CX-072, a PD-L1 Probody Therapeutic, as Monotherapy in Patients With Advanced Solid Tumors: Preliminary Results of PROCLAIM-CX-072

Aung Naing,[1] Fiona Thistlethwaite,[2] Alexander Spira,[3] Javier Garcia-Corbacho,[4] Manreet Randhawa,[5] Ferry ALM Eskens,[6] Greg Durm,[7] Nataliya Uboha,[8] Omid Hamid,[9] Anthony El-Khoueiry,[10] Vanessa Huels,[11] Will Garner,[11] Beverly Benson,[11] Hendrik-Tobias Arkenau,[12] Patricia LoRusso[13]

[1]MD Anderson Cancer Center, Houston, TX; [2]The Christie NHS Foundation Trust and University of Manchester, Manchester, UK; [3]Virginia Cancer Specialists, Fairfax, VA; [4]Hospital Clinic Barcelona/IDIBAPS, Barcelona, Spain; [5]Beatson West of Scotland Cancer Centre, Glasgow, UK; [6]Erasmus MC Cancer Center, Rotterdam, Netherlands; [7]Indiana University, Melvin and Bren Simon Cancer Center, Indianapolis, IN; [8]University of Wisconsin, Carbone Cancer Center, Madison, WI; [9]The Angeles Clinic and Research Institute, Los Angeles, CA; [10]USC Norris Comprehensive Cancer Center, Los Angeles, CA; [11]CytomX Therapeutics, Inc., South San Francisco, CA; [12]Sarah Cannon Research Institute UK Limited, London, UK; [13]Yale University School of Medicine, Yale Cancer Center, New Haven, CT

Introduction

- The programmed cell death 1 (PD-1) signaling pathway plays an important role in dampening immune surveillance and the development of malignancies; therapies that target this pathway, such as anti-PD-1 and anti-programmed cell death ligand-1 (PD-L1) monoclonal antibodies, have become part of standard therapy for multiple tumor types based upon results of large randomized trials with proven progression-free and/or overall survival benefit[1]
- Anti-cancer activity for the PD-pathway inhibitors varies depending on the cancer type and stage of disease, with single-agent overall response rates of 5% in previously treated advanced or metastatic triple-negative breast cancer (TNBC) unselected for PD-L1 status (N=58),[2] 24% in previously untreated PD-L1-postive TNBC (N=21),[3] 5% in patients with metastatic sarcoma unselected for PD-L1 status (N=42),[4] and 17% in patients with recurrent PD-L1-positive squamous cell carcinoma of the anal canal (N=24)[5]
- The optimal use of PD-pathway inhibitors is limited by potentially life-threatening toxicities, particularly when used in combination
 - Nivolumab monotherapy was associated with a rate of grade ≥3 treatment-related adverse events (TRAEs) of 16%, a rate of discontinuation for TRAEs of 8%, and a rate of discontinuation due to grade ≥3 TRAEs of 5% in patients with melanoma (N=313)[6]
 - Furthermore, when nivolumab was combined with ipilimumab in the same study (N=313), the corresponding rates were 55%, 36%, and 29%, respectively, underscoring the need for potentially safer alternatives
- Probody™ therapeutics are recombinant antibody prodrugs that are designed to activate in the presence of proteases in the tumor microenvironment and to preferentially bind to the tumor rather than healthy tissue (**Figure 1**)[7,8]

