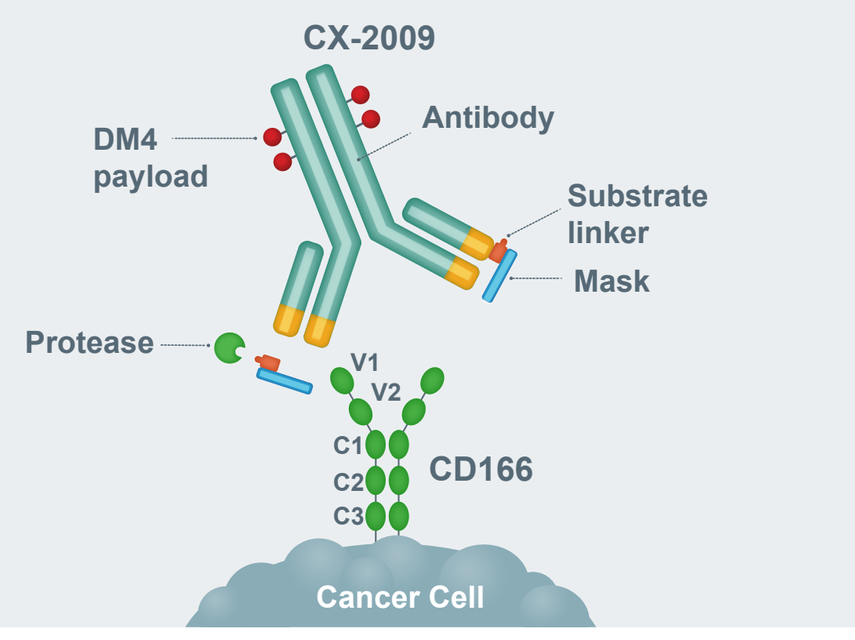
CX-2009
DM4 payload
Antibody
Substrate linker
Mask
Protease
V1
V2
C1
C2
C3
CD166
Cancer Cell

Figure 2. ALCAM (CD166) expression by receptor status in three large breast cancer datasets[4-6]

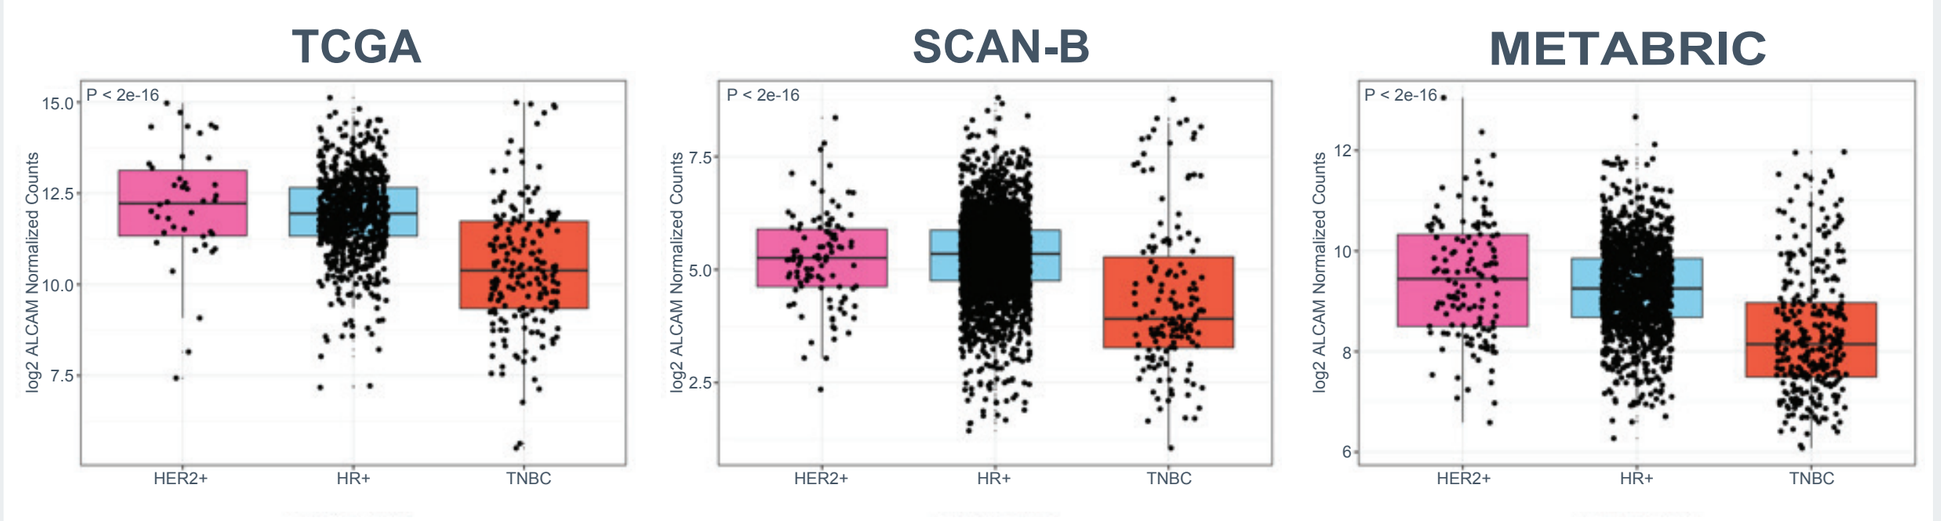
TCGA
SCAN-B
METABRIC
P < 2e-16
log2 ALCAM Normalized Counts
HER2+
HR+
TNBC

ALCAM (CD166) is highly expressed in HR+ breast cancer and variably expressed in TNBC

- In the CX-2009 phase 1 study, patients with advanced solid tumors were enrolled at doses ranging from 0.25-10 mg/kg IV Q3W and at 4 and 6 mg/kg Q2W resulting in a recommended phase 2 dose of 7 mg/kg Q3W[7]
- This phase 2 study evaluated CX-2009 as monotherapy (Arm A: patients with advanced HR+/HER2− BC; Arm B: patients with TNBC) and in combination with pacmilimab (a conditionally activated PD-L1 inhibitor) in TNBC (Arm C)
- Enrollment to the study is complete. This poster will only discuss the patients in the monotherapy arms (Arms A and B). Data is presented as of 25Aug2022

STUDY DESIGN

Phase 2, prospective, open-label, parallel-cohort, multicenter, 3 arm study of single-agent CX-2009 or in combination with CX-072 in patients with advanced breast cancer (NCT04596150)

Inclusion:
- ECOG PS 0 or 1
- Adequate hematologic, renal, and hepatic function
- Measurable disease
- TNBC – available tumor tissue (archival or fresh biopsy) for CD166 analysis

Exclusion:
- Untreated symptomatic CNS metastases
- Prior malignancy within past 2 years unless considered low risk for recurrence
- Prior maytansinoid-containing drug conjugate treatment
- Arm C: history of intolerance to prior I/O treatment and/or active autoimmune disease

Arm A
CX-2009 (7 mg/kg[a]) Q3W
HR+/HER2- (n=60)

Arm B
Selected for CD166 Expression
CX-2009 (7 or 6 mg/kg[b]) Q3W
TNBC (n=55)

Arm C[†]
Selected for CD166 + PD-L1 Expression
CX-2009 (7 mg/kg) +
pacmilimab (1200 mg) Q3W
TNBC (n=10)

Endpoints
Primary:
ORR by Central Radiology Review (CRR)
Secondary:
ORR (inv), PFS, DCR, CBR24, DoR, OS, safety, PK, ADA
Exploratory:
Biomarker correlation with outcome

[a]All patients starting dose was 7 mg/kg Q3W.
[b]19 patients had a starting dose of 7 mg/kg Q3W; the protocol was subsequently amended, and the dose was reduced to 6 mg/kg Q3W. An additional 36 patients in Arm B initiated treatment at the reduced 6 mg/kg Q3W dose.
[†]Data from Arm C is not presented in the poster.

