435P

Preliminary Results of PROCLAIM-CX-072: The First-in-Human, Dose-Finding Trial of PD-L1 Probody Therapeutic CX-072 as Monotherapy in Patients With Advanced Solid Tumors

Valentina Boni,[1] Javier Garcia-Corbacho,[2] Patrick A. Ott,[3] Daniel Cho,[4] Karen A. Autio,[5] Nataliya Uboha,[6] Alexander I. Spira,[7] Rachel Humphrey,[8] Matthias Will,[8] Aung Naing,[9] Fiona Thistlethwaite,[10] Elisabeth de Vries,[11] Hendrik-Tobias Arkenau[12]

[1]START Madrid–CIOCC HM University Hospital Sanchinarro, Madrid, Spain; [2]Hospital Clinic Barcelona, Barcelona, Spain; [3]Dana-Farber Cancer Institute, Boston, MA, USA; [4]New York University, New York, NY, USA; [5]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [6]University of Wisconsin Carbone Cancer Center, Madison, WI, USA; [7]Virginia Cancer Specialists, Fairfax, VA, USA; [8]CytomX Therapeutics, South San Francisco, CA, USA; [9]The MD Anderson Cancer Center, Houston, TX, USA; [10]The Christie NHS Foundation Trust and University of Manchester, Manchester, United Kingdom; [11]University Medical Center, Groningen, Netherlands; [12]Sarah Cannon Research Institute UK Limited, London, United Kingdom

BACKGROUND

- Programmed cell death ligand 1 (PD-L1) is expressed on many cancer and immune cells; by binding the programmed cell death 1 (PD-1) receptor, a negative regulator of T-lymphocyte activation, PD-L1 can block cancer immune detection[1]
- Monoclonal antibodies targeting the PD-1 pathway and other immune-checkpoint inhibitors have shown anticancer activity in different tumor types[1]
- Immune-related adverse events (irAEs), including interstitial pneumonitis, colitis, and transaminitis, are known toxicities of PD-1/PD-L1 axis-blocking antibodies and are commonly treated with steroids[2,3]
- irAEs are widely observed with the use of inhibitors of the PD-1/PD-L1 axis[3,4]
- Probody™ therapeutics are fully recombinant antibody prodrugs that are designed to remain relatively inactive systemically and in healthy tissue, minimizing binding to the target antigen, and to be activated specifically in the tumor microenvironment by tumor-associated protease activity[5,6]
- CX-072 is an investigational Probody therapeutic directed against PD-L1 and designed to have anticancer activity with potentially reduced irAEs
- The PROCLAIM-CX-072 (**PRO**body **CL**inical **A**ssessment **I**n **M**an) study is evaluating the tolerability and preliminary antitumor activity of multiple doses and the expansion of Probody CX-072 as monotherapy (Parts A[7] and A2) and various other regimens, including combination therapy with ipilimumab (ClinicalTrials.gov identifier, NCT03013491)
- Here we report interim safety and efficacy results of the monotherapy dose-escalation cohorts 0.03 to 30 mg/kg (Parts A and A2). Translational data will be presented at a future date. See poster 436P for an update on Part B1 (combination with ipilimumab)

OBJECTIVES AND END POINTS

- Primary objectives of Parts A and A2 of the study are to assess the safety and tolerability and to determine the maximum tolerated dose (MTD) and the dose-limiting toxicity of CX-072 when administered as monotherapy
 - Part A (define MTD): Dose escalation (3+3 design; 0.03-30 mg/kg) in unselected solid tumors
 - Part A2 (biomarker analyses): Biomarker cohorts in PD-L1–positive solid tumors (n = 6 per dose level) in select dose levels (0.3-10 mg/kg)
- Secondary objectives are to obtain preliminary evidence of anticancer activity in patients treated with CX-072 using
 - Response rate (Response Evaluation Criteria in Solid Tumors [RECIST] v1.1)
 - Time to response and duration of response
 - Progression-free survival
- Secondary objectives also include characterization of the single-dose and multidose pharmacokinetic (PK) profiles of CX-072 when administered alone
- Exploratory objectives include investigation of the immunomodulatory activity of CX-072 in on-treatment biopsy specimens

METHODS

Study Design

- This is a first-in-human, open-label, dose-escalation, multicenter, phase 1/2a study of CX-072 designed to evaluate the safety and determine the MTD and/or maximum achieved dose of CX-072 as monotherapy (**Figure 1**; Part A, dose escalation; Part A2, biomarker cohort)

Figure 1. PROCLAIM-CX-072 Parts A and A2 study design.