Figure 1. Probody Structure

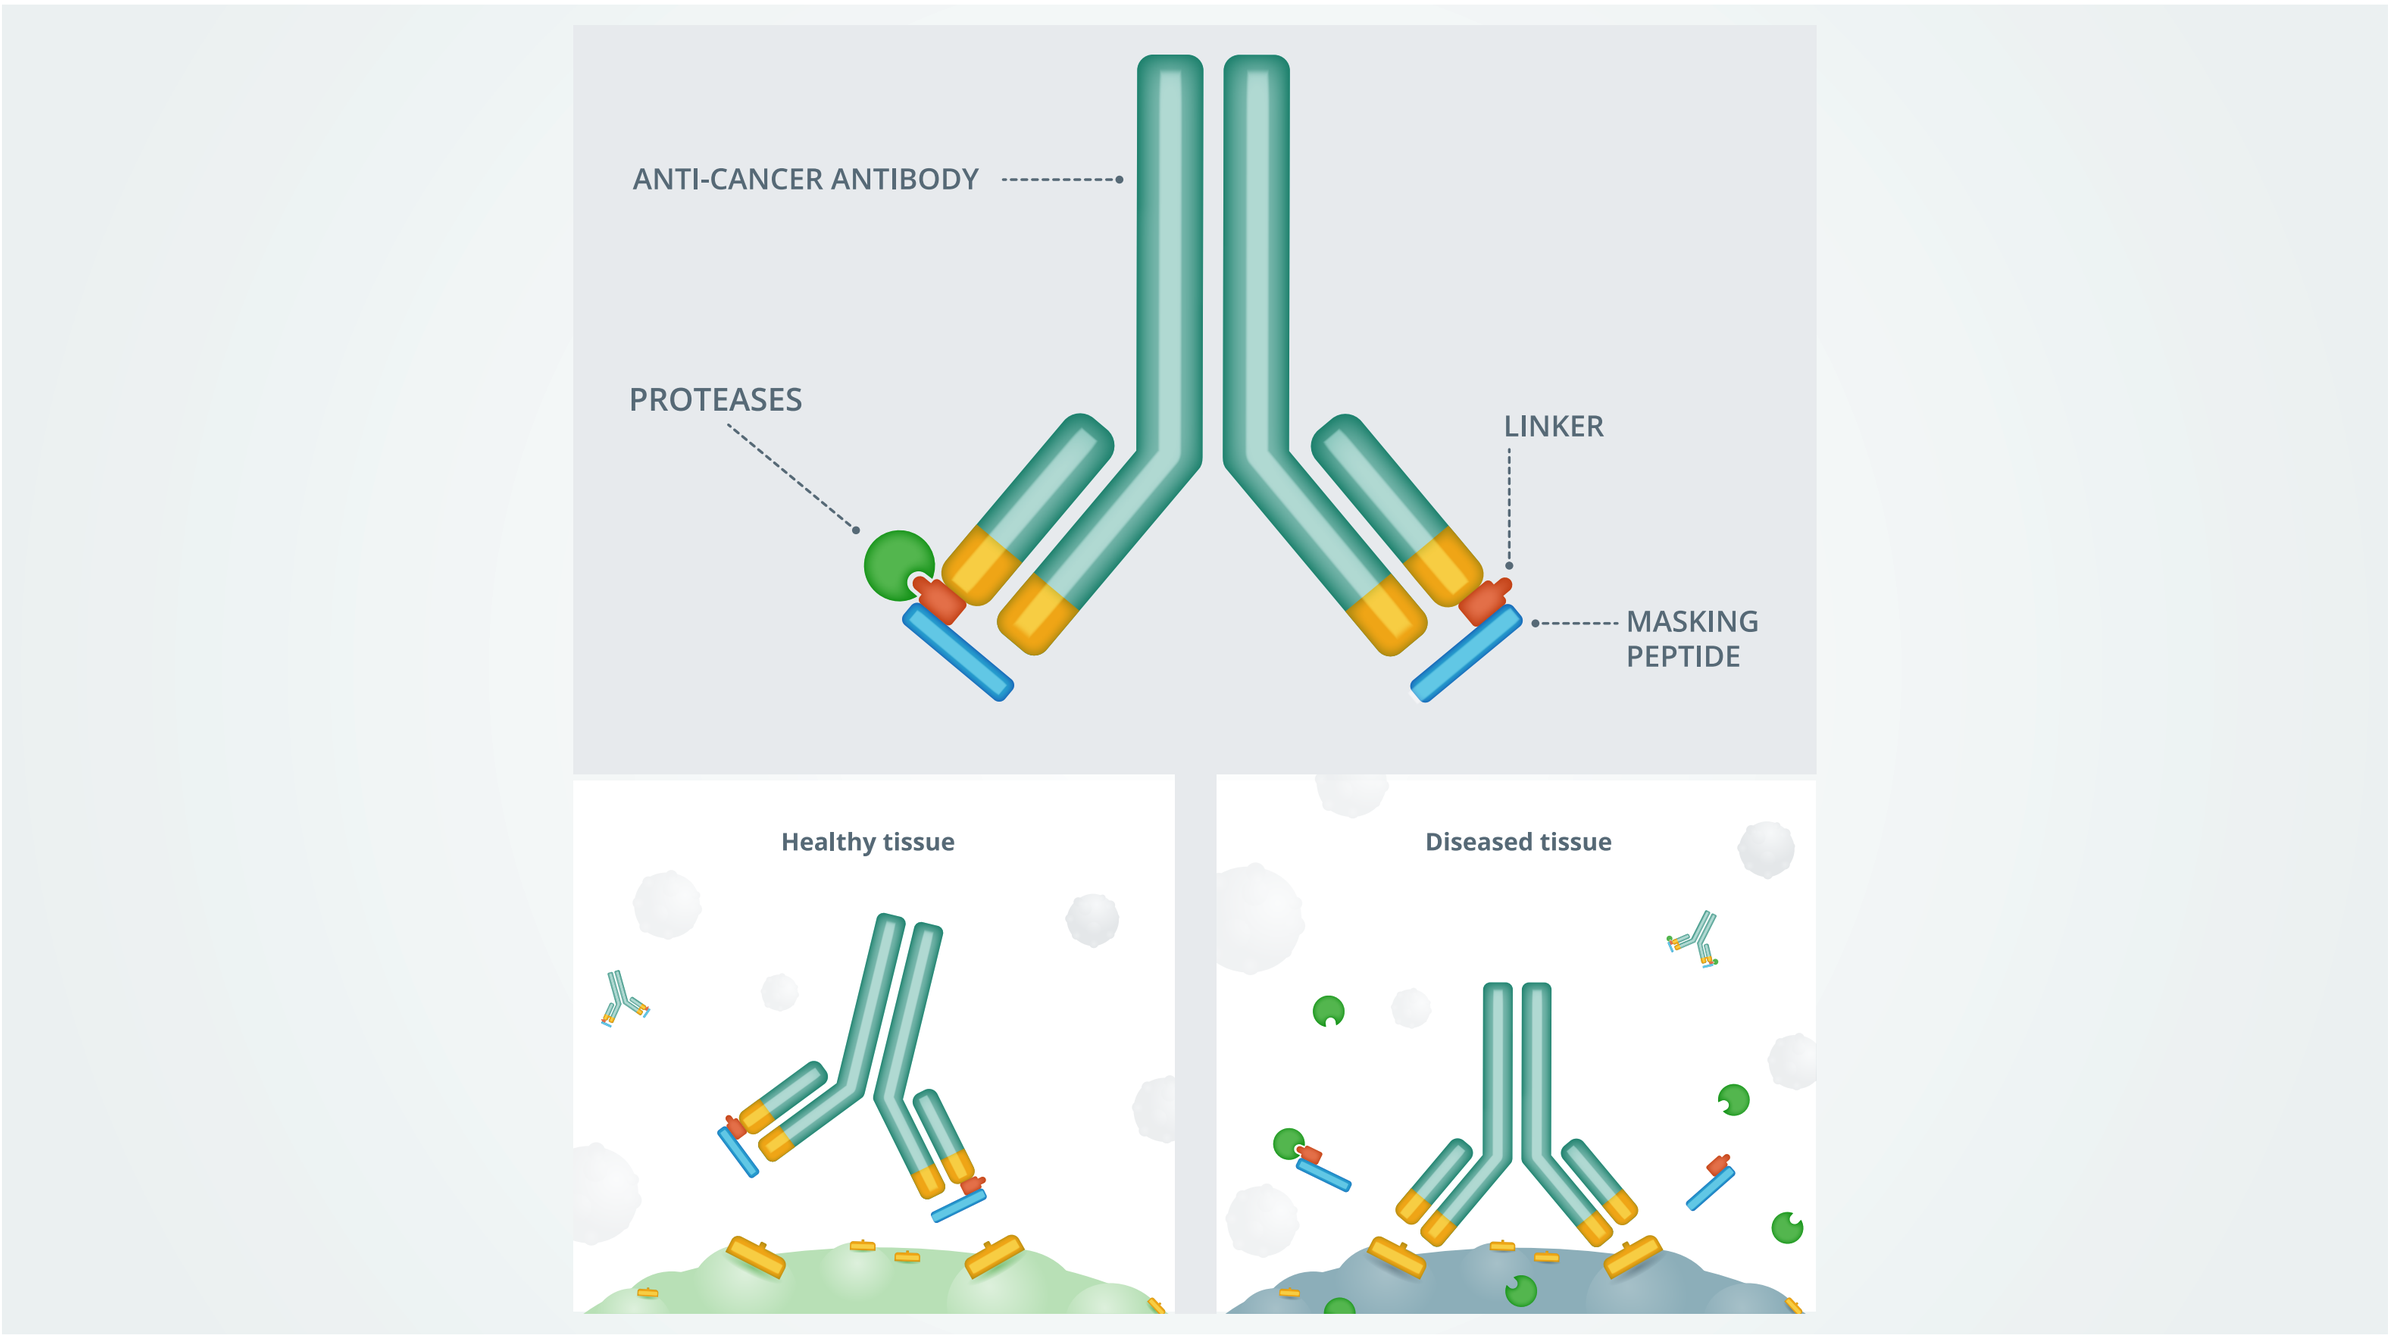
ANTI-CANCER ANTIBODY
PROTEASES
LINKER
MASKING PEPTIDE
Healthy tissue
Diseased tissue