METHODS

Statistical Methods

- **Safety-evaluable population:** all enrolled patients who received at least 1 dose of CX-2009 regardless of the duration of treatment
- **Efficacy-evaluable population:** all patients in the safety-evaluable population who have expression of CD166 by IHC and at least 1 measurable lesion on the screening radiology scans (per protocol this population would only be used to perform sensitivity analyses related to objective response)
- Due to established high CD166 expression in HR+ breast cancer no CD166 testing was performed during screening for enrollment in Arm A, but was implemented for Arm B due to known variability of CD166 expression in TNBC
- **Modified efficacy-evaluable population (MEE):** all patients in the efficacy-evaluable population who have at least 1 post-baseline tumor scan centrally assessed by the CRR
- All Arms: the primary endpoint was ORR according to RECIST v1.1 based on assessment by CRR. The analysis will include the MEE population. The analyses of ORR will be performed separately for each arm
- No futility rule was set for Arm A; a futility rule of less than 10% ORR was set for Arm B (TNBC)

Population PK Model

- A 3 compartmental population PK (POPPK) model with linear elimination from the central compartment was fitted to the PK data for intact and total CX-2009 from 97 phase 1 patients and 115 phase 2 patients. A 2-compartmental PK model was also developed and fitted to DM4 and Me-DM4 PK data
- Formal exposure-response analysis was performed to identify exposure parameters correlated with selected toxicity endpoints

Biomarker Correlative Methods

- CD166 was measured by IHC as performed using a validated assay by central laboratory
- Gene expression (ALCAM) was measured by RNA sequencing from archival FFPE tumor tissues. Read counts were upper quantile normalized and log2 transferred
- Correlation between target expression and tumor lesion reduction (by investigator review) was calculated by Spearman's rank-order correlation

DEMOGRAPHICS

Table 1. Baseline Demographic and Disease Characteristics – Arms A and B

- 7 mg/kg Q3W was the starting dose for all patients in Arm A and 19 patients in Arm B; the protocol was amended, and the dose was reduced to 6 mg/kg Q3W. An additional 36 patients in Arm B initiated treatment at the reduced 6 mg/kg Q3W dose

	CX-2009 Monotherapy		
	Arm A: 7 mg/kg (n=60)	Arm B: 7 mg/kg (n=19)	Arm B: 6 mg/kg (n=36)
Median age, years (range)	60.5 (36-83)	57.0 (37-75)	52.5 (25-72)
Sex, n (%)			
Male	2 (3.3)	0 (0.0)	0 (0.0)
Female	58 (96.7)	19 (100.0)	36 (100.0)
ECOG performance status, n (%)			
0	29 (48.3)	12 (63.2)	23 (63.9)
1	31 (51.7)	7 (36.8)	13 (36.1)
CD166 IHC H-score at baseline, n (%)[a]			
0-200	21 (35.0)	15 (78.9)	24 (66.7)
201-300	32 (53.3)	4 (21.1)	12 (33.3)
Missing	7 (11.7)	0 (0.0)	0 (0.0)
No. of prior regimens for advanced disease, median (range)[b]	3.5 (1-6)	1.5 (1-3)	2 (1-3)
Types of prior treatments, n (%)			
Platinum-containing compounds	4 (6.7)	11 (57.9)	18 (50.0)
Microtubule inhibitors	42 (70.0)	19 (100.0)	36 (100.0)
mTOR inhibitors	29 (48.3)	0 (0.0)	0 (0.0)
PD-1/PD-L1 inhibitors	4 (6.7)	11 (57.9)	12 (33.3)
Endocrine therapy[c]	59 (98.3)	3 (15.8)	8 (22.2)
Sacituzumab therapy	0 (0.0)	4 (21.1)	4 (11.1)
CDK4/6 inhibitors	60 (100.00	1 (5.3)	1 (2.8)

[a]Patients in arm A missing CD166 data were included into MEE population
[b]Included single-agent hormonal therapy, doublet hormonal therapy, targeted therapy, and cytotoxic therapy
[c]Prior hormone therapy for one additional patient in Arm A was confirmed after data cutoff

SAFETY RESULTS: ARMS A and B

- Grade 3+ AEs related to CX-2009 and serious treatment-related AEs (TRAEs) occurred less frequently at 6 mg/kg than 7 mg/kg
- Treatment discontinuations were lower in patients dosed with 6 mg/kg compared with those dosed with 7 mg/kg (8.3% vs 26.6%)
- Of 27 patients who discontinued treatment due to AEs, 24 were due to TRAE; 13 were caused by neurological events and 9 by ocular events (remaining AEs were nausea, diarrhea and fatigue, all grade 3)
- Five patients experienced grade 4 TRAEs for a total of 7 events. One patient (0.9%) experienced grade 4 severe panenteritis and severe blistering rash as well as grade 5 neutropenia in Arm A. Remaining grade 4 events were neutrophil count decreased, blurred vision, acute kidney injury and hypercalcemia(x2) all of which resolved

Table 2. TRAEs – Arms A and B

	CX-2009 Monotherapy						
Patients reporting AE, n (%)	7 mg/kg (n=79)*			6 mg/kg (n=36)			Total (N=115)
Treatment-related SAEs	9 (11.4)			1 (2.8)			10 (8.7)
TRAEs leading to discontinuation	21(26.6)			3 (8.3)			24 (20.9)
TEAE related to CX-2009 in >10% of patients by preferred term	Grade 2	Grade 3+	All	Grade 2	Grade 3+	All	All
Ocular toxicities	20 (25.3)	11 (13.9)	50 (63.3)	1 (2.8)	1 (2.8)	14 (38.9)	64 (55.7)
Neuropathy/neurotoxicity	26 (32.9)	9 (11.4)	44 (55.7)	7 (19.4)	1 (2.8)	16 (44.4)	60 (52.2)
Fatigue	6 (7.6)	1 (1.3)	28 (35.4)	2 (5.6)	2 (5.6)	8 (22.2)	36 (31.3)
Nausea	10 (12.7)	3 (3.8)	30 (38.0)	2 (5.6)	0	5 (13.9)	35 (30.4)
Infusion-related reaction	9 (11.4)	0	17 (21.5)	7 (19.4)	1 (2.8)	11 (30.6)	28 (24.3)
Diarrhea	2 (2.5)	1 (1.3)	19 (24.1)	1 (2.8)	0	3 (8.3)	22 (19.1)
Decreased appetite	6 (7.6)	0	15 (19.0)	0	0	4 (11.1)	19 (16.5)
ALT increased	0	4 (5.1)	15 (19.0)	0	0	2 (5.6)	17 (14.8)
AST increased	2 (2.5)	2 (2.5)	14 (17.7)	0	0	2 (5.6)	16 (13.9)
Vomiting	2 (2.5)	2 (2.5)	12 (15.2)	1 (2.8)	0	4 (11.1)	16 (13.9)
Myalgia	9 (11.4)	0	14 (17.7)	0	0	1 (2.8)	15 (13.0)
Headache	3 (3.8)	0	12 (15.2)	0	0	1 (2.8)	13 (11.3)