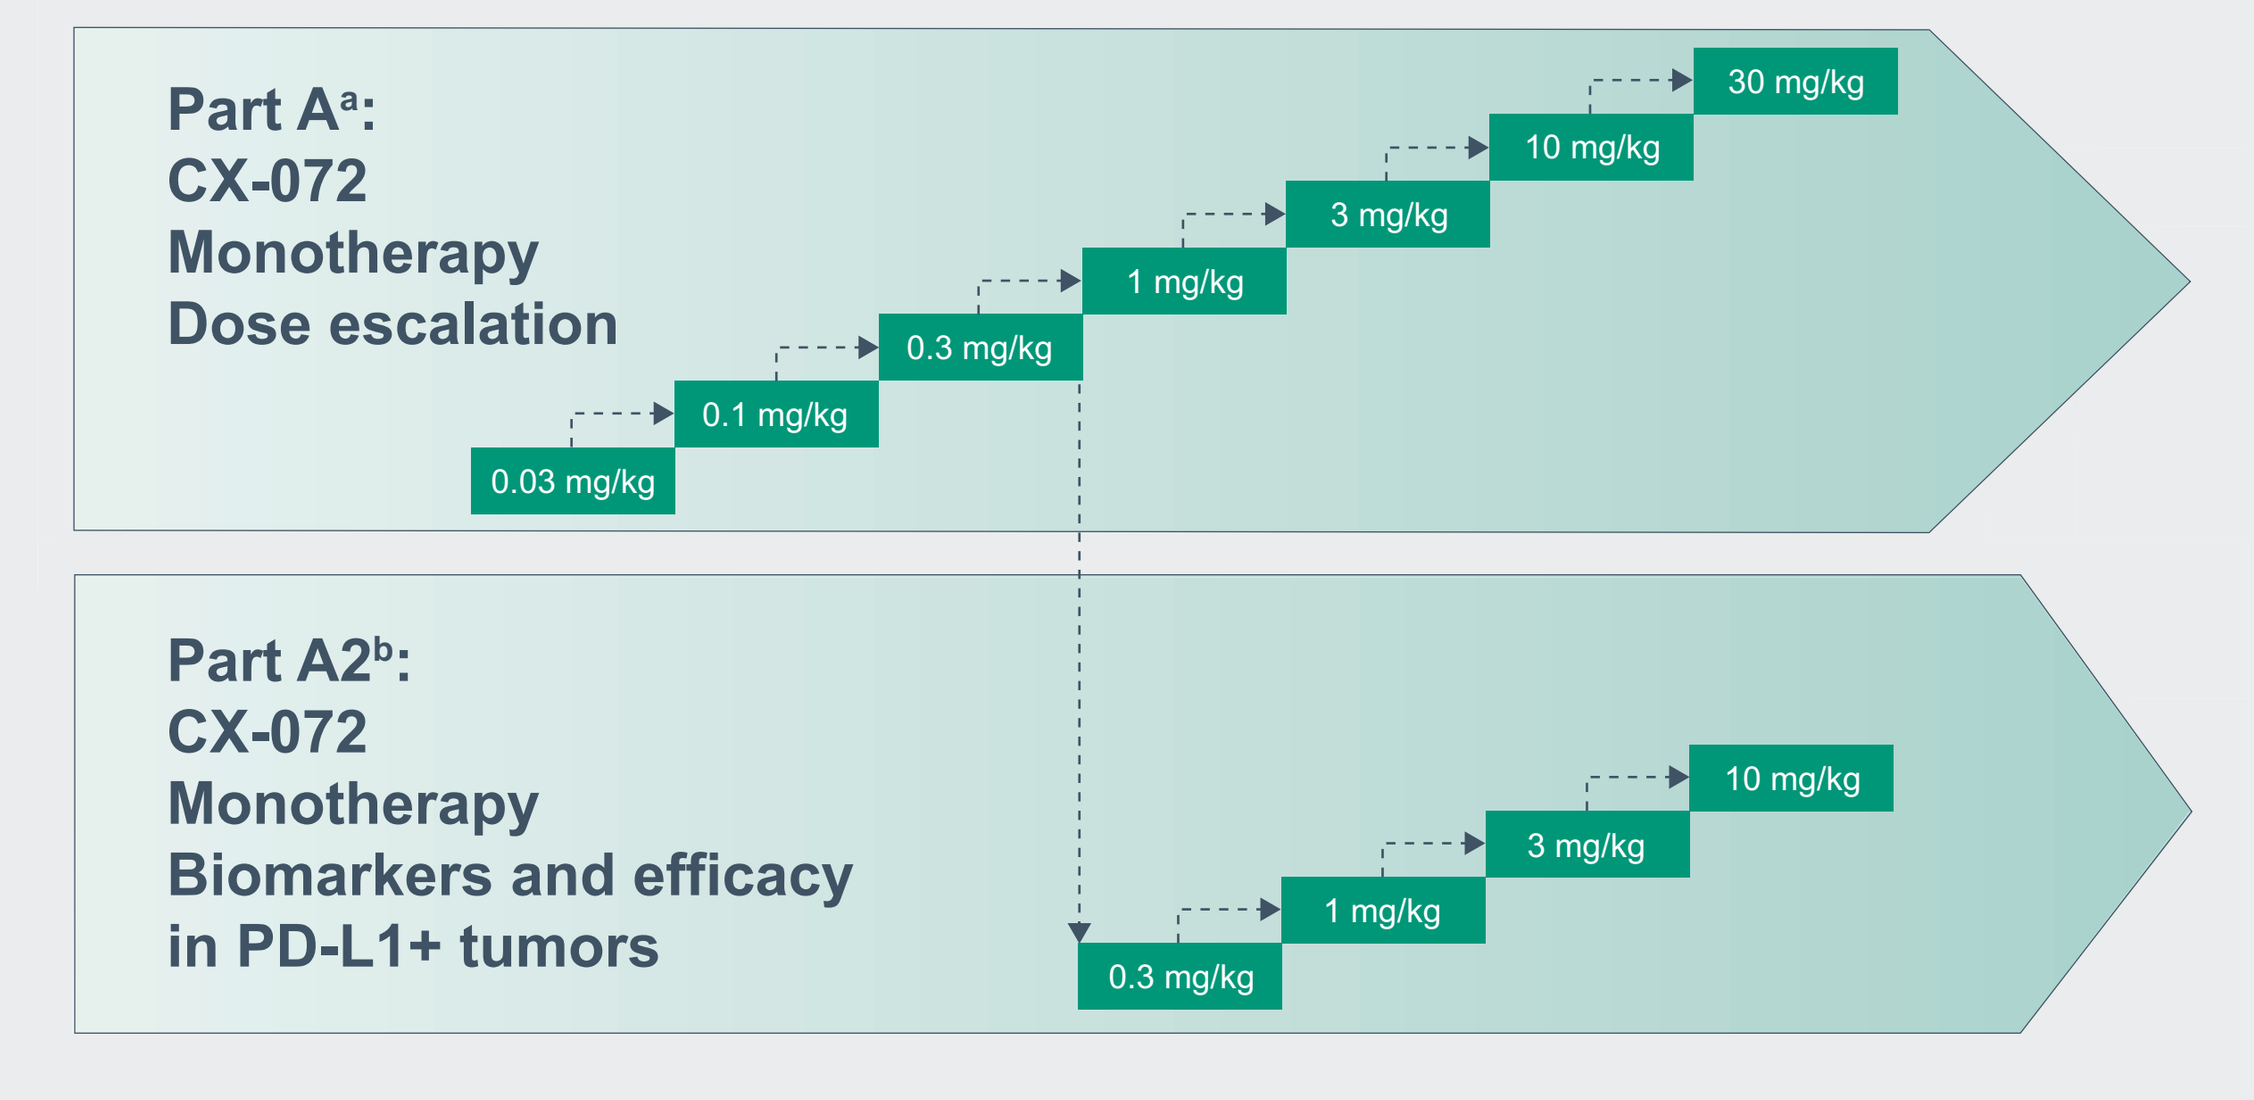
Part A[a]:
CX-072
Monotherapy
Dose escalation
0.03 mg/kg
0.1 mg/kg
0.3 mg/kg
1 mg/kg
3 mg/kg
10 mg/kg
30 mg/kg
Part A2[b]:
CX-072
Monotherapy
Biomarkers and efficacy
in PD-L1+ tumors
0.3 mg/kg
1 mg/kg
3 mg/kg
10 mg/kg

[a]CX-072 monotherapy was administered intravenously every 14 days. One patient each was enrolled in the 0.03, 0.1, and 0.3 mg/kg dosing cohorts, and subsequent dose levels follow a 3+3 design, which is also used for all other dose-escalation groups.
[b]After successful completion of the monotherapy dose level in Part A, Part A2 has enrolled 6 additional patients with PD-L1–positive cancer at each indicated dose to refine the maximum tolerated dose/multiple ascending dose and to evaluate the relationship between dose/exposure and safety, efficacy, and pharmacodynamic biomarkers.