- In an ongoing phase 1 clinical program, CX-072, an investigational anti-PD-L1 Probody therapeutic, has been shown to perform as designed:
 - Circulating predominantly in the intact form[9]
 - Activating in tumor tissues at high levels (with >98% receptor coverage at doses of ≥3 mg/kg)[9]
 - Having a level of anticancer activity expected from the class of PD-pathway inhibitors,[10] and
 - Having a favorable toxicity profile, particularly in combination with ipilimumab[10,11]
- PROCLAIM-CX-072 (PRObody CLinical Assessment In Man) is an ongoing phase 1/2a study evaluating the tolerability and preliminary antitumor activity of multiple doses and the expansion of Probody CX-072 as monotherapy and various other regimens, including combination therapy with ipilimumab (ClinicalTrials.gov identifier NCT03013491)

Objective

- To report preliminary results of antitumor activity of CX-072 monotherapy at its recommended dose of 10 mg/kg every 2 weeks in patients with TNBC with skin lesions, anal squamous cell carcinoma (SCC), cutaneous squamous cell carcinoma (cSCC), undifferentiated pleomorphic sarcoma (UPS), or small bowel adenocarcinoma (SBA)

Methods

Study Design and Treatment

- This first-in-human, open-label, dose-escalation, multicenter, phase 1/2a study of CX-072 was designed to evaluate the safety and determine the maximum tolerated dose and/or maximum achieved dose of CX-072 as monotherapy, in combination with ipilimumab, and in combination with vemurafenib; here we report preliminary results from selected cohorts in the monotherapy dose-expansion phase
- Patients received CX-072 monotherapy 10 mg/kg intravenously every 14 days until confirmed disease progression or unacceptable toxicity

Patients

- Eligible patients were ≥18 years of age with Eastern Cooperative Oncology Group performance status 0 or 1 and adequate hematologic, hepatic, and renal function
- Patients were required to be immunotherapy naïve, including checkpoint inhibitors
- Patients were not selected for PD-L1 expression
- Eligible patients had selected metastatic or locally advanced tumor types with measurable disease and tumor progression despite standard treatment (or no standard treatment available, ineligible for standard treatment, or unwilling to undergo standard treatment)
 - TNBC patients were required to have locally advanced and locally recurrent skin or subcutaneous metastases not suitable for surgical resection or radiotherapy and to have received ≥1 and ≤3 systemic chemotherapy regimens for metastatic breast cancer with documented disease progression
 - Anal SCC patients were required to have metastatic or locally advanced unresectable disease and prior radiation therapy and/or chemotherapy
 - cSCC patients were required to have metastatic or locally advanced unresectable primary disease
 - UPS patients were required to have tumor proportion score (TPS) ≥1% membranous staining or unknown PD-L1 status, prior standard surgery or radiation therapy, and ≥1 prior systemic therapy
 - SBA patients were required to have ≥1 and ≤3 prior lines of systemic chemotherapy for metastatic or advanced unresectable disease