*Patients dosed at 7 mg/kg in Arms A and B were combined for AE reporting purposes

EFFICACY RESULTS ARM A: HR+/HER- Breast Cancer

- Arm A met the primary efficacy endpoint with a confirmed ORR by central radiology review of 14.3% (n=49) (**Table 3**)
- All 60 patients received 7 mg/kg Q3W as their starting dose
- Significant tumor reduction noted at six week assessment

Arm A: HR+/HER2- Breast Cancer

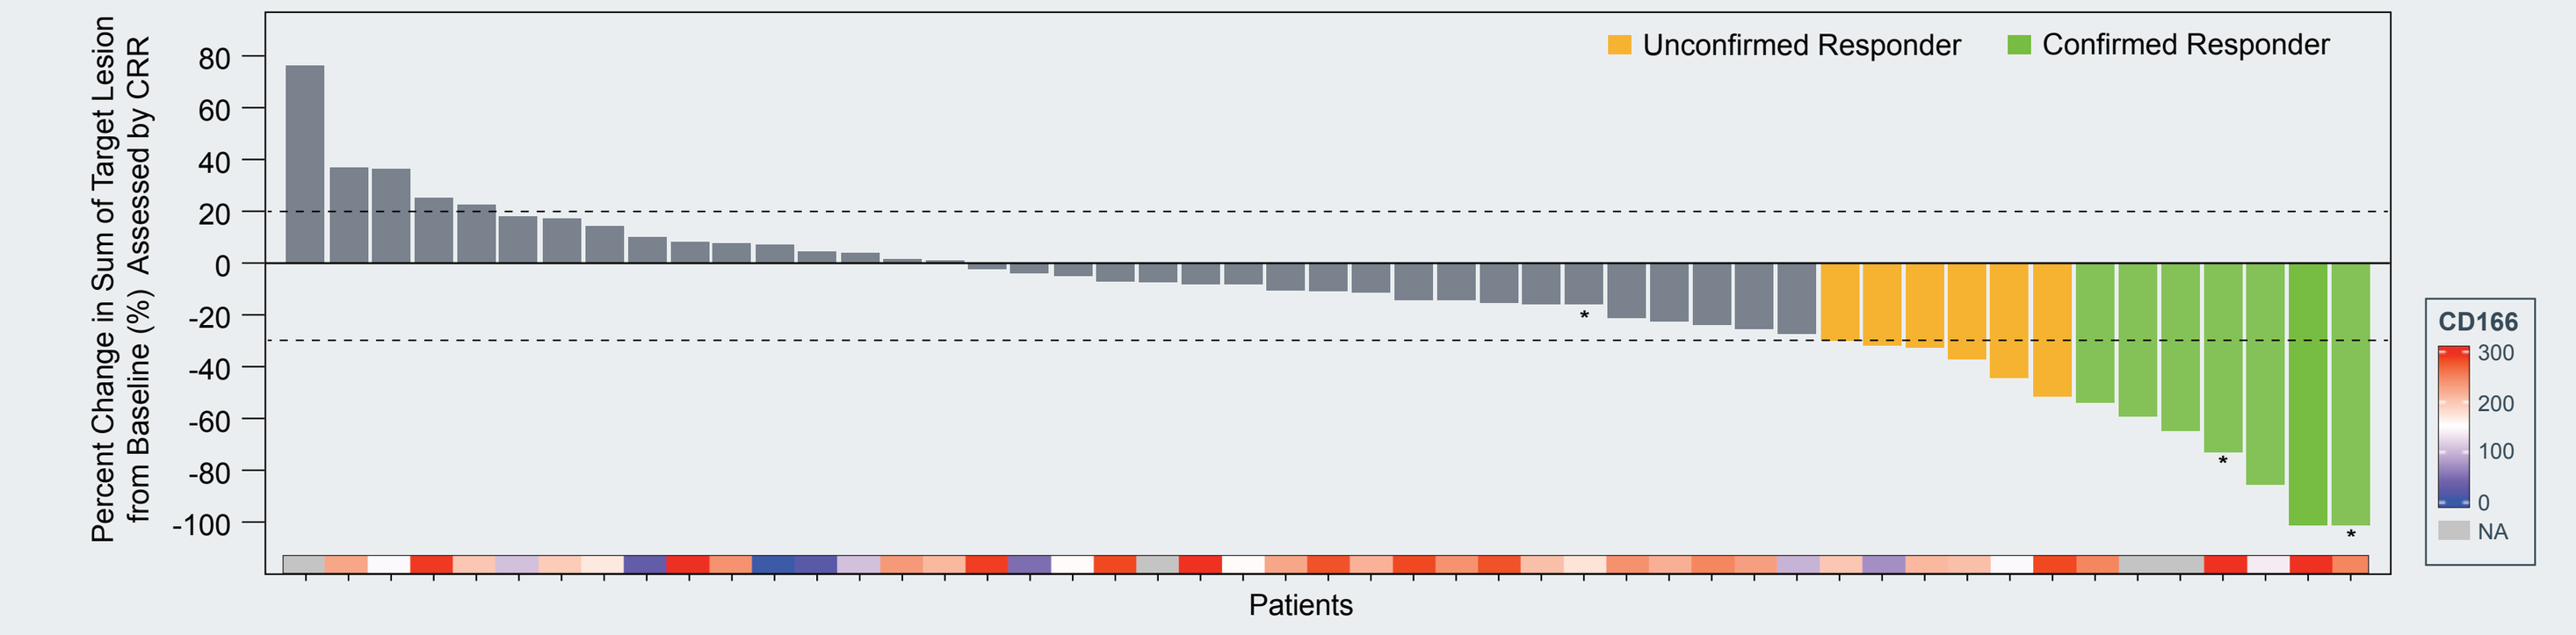
Unconfirmed Responder
Confirmed Responder
Percent Change in Sum of Target Lesion from Baseline (%) Assessed by CRR
Patients
CD166
300
200
100
0
NA