Patients

- Patients are ≥18 years of age and have Eastern Cooperative Oncology Group performance status 0 to 1
- To be included in Part A, patients are required
 - To have any type of metastatic or advanced unresectable solid tumor or lymphoma (measurable or nonmeasurable disease)
 - To be naive to immunotherapy, including PD-1/PD-L1 and cytotoxic T-lymphocyte antigen 4 inhibitor therapy, and to have a tumor type for which a checkpoint inhibitor is not available for therapy
- To be included in Part A2, patients are additionally required
 - To participate in biomarker analysis and have a tumor site that is safe to biopsy
 - To have PD-L1 expression ≥1%, defined as tumor proportion score ≥1% of membranous staining based on the DAKO PD-L1 IHC 22C3 pharmDx, in order to measure activation in the tumor tissue
- CX-072 monotherapy (0.03, 0.1, 0.3, 1, 3, 10, 30 mg/kg) is administered intravenously every 14 days without any premedication

Safety Assessments

- Adverse events (AEs) are assessed and reviewed before each infusion and at any other visit that includes a physical examination. AEs are coded using MedDRA v19.1 and graded using the National Cancer Institute Common Terminology Criteria for Adverse Events v4.03
- irAEs are defined as treatment-related AEs (TRAEs) that were on a predefined list of >300 preferred terms and that required treatment with systemic steroids within 30 days of the onset of the relevant AE

Tumor Response and Translational Evaluation

- Imaging for tumor response assessment using RECIST v1.1 is performed every 8 weeks for the first 12 months, then every 12 weeks thereafter
- After the last dose of study medication, patients who do not discontinue because of progressive disease will be evaluated every 3 months for disease progression
- All patients will be evaluated for overall survival until study withdrawal or death
- Archival tissue or fresh biopsy samples are provided at baseline. On-treatment biopsies were optional in Part A and required for participation in Part A2

RESULTS

Patients

- As of August 3, 2018, 46 patients were enrolled and 11 patients (24%) were still receiving treatment
 - 35 patients discontinued treatment because of radiologic or clinical disease progression (n = 28), AE (n = 5), or voluntary withdrawal (n = 2)
- Baseline characteristics are presented in **Table 1**

Table 1. Baseline Characteristics of Patients Treated With CX-072

Baseline Patient Characteristics	All Patients N = 46
Median age, years (range)	64 (22-81)
Sex, n (%)	
Female	24 (52)
Male	22 (48)
Race, n (%)	
White	37 (80)
African American	1 (2)
Asian	1 (2)
Not reported/unknown/other	7 (15)
ECOG performance status, n (%)	
0	19 (41)
1	27 (59)
No. of previous cancer treatments, median (range)	3 (1-13)
Cancer type, n (%)	
Thymoma or thymic cancer	10 (22)
Castration-resistant prostate cancer	4 (9)
Esophageal carcinoma	4 (9)
Cervix carcinoma	3 (7)
Pancreatic carcinoma	3 (7)
Uterine carcinoma	3 (7)
Gastric cancer	2 (4)
Rectal carcinoma	2 (4)
Triple-negative breast cancer	2 (4)
Uterine sarcoma	2 (4)
Other[a]	11 (24)
PD-L1 expression status, n (%)	
None (<1%)	11 (24)
Low (1-49%)	17 (37)
High (≥50%)	13 (28)
Unknown	5 (11)

ECOG, Eastern Cooperative Oncology Group; ER, estrogen receptor; PD-L1, programmed cell death-ligand 1.
Percentages may not add up to 100% due to rounding.
[a]One patient each had adrenal carcinoma, anal carcinoma, appendix carcinoma, breast (ER+) carcinoma, cholangiocellular carcinoma, colon carcinoma, head and neck squamous cell carcinoma, mesothelioma, ovarian carcinoma, peritoneal carcinoma, or salivary gland carcinoma.

Duration of Treatment

- Median (range) durations of treatment are reported in **Table 2**

Table 2. Duration of CX-072 Treatment

CX-072 Dose, mg/kg	≤1 n = 21	3 n = 13	10 n = 9	30 n = 3	All Patients N = 46
Median treatment duration (range), months	3.6 (1-7)	1.5 (0-9)	1.8 (1-11)	2.4 (2-4)	2.0 (0-11)

Pharmacokinetic Analysis

- Preliminary single-agent, single-dose CX-072 PK data (**Figure 2A**) suggest that CX-072
 - Circulates predominantly as the intact prodrug species (96% intact at 30 mg/kg)
 - Is likely only minimally influenced by target-mediated drug disposition (TMDD) at low doses
- In contrast, the PD-L1 inhibitor atezolizumab exhibited nonlinear PK below the 1 mg/kg dose level, which may be attributable to TMDD (**Figure 2B**)

Figure 2. (A) Dose 1 median concentration of intact (solid colored lines) and total (dashed colored lines) CX-072 versus time after administration of up to 30 mg/kg CX-072 every 2 weeks.[a,b] (B) Dose 1 mean serum concentrations of atezolizumab.