Safety Assessments

- Adverse events (AEs) were assessed and reviewed before each infusion and at any other visit that included a physical examination. AEs were coded using MedDRA v21.1 and graded using the National Cancer Institute CTCAE v4.03
 - Immune-related AEs (irAEs) were defined as TRAEs from a list of >300 terms relating to inflammation that required treatment with immunosuppressive agents, including but not limited to systemic steroids within 30 days of the onset of the relevant AE with no clear alternate etiology, or requiring use of systemic hormonal replacement

Tumor Response Assessments

- Imaging for tumor response assessment using Response Evaluation Criteria in Solid Tumors (RECIST) v1.1 was performed every 8 weeks for the first 12 months, then every 12 weeks thereafter

RESULTS

- As of the April 5, 2019 data cut-off date, the median duration of follow-up in the 5 selected cohorts was 10.1 weeks (range: 2.1–76 weeks)
- A total of 72 patients were enrolled into the selected cohorts and treated, with a mean age of 58.5 years (range: 32–80); most were female (67%) and white (71%) (**Table 1**)
- Patients had received a median of 3 prior treatment regimens (range 1–10)
- 33% of patients had any PD-L1 expression (ie, high [tumor proportion score >50%] or low [tumor proportion score ≥1%–50%] expression) by the central assay on archival tissue using the DAKO PD-L1 IHC 22c3 assay

Table 1. Patient Demographic and Baseline Characteristics

Characteristic	TNBC (n=15)	Anal SCC (n=15)	cSCC (n=8)	UPS (n=20)	SBA (n=14)	Total* (n=72)
Mean age (range), years	53.5 (32–72)	61.0 (36–80)	69.6 (61–79)	59.8 (33–80)	53.0 (34–67)	58.5 (32–80)
Male, n (%)	0	2 (13)	2 (25)	12 (60)	8 (57)	24 (33)
Female, n (%)	15 (100)	13 (87)	6 (75)	8 (40)	6 (43)	48 (67)
Race, n (%)						
White	7 (47)	11 (73)	8 (100)	16 (80)	9 (64)	51 (71)
Black/African American	2 (13)	0	0	2 (10)	2 (14)	6 (8)
Asian	0	0	0	1 (5)	0	1 (1)
Unknown/not reported	6 (40)	4 (27)	0	1 (5)	3 (21)	14 (19)
Median prior regimens (range), n	3 (1–10)	2 (1–5)	1.5 (1–4)	3 (1–6)	2.5 (1–6)	3 (1–10)
Baseline ECOG PS, n (%)						
0	6 (40)	9 (60)	4 (50)	1 (5)	6 (43)	26 (36)
1	8 (53)	6 (40)	4 (50)	19 (95)	8 (57)	45 (63)
Missing	1 (7)	0	0	0	0	1 (1)
PD-L1 status at baseline, n (%)						
High expression	0	1 (7)	0	4 (20)	0	5 (7)
Low expression	1 (7)	7 (47)	3 (38)	6 (30)	2 (14)	19 (26)
No expression	11 (73)	0	1 (13)	9 (45)	8 (57)	29 (40)
Unknown	3 (20)	7 (47)	4 (50)	1 (5)	4 (29)	19 (26)

TNBC=triple-negative breast cancer with skin lesions; SCC=squamous cell carcinoma; cSCC=cutaneous squamous cell carcinoma; UPS=undifferentiated pleomorphic sarcoma; SBA=small bowel adenocarcinoma; ECOG PS=Eastern Cooperative Oncology Group Performance Status; PD-L1=programmed cell death ligand-1.
*Total refers to the total of the 5 selected indications. All patients are from dose expansion, except for 2 patients who met the tumor indications and were dosed at 10 mg/kg in the dose-escalation part (TNBC [n=1] and anal SCC [n=1]).