Arm A: HR+/HER2- Breast Cancer

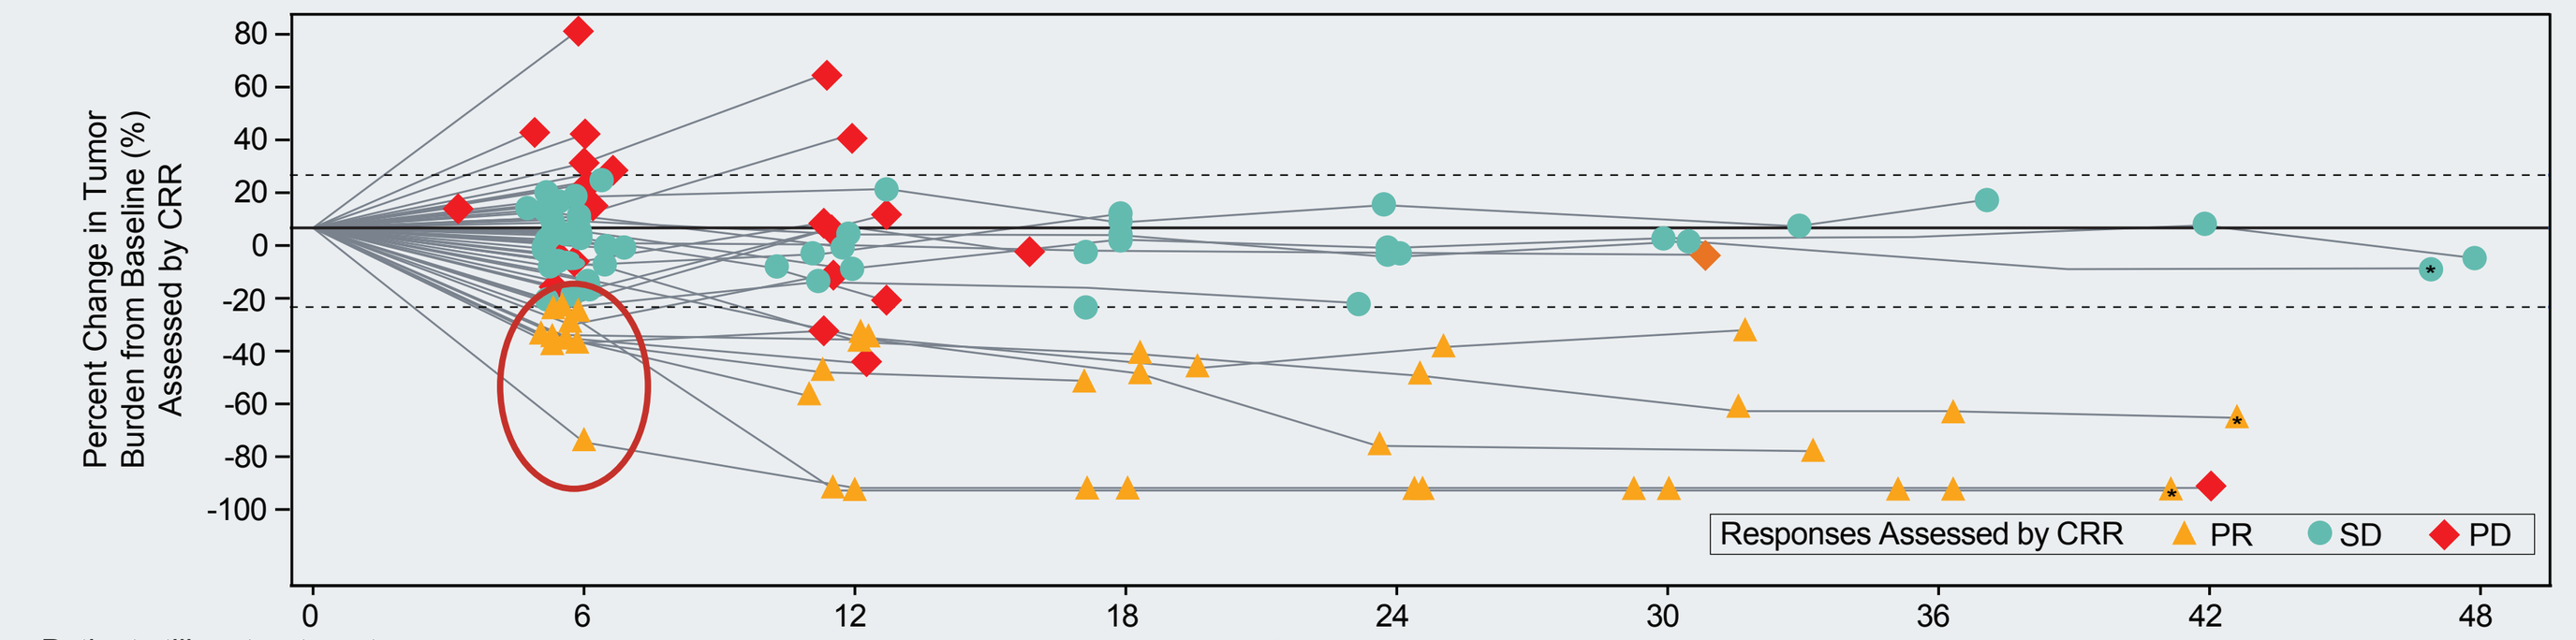
Percent Change in Tumor Burden from Baseline (%) Assessed by CRR
Weeks Since Treatment Initiation
Responses Assessed by CRR
PR
SD
PD

* Patient still on treatment.
Red circle shows significant tumor reduction at six-week assessment.

Table 3. Response in Modified Efficacy Evaluable Patient Population as Assessed by Investigator and Central Radiology

Population, n (%)	Investigator n=52*	Central Radiology n=49**
ORR	8 (15.4%)	7 (14.3%)
CR (confirmed or unconfirmed)	0	0
PR (confirmed or unconfirmed)	17 (32.7%)	13 (26.5%)
Confirmed PR	8 (15.4%)	7 (14.3%)
Unconfirmed PR	9 (17.3%)	6 (12.2%)
SD (%)	17 (32.7%)	24 (49.0%)
PD (%)	18 (34.6%)	11 (22.4%)
CBR24 (confirmed or unconfirmed) (95% CI)	21 (27.0, 54.9)	17 (21.7, 49.6)
Confirmed	12 (23.1%)	11 (22.4%)
Unconfirmed	13 (25.0%)	10 (20.4%)
PFS, median in months (95% CI) [min, max]	2.6 (2.1, 3.2) [1.1, 11.1]	2.6 (1.5, 2.7) [1.4, 7.1]

*Of 60 patients in Arm A safety population, 6 discontinued treatment prior to first assessment due to AE, 1 withdrew consent and 1 started different systemic therapy making them not evaluable for tumor response. **Three additional patients could not be assessed by CRR due to not having imaging of measurable disease at baseline.