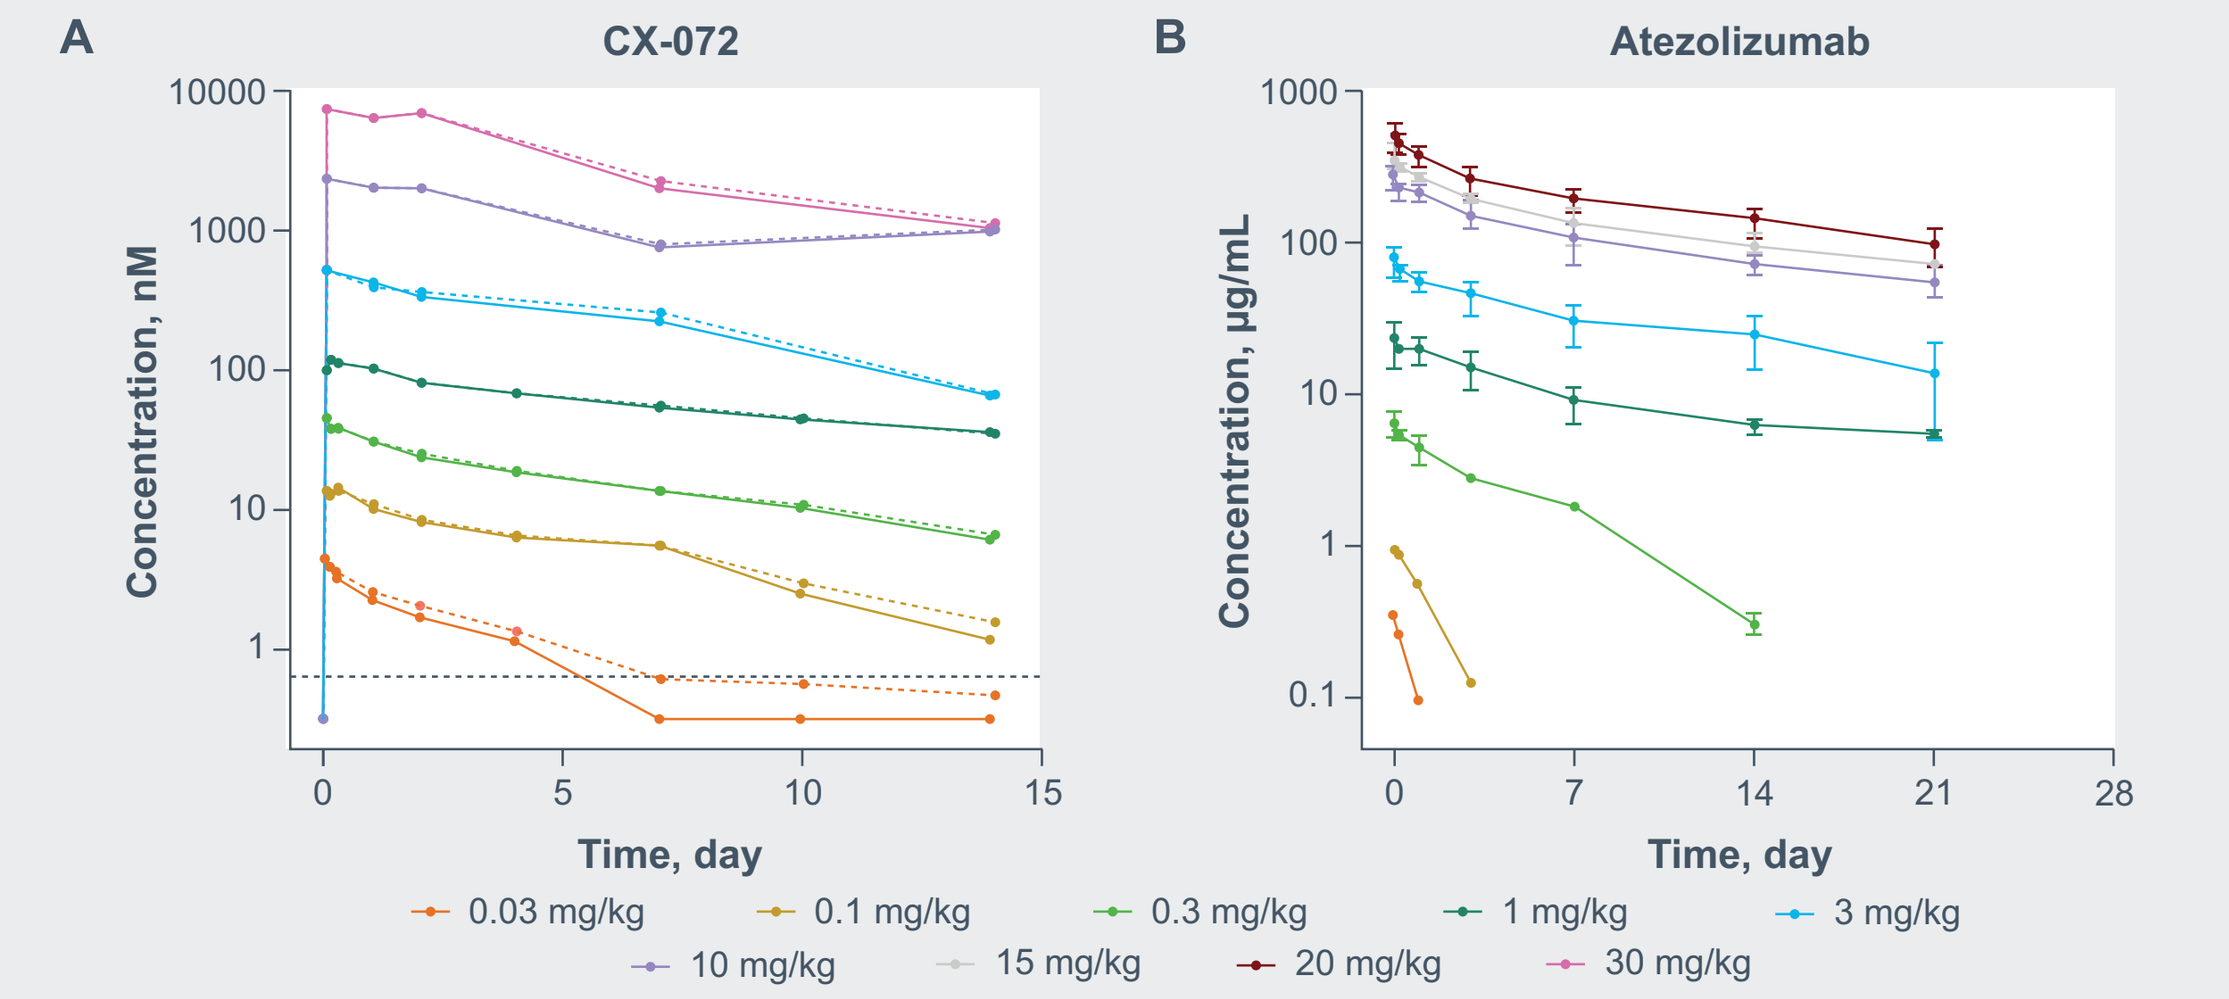
A
CX-072
Concentration, nM
Time, day
B
Atezolizumab
Concentration, µg/mL
Time, day
0.03 mg/kg
0.1 mg/kg
0.3 mg/kg
1 mg/kg
3 mg/kg
10 mg/kg
15 mg/kg
20 mg/kg
30 mg/kg

LOQ, limit of quantitation.
[a]Gray dashed line represents LOQ for CX-072 assay, and data below the LOQ are assigned a value of LOQ/2.
[b]Cohort A, cycle 1, dose 1.
Figure 1B is reprinted by permission from Springer Nature. Herbst RS et al. Predictive correlates of response to the anti-PD-L1 antibody MPDL3280A in cancer patients. *Nature.* 2014;515:563-567. Copyright © 2014.