Preliminary Antitumor Response

- Antitumor activity was assessed in 65 response-evaluable patients, which included patients in the safety population but excluded those who are ongoing with no post-baseline response assessment at data cut-off
- A summary of best overall responses is shown in **Table 2**
- Confirmed partial responses were observed in TNBC (n=2), cSCC (n=1), and UPS (n=1); partial response that was unconfirmed at data cut-off was subsequently confirmed in one patient with anal SCC
- Disease control rates (defined as complete response + partial response + stable disease) were 53% (8/15) in TNBC, 58% (7/12) in anal SCC, 67% (4/6) in cSCC, 25% (5/20) in UPS, and 17% (2/12) in SBA

Table 2. Summary of Best Overall Responses by Tumor Type in All Patients Treated*

Best Overall Response, n (%)	TNBC (n=15)	Anal SCC (n=12)	cSCC (n=6)	UPS (n=20)	SBA (n=12)
ORR	2 (13)	1 (8)‡	1 (17)	1 (5)	0
PR	2 (13)	1 (8)‡	1 (17)	1 (5)	0
SD	6 (40)	6 (50)	3 (50)	4 (20)	2 (17)
PD	4 (27)	4 (33)	2 (33)	11 (55)	9 (75)
Early withdrawal§	3 (20)	1 (8)	0	4 (20)	1 (8)
Disease control rate (CR+PR+SD)	8 (53)	7 (58)	4 (67)	5 (25)	2 (17)

TNBC=triple-negative breast cancer with skin lesions; SCC=squamous cell carcinoma; cSCC=cutaneous squamous cell carcinoma; UPS=undifferentiated pleomorphic sarcoma; SBA=small bowel adenocarcinoma; ORR=overall response rate; PR=partial response; SD=stable disease; PD=progressive disease; CR=complete response.
*Excluding newly enrolled patients who were on treatment and had not provided a postbaseline assessment as of data cut-off; all patients are from dose expansion, except for 2 patients who met the tumor indications and were dosed at 10 mg/kg in the dose-escalation part (TNBC [n=1] and anal SCC [n=1]).
‡At data cutoff, the patient had an unconfirmed partial response that was subsequently confirmed.
§Discontinued study without providing a post-baseline scan.

- Decreases in target lesion size were observed in patients with TNBC, anal SCC, cSCC, and UPS, as shown in waterfall plots by tumor type in **Figure 2** (top panel)
- In patients who responded, decreases in tumor size from baseline were observed in the first 8 to 16 weeks (**Figure 2**, bottom panel)

Figure 2. Percent Change in Target Lesions From Baseline (top panel) and Percent Change in Tumor Burden Over Time (bottom panel) by Tumor Type