EFFICACY RESULTS ARM B: TNBC

- Arm B did not pass protocol-defined futility boundary of 10% ORR in TNBC
- Confirmed partial responses were observed at 6 mg/kg Q3W

Arm B: TNBC, Started CX-2009 6 mg/kg

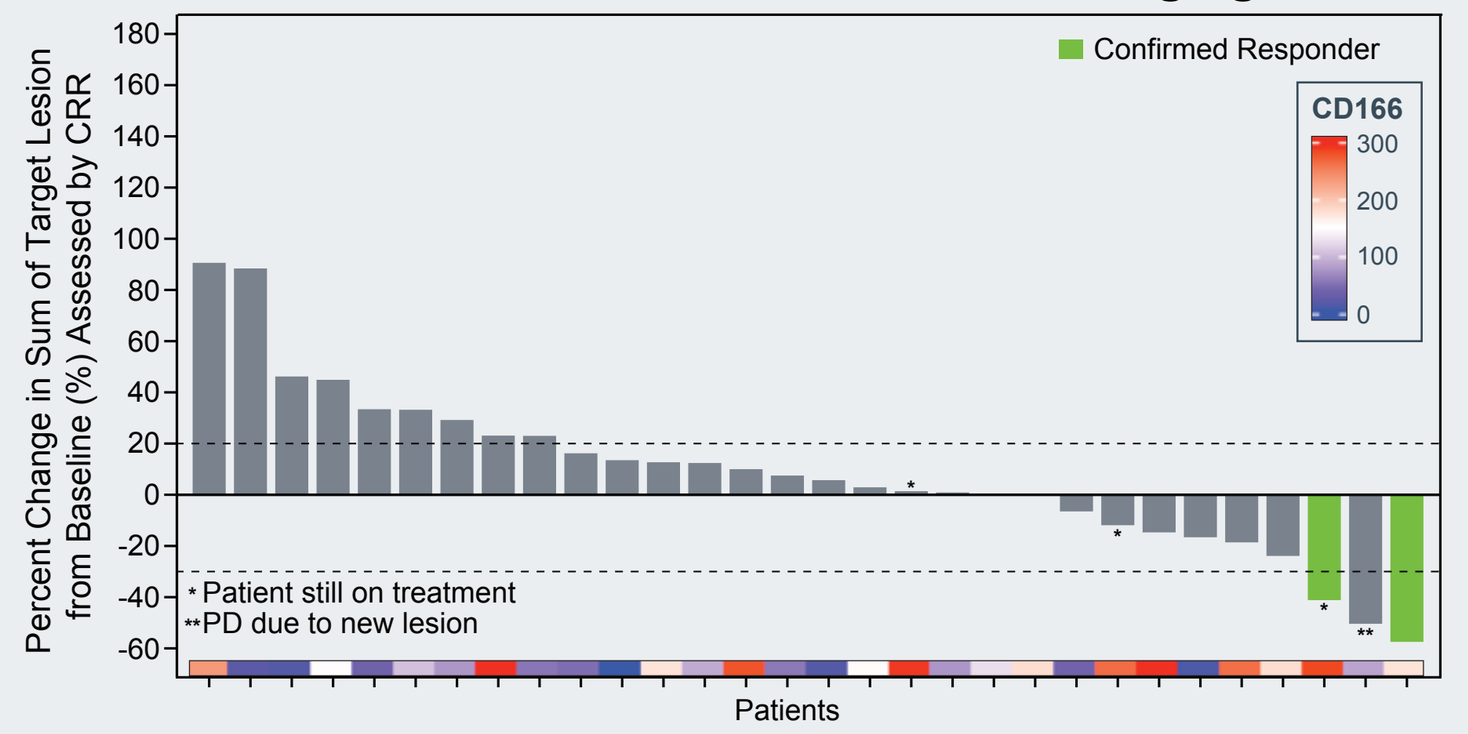
Confirmed Responder
CD166
Percent Change in Sum of Target Lesion from Baseline (%) Assessed by CRR
* Patient still on treatment
** PD due to new lesion
Patients

Of 36 patients in Arm B 6 mg/kg Q3W safety population, 1 patient discontinued treatment prior to first assessment due to AE, 2 patients could not be assessed by CRR due to not having imaging of measurable disease at baseline, and 3 patients did not have tumor measurements recorded prior to data cutoff.

Arm B: TNBC, Started CX-2009 7 mg/kg

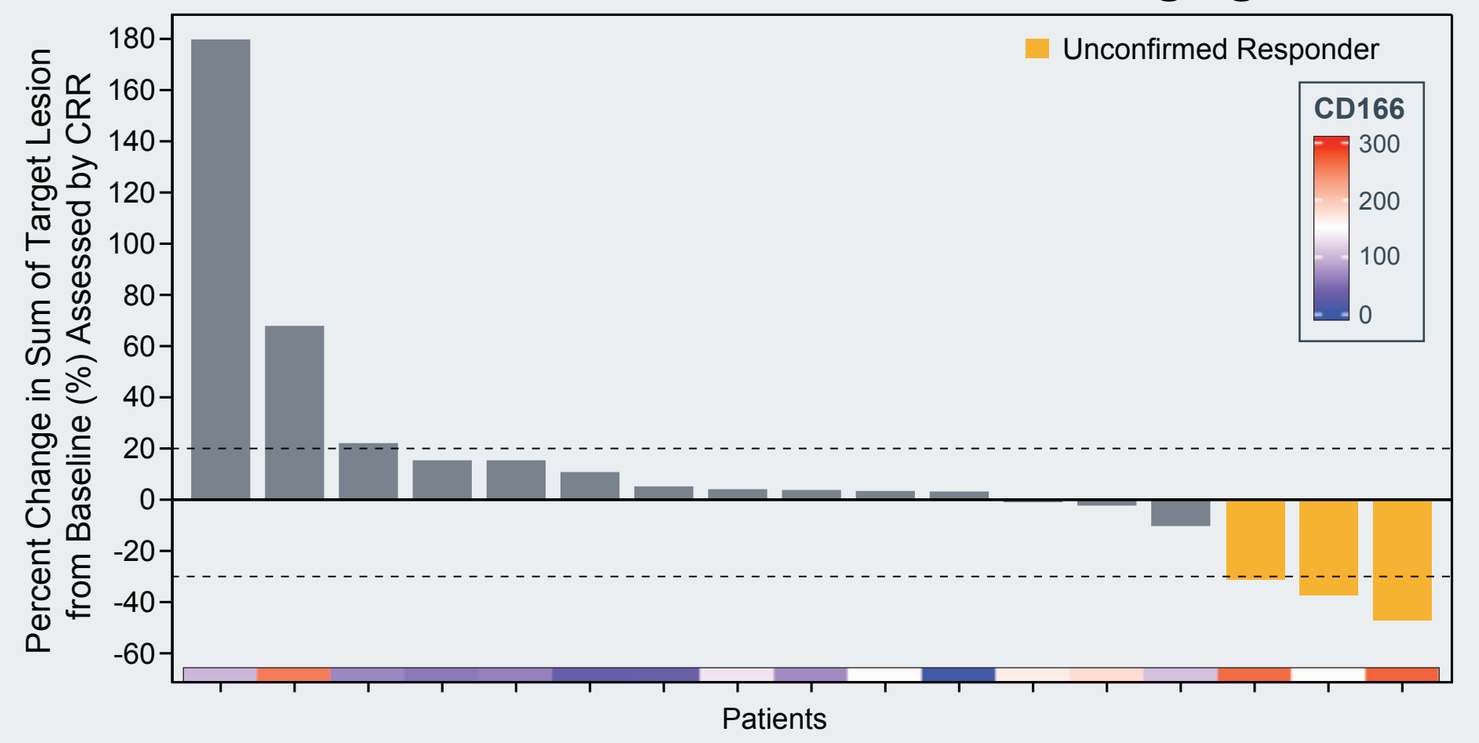
Unconfirmed Responder
CD166
Percent Change in Sum of Target Lesion from Baseline (%) Assessed by CRR
Patients

Of 19 patients in Arm B 7 mg/kg Q3W safety population, 1 patient could not be assessed by CRR due to not having imaging of measurable disease at baseline, and 1 patient did not have tumor measurements recorded prior to data cutoff.