Safety

- AEs are summarized in **Table 3**
- The MTD was not reached
- All-grade TRAEs, grade 3-4 TRAEs, and irAEs occurred in 54%, 11%, and 7% of patients, respectively
 - 3 patients (7%) experienced a treatment-related serious AE
 - Neutropenia and thrombocytopenia (grade 3) in a patient with thymic cancer
 - Dyspnea and pneumonitis (grade 3) in a patient with thymoma
 - Stress-induced cardiomyopathy (grade 4) as a result of an infusion-related reaction (IRR) in a patient with prostate cancer and a preexisting heart condition
- The rate of TRAEs leading to discontinuation of CX-072 was 11%
- IRRs were observed in 10 of 46 patients (22%), 2 of whom experienced a grade 3-4 IRR; only 1 patient discontinued treatment because of an IRR
- The most common treatment-emergent AEs (TEAEs) and all irAEs are reported in **Table 4** and **Table 5**, respectively

Table 3. Safety Summary by Dose, Patients Experiencing Event, n (%)

CX-072 Dose Level, mg/kg	≤1 n = 21	3 n = 13	10 n = 9	30 n = 3	All Dose Cohorts N = 46
TEAE					
All grades	21 (100)	13 (100)	8 (89)	3 (100)	45 (98)
Grade 3-4	13 (62)	8 (62)	5 (56)	2 (67)	28 (61)
SAE	6 (29)	7 (54)	3 (33)	0	16 (35)
TRAE					
All grades	9 (43)	8 (62)	5 (56)	3 (100)	25 (54)
Grade 3-4	2 (10)	2 (15)	0	1 (33)	5 (11)
SAE	1 (5)	2 (15)	0	0	3 (7)
irAE					
All grades	0	2 (15)	0	1 (33)	3 (7)
Grade 3-4	0	2 (15)	0	1 (33)	3 (7)
IRR					
All grades	5 (24)	3 (23)	1 (11)	1 (33)	10 (22)
Grade 3-4	2 (10)	0	0	0	2 (4)
TEAE leading to death					
All grades	0	0	0	0	0

AE, adverse event; irAE, immune-related adverse event; IRR, infusion-related reaction; SAE, serious adverse event; TEAE, treatment-emergent adverse event; TRAE, treatment-related adverse event.
Deaths due to disease progression (not considered related to study drug) were excluded from all safety analyses.
AEs were coded using MedDRA v19.1.

Table 4. Most Common TEAEs in ≥10% of Patients, Patients Experiencing Event, n (%)

CX-072 Dose Level, mg/kg	≤1 n = 21		3 n = 13		10 n = 9		30 n = 3		All Dose Cohorts N = 46		
Grade	1-2	3-4	1-2	3-4	1-2	3-4	1-2	3-4	1-2	3-4	Total
Fatigue	8 (38)	0	2 (15)	1 (8)	4 (44)	0	0	0	14 (30)	1 (2)	15 (33)
Cough	4 (19)	0	4 (31)	0	2 (22)	0	1 (33)	0	11 (24)	0	11 (24)
Anemia	2 (10)	1 (5)	2 (15)	2 (15)	1 (11)	0	1 (33)	1 (33)	6 (13)	4 (9)	10 (22)
Decreased appetite	6 (29)	0	2 (15)	1 (8)	1 (11)	0	0	0	9 (20)	1 (2)	10 (22)
IRR	3 (14)	2 (10)	3 (23)	0	1 (11)	0	1 (33)	0	8 (17)	2 (4)	10 (22)
Nausea	3 (14)	0	4 (31)	0	3 (33)	0	0	0	10 (22)	0	10 (22)
Constipation	6 (29)	0	2 (15)	0	1 (11)	0	0	0	9 (20)	0	9 (20)
Dyspnea	1 (5)	1 (5)	3 (23)	1 (8)	2 (22)	0	0	0	6 (13)	2 (4)	8 (17)
Back pain	0	1 (5)	2 (15)	1 (8)	2 (22)	0	1 (33)	0	5 (11)	2 (4)	7 (15)
Arthralgia	2 (10)	0	1 (8)	1 (8)	1 (11)	0	1 (33)	0	5 (11)	1 (2)	6 (13)
Diarrhea	0	0	3 (23)	0	2 (22)	0	1 (33)	0	6 (13)	0	6 (13)
Asthenia	2 (10)	0	3 (23)	0	0	0	0	0	5 (11)	0	5 (11)
Dizziness	3 (14)	0	1 (8)	0	1 (11)	0	0	0	5 (11)	0	5 (11)
Headache	2 (10)	0	1 (8)	0	2 (22)	0	0	0	5 (11)	0	5 (11)
Pain in extremity	2 (10)	0	2 (15)	0	0	1 (11)	0	0	4 (9)	1 (2)	5 (11)
Pyrexia	3 (14)	0	1 (8)	0	1 (11)	0	0	0	5 (11)	0	5 (11)
Vomiting	2 (10)	0	2 (15)	0	1 (11)	0	0	0	5 (11)	0	5 (11)

AE, adverse event; IRR, infusion-related reaction; TEAE, treatment-emergent adverse event.
Patients were grouped according to the most severe grade experienced for a particular AE. AEs were coded according to MedDRA v19.1.

Table 5. irAEs, Patients Experiencing Event, n (%)

CX-072 Dose Level, mg/kg	≤1 n = 21		3 n = 13		10 n = 9		30 n = 3		All Dose Cohorts N = 46		
Grade	1-2	3-4	1-2	3-4	1-2	3-4	1-2	3-4	1-2	3-4	Total
Any irAE	0	0	0	2 (15)	0	0	0	1 (33)	0	3 (7)	3 (7)
Liver	0	0	0	0	0	0	0	1 (33)	0	1 (2)	1 (2)
ALT increased[a]	0	0	0	0	0	0	0	1 (33)	0	1 (2)	1 (2)
AST increased[a]	0	0	0	0	0	0	0	1 (33)	0	1 (2)	1 (2)
Lung	0	0	0	1 (8)	0	0	0	0	0	1 (2)	1 (2)
Pneumonitis	0	0	0	1 (8)	0	0	0	0	0	1 (2)	1 (2)
Other	0	0	0	1 (8)	0	0	0	0	0	1 (2)	1 (2)
Febrile neutropenia[b]	0	0	0	1 (8)	0	0	0	0	0	1 (2)	1 (2)
Thrombocytopenia[b]	0	0	0	1 (8)	0	0	0	0	0	1 (2)	1 (2)

AE, adverse event; ALT, alanine aminotransferase; AST, aspartate aminotransferase; irAE, immune-related adverse event.
[a]ALT and AST elevation occurred in the same patient.
[b]Febrile neutropenia and thrombocytopenia occurred in the same patient.
irAEs were defined as treatment-related AEs that were on a predefined list of >300 preferred terms and that required treatment with systemic steroids within 30 days of the onset of the relevant AE.
Patients were grouped according to the most severe grade experienced for a particular AE. AEs were coded using MedDRA v19.1.