CONCLUSIONS

- Arm A met the study primary endpoint with an ORR of 14.3% by central radiology review in patients with HR+/HER2- ABC (median of 3.5 prior regimens); mPFS of 2.6 (range, 1.4 - 7.1) months was observed
- Durability of response appeared to be limited by the higher than anticipated toxicity at the 7mg/kg dose
- A lower dose of 6 mg/kg Q3W appears to be better tolerated than 7 mg/kg Q3W
 - Fewer AEs leading to discontinuation (8.3% vs 26.6%)
 - Decreased incidence of grade 3 and higher ocular toxicity, as well as grade 2 and above neuropathy
 - Anti-tumor activity was observed when dosing with 6 mg/kg dose (Arm B)
- No on-target, off-tumor toxicities were observed, suggesting antibody masking was effective in normal tissues, and overall toxicity profile was generally consistent with a DM4 payload
- CD166 is expressed significantly higher at the RNA and protein level in HR+ compared with TNBC
- In TNBC, CD166 protein level by IHC is significantly correlated with tumor lesion reduction, suggesting a patient selection strategy may be effective
- Additional clinical studies in HR+ advanced breast cancer, incorporating a starting dose of 6 mg/kg and possible biomarker strategies, are warranted

POPULATION PK RESULTS

- Grade 3 ocular toxicity was highly correlated with intact CX-2009 exposure and to a lower extent with C_{trough} of DM4 (p=0.04). AUC was used for building the regression model (p<0.001)
- Grade 2 neuropathy was correlated with C_{trough} of DM4 (p=0.04) and CX-2009 (p=0.03)
- Probability of discontinuation due to any AE was correlated with intact CX-2009 exposure. AUC of CX-2009 was used in the final exposure-response model (p<0.001)
- This modeling supports the clinical data observed in Arm B and suggests that reducing the dose to 6 mg/kg could lower probabilities of grade 3 and higher ocular toxicity, grade 2 and higher neuropathy, and discontinuation due to any AE

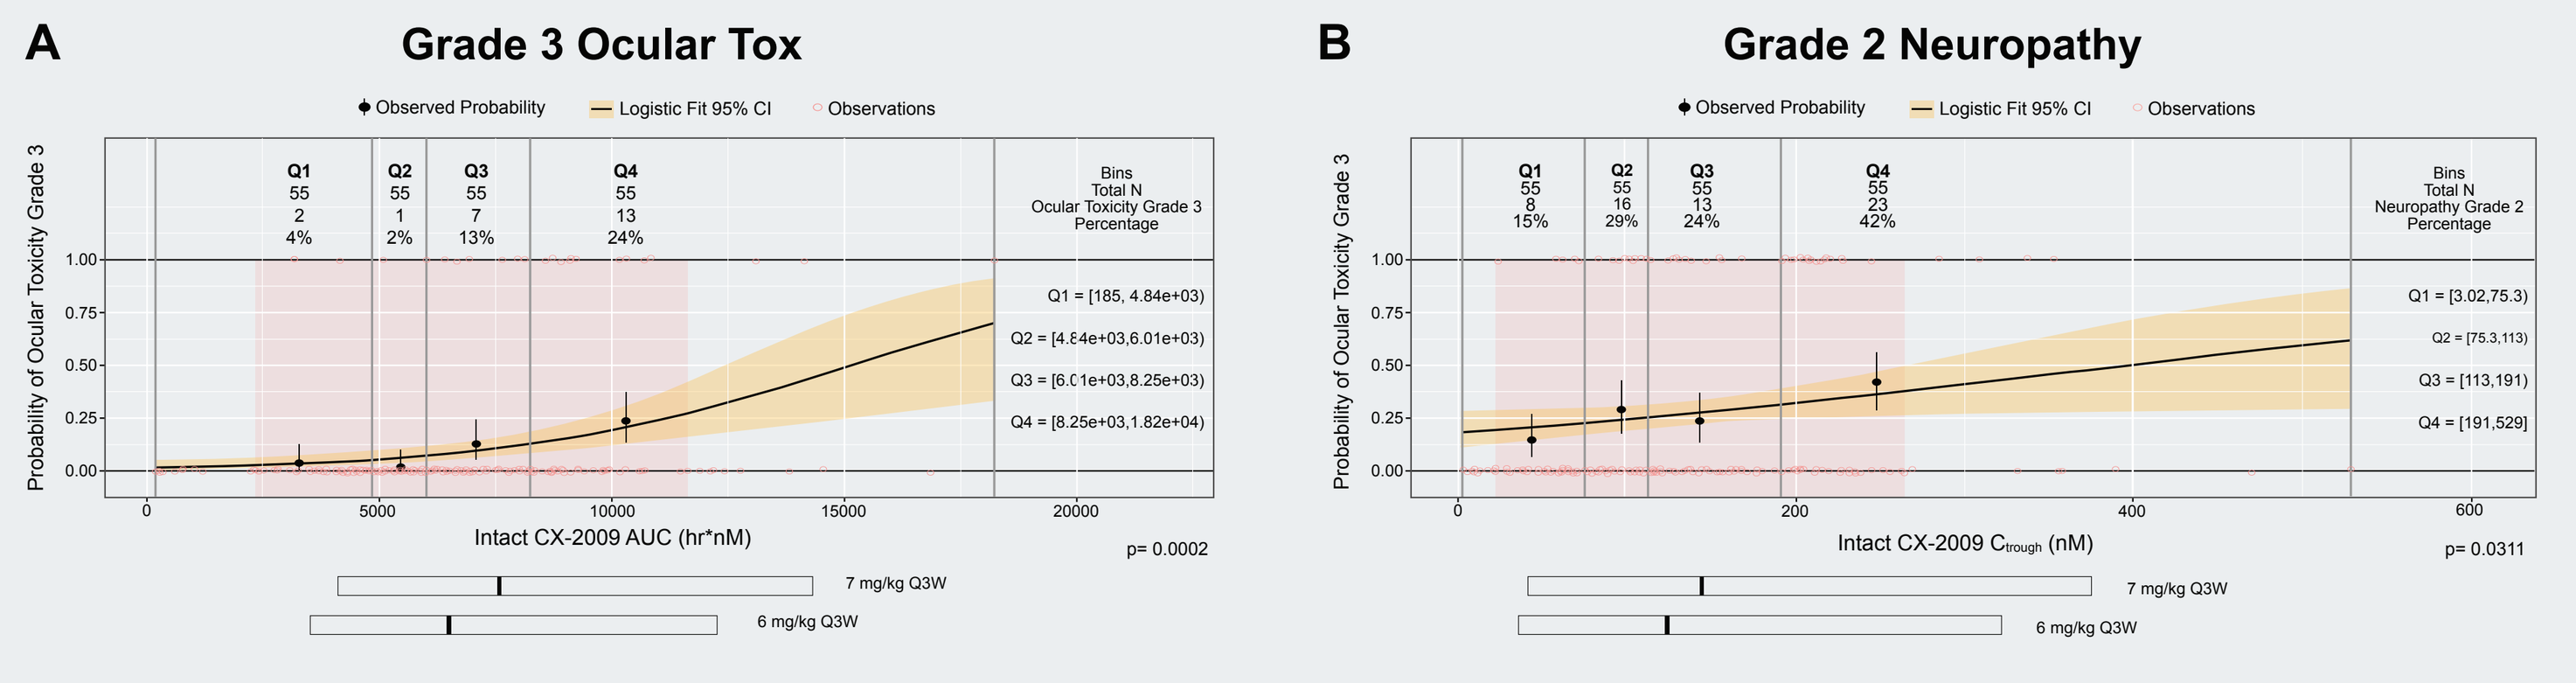
A
Grade 3 Ocular Tox
Intact CX-2009 AUC (hr*nM)
p= 0.0002
7 mg/kg Q3W
6 mg/kg Q3W
B
Grade 2 Neuropathy
Intact CX-2009 Ctrough (nM)
p= 0.0311