Tumor Response

- Tumor response rates among evaluable patients (n = 38) who had ≥1 postbaseline tumor assessment are provided in **Table 6**
 - The disease control rate (complete response, partial response [PR], or stable disease) was 18 of 38 (47%) of all evaluable patients; in patients receiving CX-072 ≥3 mg/kg, the disease control rate was 11 of 18 (61%)
- Target lesions decreased from baseline in 14 of 37 (38%) evaluable patients with measurable disease at baseline (**Figure 3**)
 - Target lesions decreased from baseline at dose levels ≥3 mg/kg in 10 of 17 patients (59%) with measurable disease at baseline
- 3 patients achieved PR as best response:
 - Confirmed PR: Triple-negative breast cancer (10 mg/kg): PD-L1 negative, low tumor mutational burden, microsatellite-stable (**Figure 4**)
 - Unconfirmed PRs: Thymic cancer (3 mg/kg) and cervical cancer (10 mg/kg)

Table 6. Best Tumor Response in Evaluable Patients[a] per RECIST v1.1, n (%)

CX-072 Dose, mg/kg	≤1 n = 20	3 n = 10	10 n = 5	30 n = 3	All Evaluable Patients n = 38
Partial response[b]	0	1 (10)	2 (40)	0	3 (8)
Stable disease[c]	7 (35)	5 (50)	1 (20)	2 (67)	15 (39)
Disease control rate[d]	7 (35)	6 (60)	3 (60)	2 (67)	18 (47)
Progressive disease	13 (65)	3 (30)	2 (40)	0	18 (47)
Inevaluable[e]	0	1 (10)	0	1 (33)	2 (5)

RECIST, Response Evaluation Criteria in Solid Tumors.
[a]Evaluable patients are those with ≥1 postbaseline tumor assessment.
[b]Includes 2 patients with unconfirmed partial response.
[c]One patient with nonmeasurable disease at baseline in the 3 mg/kg cohort had a best overall response of noncomplete response/nonprogressive disease and was grouped under stable disease.
[d]Disease control rate = partial response + stable disease.
[e]Two patients each had a single postbaseline assessment that was stable disease but was assessed earlier than the protocol-defined minimum duration on study drug (7 weeks).

Figure 3. Best percentage change from baseline in target lesions.[a]

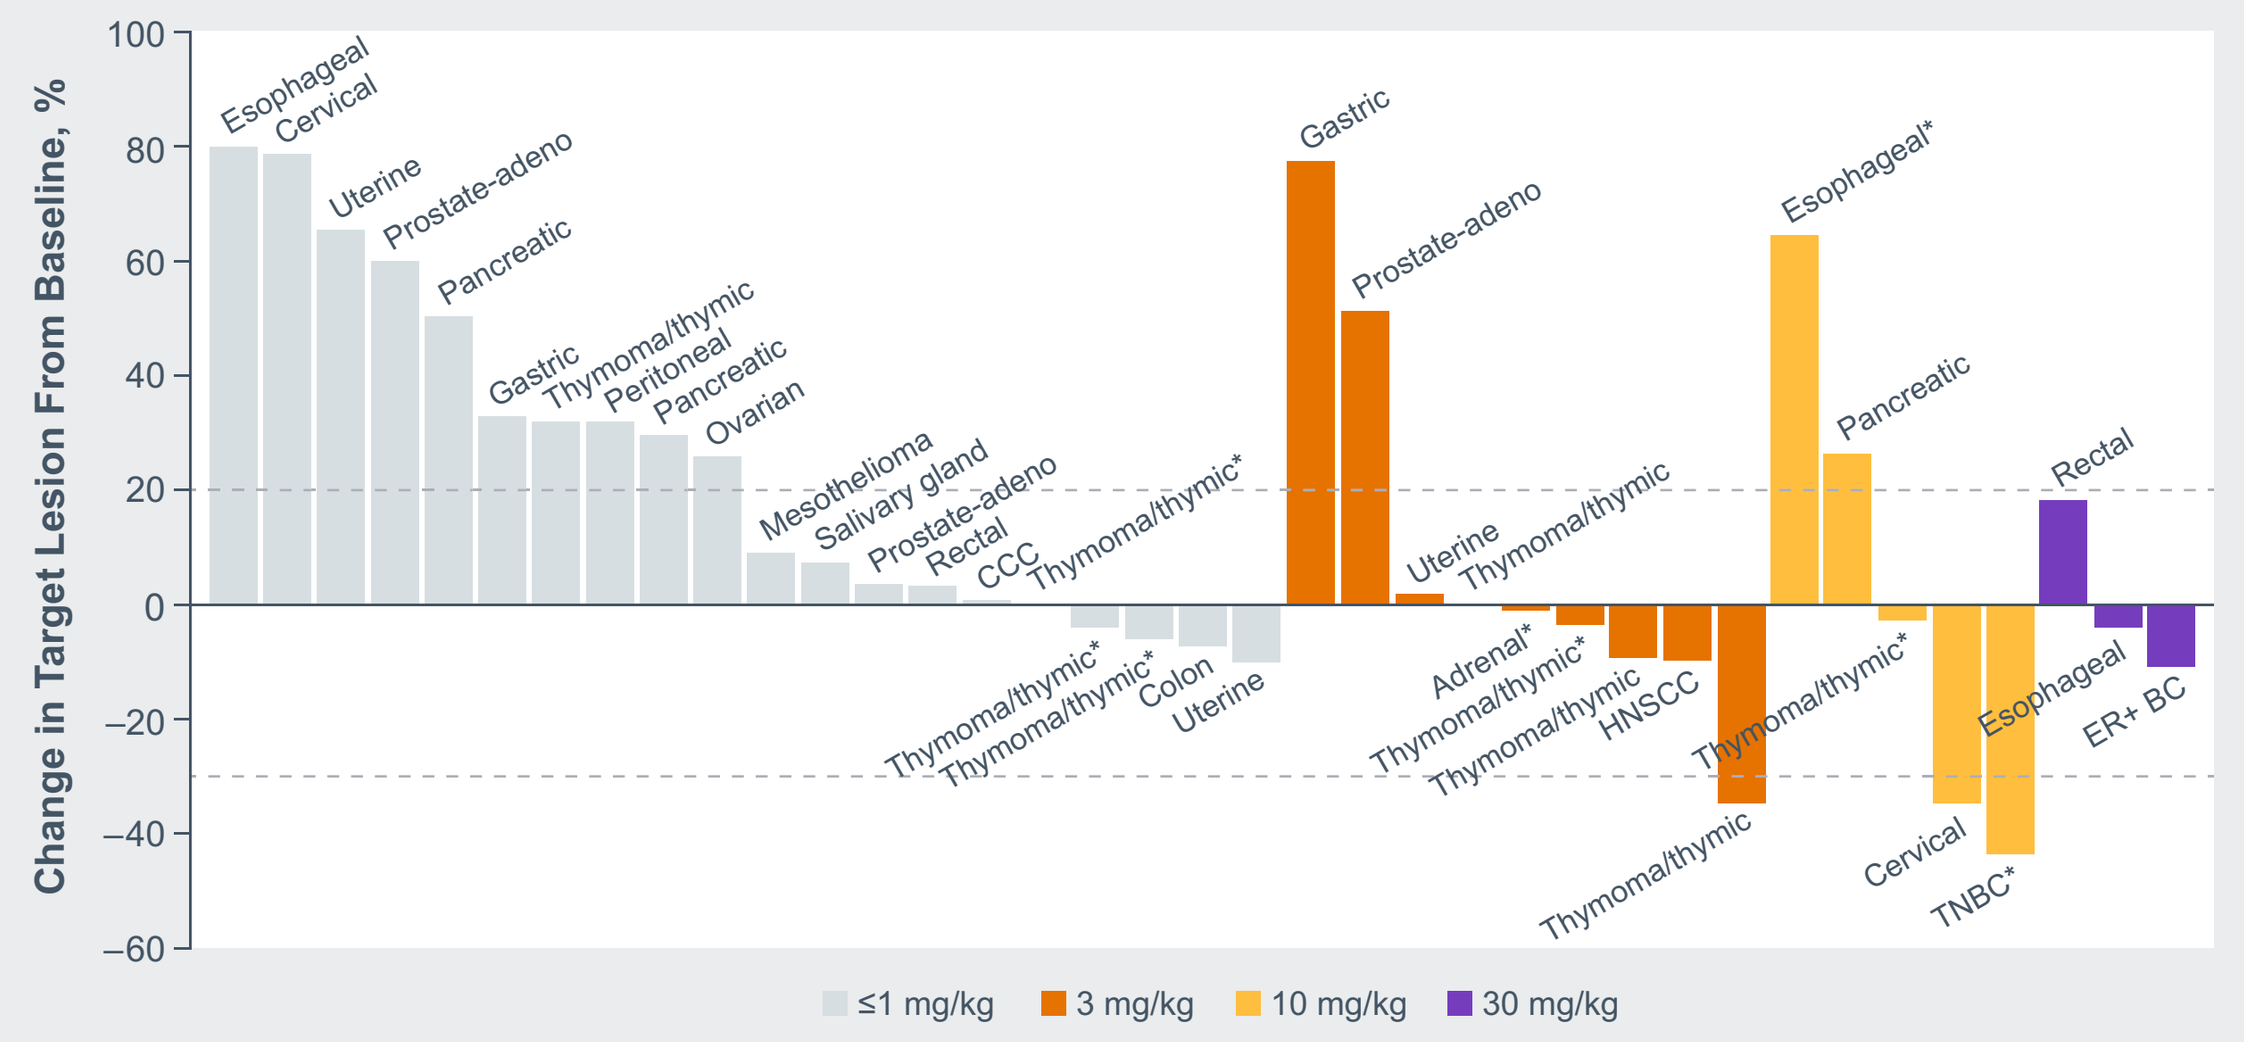
Change in Target Lesion From Baseline, %
Esophageal
Cervical
Uterine
Prostate-adeno
Pancreatic
Gastric
Thymoma/thymic
Peritoneal
Pancreatic
Ovarian
Mesothelioma
Salivary gland
Prostate-adeno
Rectal
CCC
Thymoma/thymic*
Thymoma/thymic*
Thymoma/thymic*
Colon
Uterine
Gastric
Prostate-adeno
Uterine
Thymoma/thymic
Adrenal*
Thymoma/thymic*
Thymoma/thymic
HNSCC
Thymoma/thymic
Esophageal*
Pancreatic
Thymoma/thymic*
Cervical
TNBC*
Esophageal
Rectal
ER+ BC
≤1 mg/kg
3 mg/kg
10 mg/kg
30 mg/kg

CCC, cholangiocellular carcinoma; ER+ BC, breast (ER+) carcinoma; HNSCC, head and neck squamous cell carcinoma; RECIST, Response Evaluation Criteria in Solid Tumors; TNBC, triple-negative breast cancer.
*Patient is still receiving treatment as of data cutoff.
[a]As evaluated per RECIST v1.1. Plots include evaluable patients with measurable disease at baseline.

Sample Case Study

Figure 4. Patient with triple-negative breast cancer treated with CX-072 10 mg/kg.