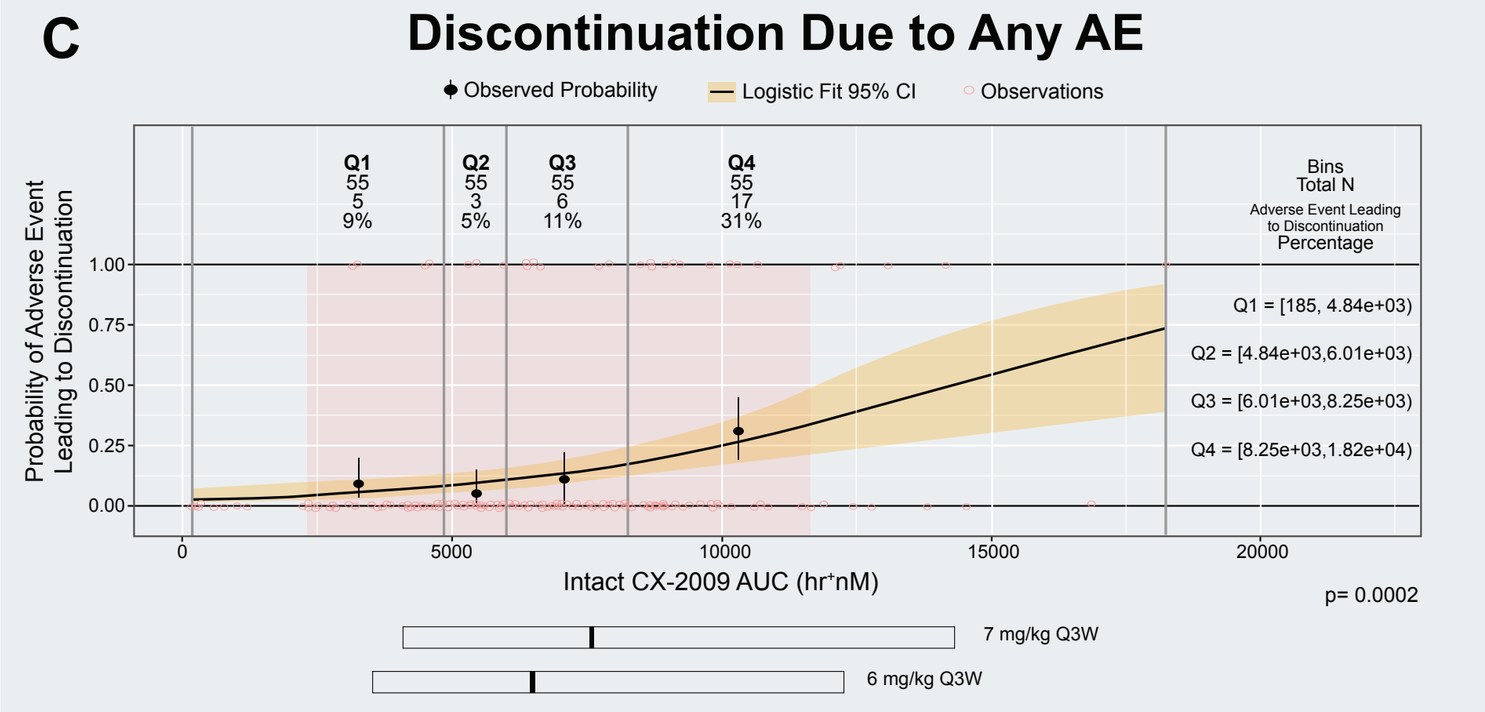
C
Discontinuation Due to Any AE
Intact CX-2009 AUC (hr*nM)
p= 0.0002
7 mg/kg Q3W
6 mg/kg Q3W

A. Logistic regression analysis to assess probability of grade 3 and above ocular tox. Quartiles of AUC are indicated. The AUC ranges for 7 mg/kg and 6 mg/kg Q3W shown, **B.** Probability of grade 2 neuropathy correlated with C_{trough} with quartiles included, **C.** Probability of discontinuation due to any AE is correlated with AUC. The ranges of exposures are shown.

BIOMARKER RESULTS

The biomarker strategy was hypothesis-generating and warrants further validation

- CD166 is expressed at significantly higher levels at the RNA and protein level in HR+ compared to TNBC
- In TNBC, CD166 protein level by IHC is significantly correlated with tumor lesion reduction

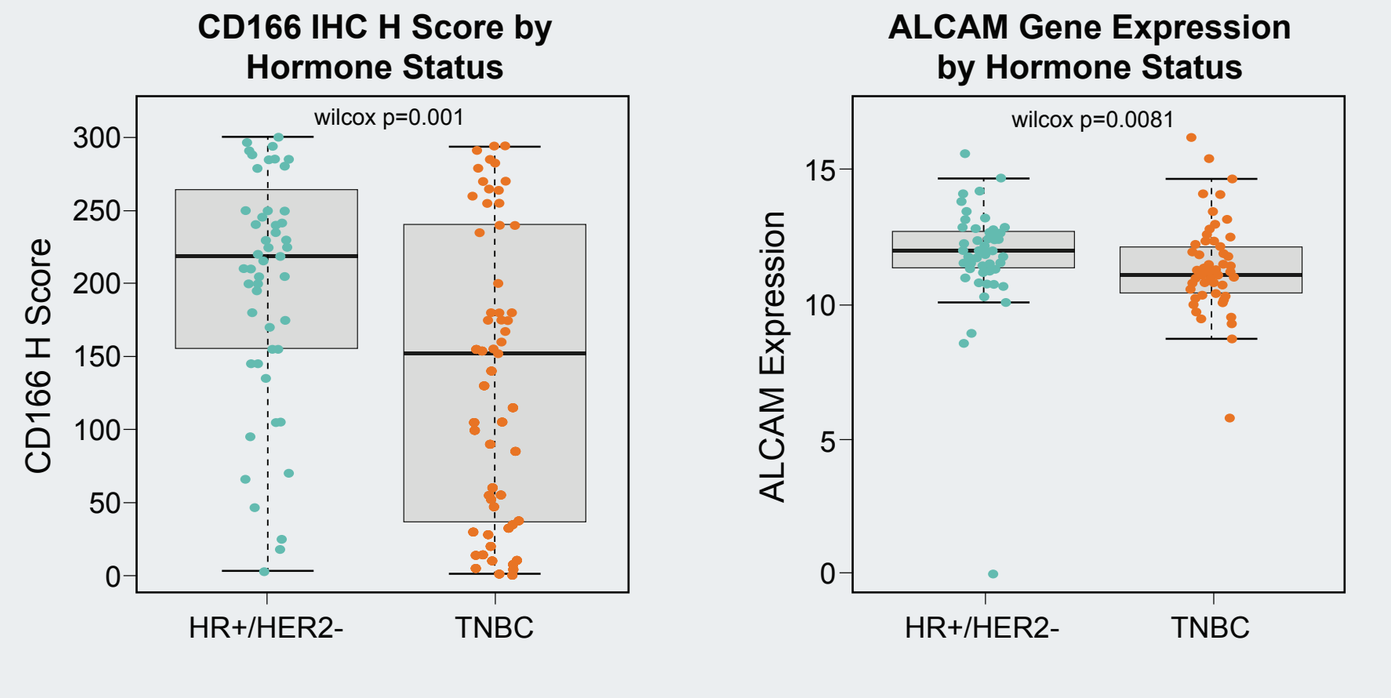
CD166 IHC H Score by Hormone Status
wilcox p=0.001
CD166 H Score
ALCAM Gene Expression by Hormone Status
wilcox p=0.0081
ALCAM Expression
HR+/HER2-
TNBC

Boxplots of target expression for HR+ and TNBC populations.
Left: CD166 IHC H scores. Right: Normalized ALCAM mRNA expression in log2.

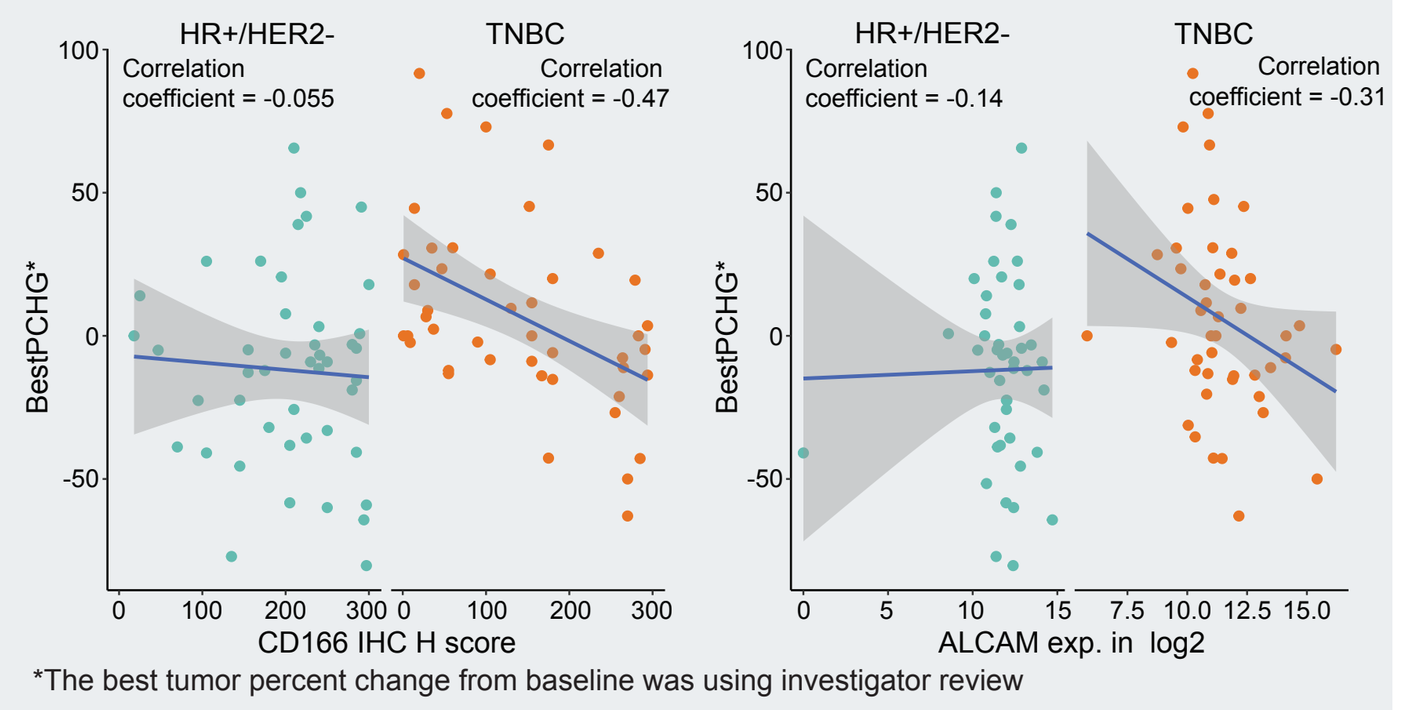
HR+/HER2-
Correlation coefficent = -0.055
TNBC
Correlation coefficent = -0.47
HR+/HER2-
Correlation coefficent = -0.14
TNBC
Correlation coefficent = -0.31
BestPCHG*
CD166 IHC H score
ALCAM exp. in log2

*The best tumor percent change from baseline was using investigator review

Scatter plots of target expression and best tumor percent change (BestPCHG). Spearman rank correlation rho is shown on the top-left corner of each plot. In TNBC, both protein and mRNA expression was significantly correlated with BestPCHG (p-value= 0.001 and 0.04, respectively), but the strong correlation was not seen in HR+. Target expression is also predictive in CBR, DCR and ORR in TNBC (Arm B), but not in HR+ (data not shown).

REFERENCES

1. Donaghy H. *mAbs.* 2016;8:659-671.
2. Deu E, et al. *Nature Struct Mol Biol.* 2012;19:9-16.
3. von Lersner A, et al. *Clin Exp Metastasis.* 2019;36:87-95.
4. Cancer Genome Atlas Research Network. *Nat Genet.* 2013;45(10):1113-1120.
5. Saal LH. *Genome Med.* 2015;7(1):20.
6. Curtis C. *Nature.* 2012;486(7403):346-352.
7. Boni V, et al. *Clin Cancer Res.* 2022;28:2020-2029.

Please contact the author at kathmill@iupui.edu for permission to reprint and/or distribute.
PROBODY is a U.S. registered trademark of CytomX Therapeutics, Inc.