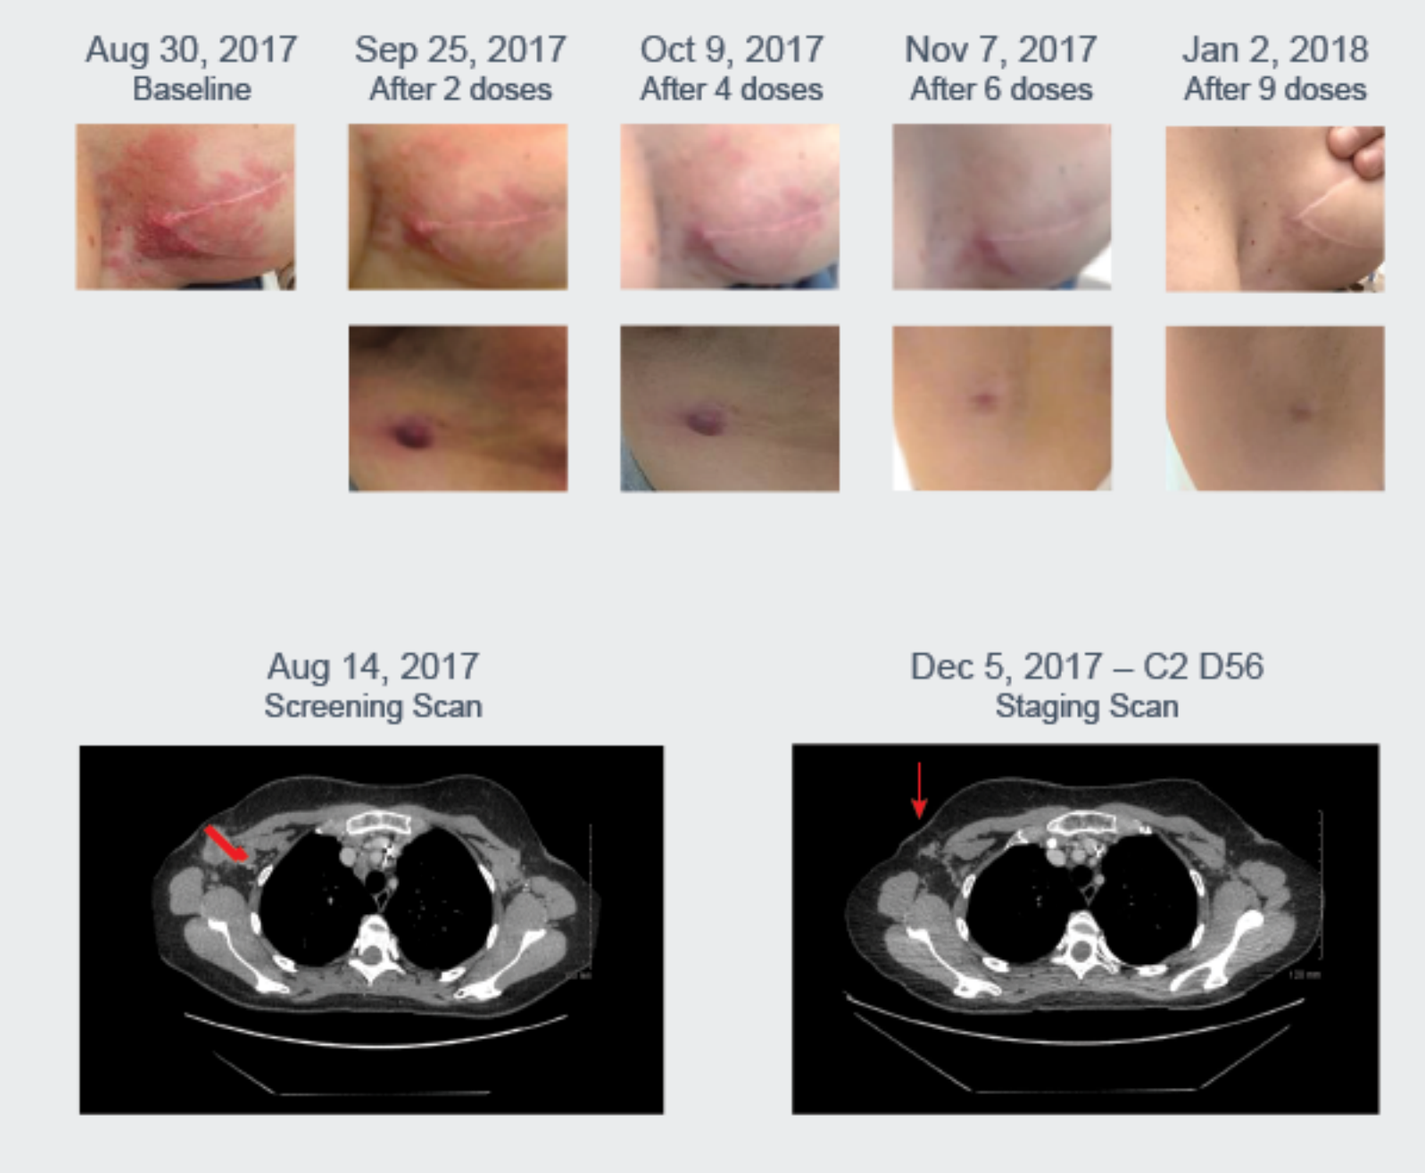
Aug 30, 2017
Baseline
Sep 25, 2017
After 2 doses
Oct 9, 2017
After 4 doses
Nov 7, 2017
After 6 doses
Jan 2, 2018
After 9 doses
Aug 14, 2017
Screening Scan
Dec 5, 2017 – C2 D56
Staging Scan

Patient

- Triple-negative breast cancer
 - Microsatellite-stable
 - Low tumor mutational burden (4 mutations/megabase)
 - PD-L1 negative
- Continues to receive CX-072 10 mg/kg as of data cutoff
- Patient has confirmed partial response

CONCLUSIONS

- The MTD has not been reached
- Probody CX-072 monotherapy demonstrated durable anticancer activity with a favorable safety profile in a patient population unlikely to respond to checkpoint inhibitors
- Of all patients treated at ≥3 mg/kg who were evaluable and had ≥1 postbaseline assessment (n = 18)
 - 3 patients (17%) experienced an objective response (1 PR and 2 unconfirmed PRs) among a variety of mostly PD-L1–negative cancer types that are not routinely treated with immunotherapy or not commonly thought to be immunogenic
 - 11 patients (61%) experienced disease control
- Of the safety population (N = 46)
 - To date, no differences in toxicity were observed across dose levels
 - 5 patients (11%) experienced a grade 3-4 TRAE
 - 3 patients (7%) experienced an irAE
- Expansion cohorts in 8 indications are in progress at the 10 mg/kg dose level in patients with known PD-1 pathway–sensitive cancer types and/or cancer types that demonstrated Probody CX-072 sensitivity in the current study

REFERENCES

1. Iwai Y et al. *J Biomed Sci.* 2017;24:26.
2. Baxi et al. *BMJ.* 2018;360:k793.
3. Pawel J et al. *Ann Oncol.* 2017;28(suppl 5).
4. Herbst RS et al. *Nature.* 2014;515:563-567.
5. Desnoyers LR et al. *Sci Transl Med.* 2013;5:207ra144.
6. Polu KR et al. *Expert Opin Biol Ther.* 2014;14:1049-1053.
7. Autio KA et al. *J Clin Oncol.* 2018;36(suppl); abstr 3071.

ACKNOWLEDGMENTS

The authors thank the participating patients and their families and all staff at participating sites. This study was sponsored by CytomX Therapeutics, Inc. Medical writing assistance was provided by Andrew Occiano, PharmD (ApotheCom, San Francisco, CA, USA) and was funded by CytomX Therapeutics, Inc.

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

PROBODY is a trademark of CytomX Therapeutics, Inc. All other brands and trademarks referenced herein are the property of their respective owners.

View Other Publications

Presented at the European Society for Medical Oncology (ESMO) Congress; October 19–23, 2018; Munich, Germany

© 2018 CytomX Therapeutics, Inc.