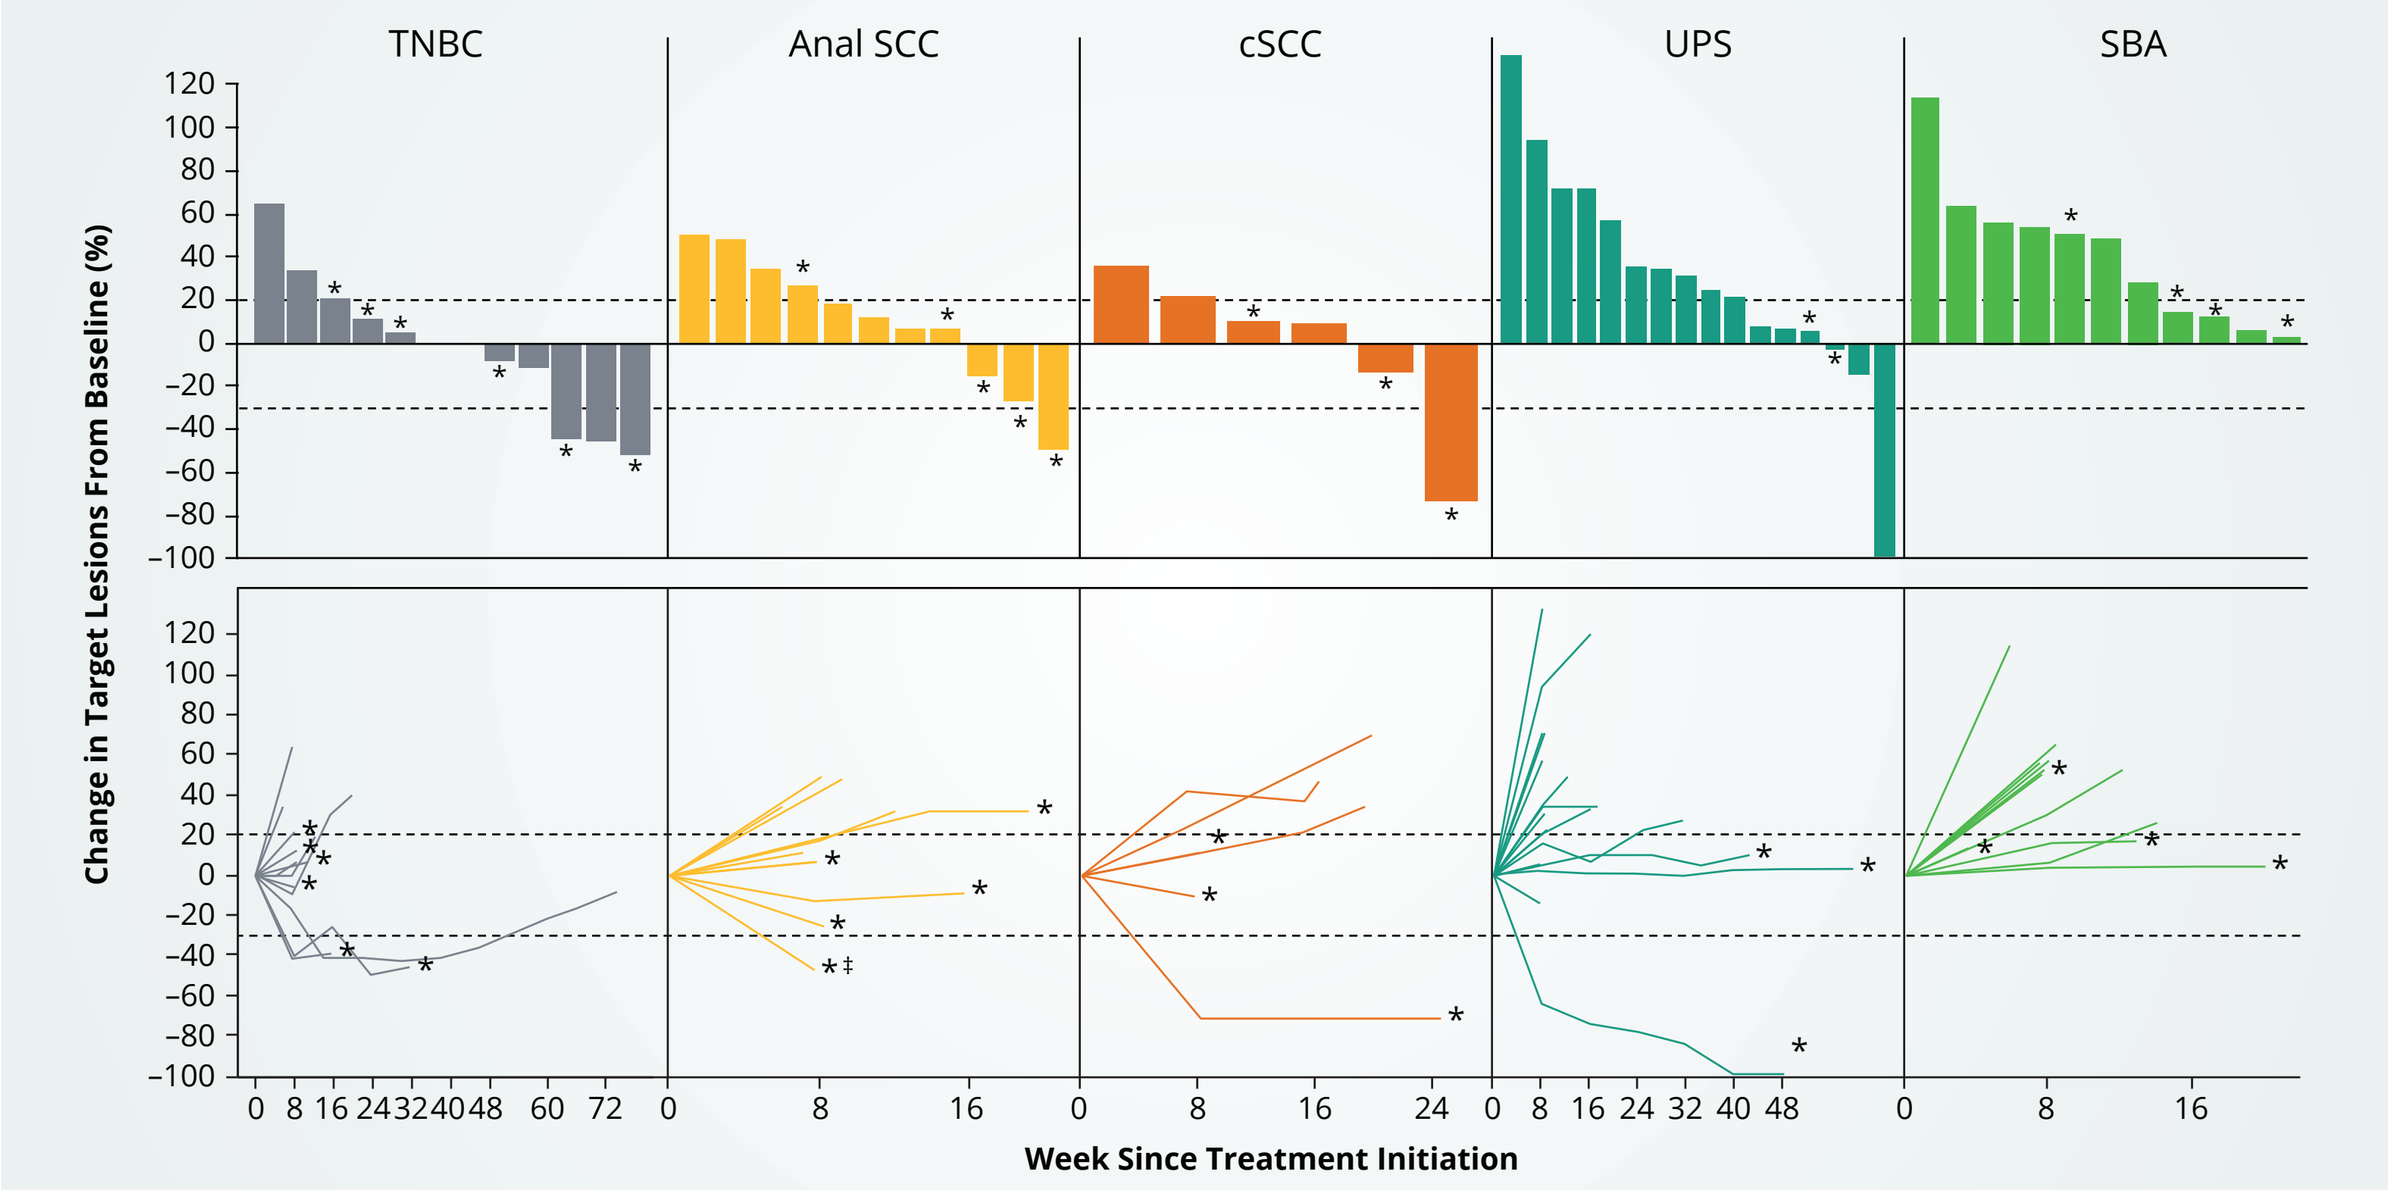
TNBC
Anal SCC
cSCC
UPS
SBA
Change in Target Lesions From Baseline (%)
Week Since Treatment Initiation

TNBC=triple-negative breast cancer with skin lesions; SCC=squamous cell carcinoma; cSCC=cutaneous squamous cell carcinoma; UPS=undifferentiated pleomorphic sarcoma; SBA=small bowel adenocarcinoma.
*Denotes patient considered to be on treatment, as no end-of-treatment date was listed in database as of data cut-off (April 5, 2019). Evaluable patients include those in the safety population except patients who are ongoing with no post-baseline response assessment as of data cut-off.
‡At data cutoff, the patient had an unconfirmed partial response that was subsequently confirmed.
All patients are from dose expansion, except for 2 patients who met the tumor indications and were dosed at 10 mg/kg in the dose-escalation part (TNBC [n=1] and anal SCC [n=1]).

- Among patients who responded per RECIST v1.1 criteria, most remain on treatment as of data cut-off (**Figure 3**)

Figure 3. Time on Treatment for Patients With RECIST v1.1 Response

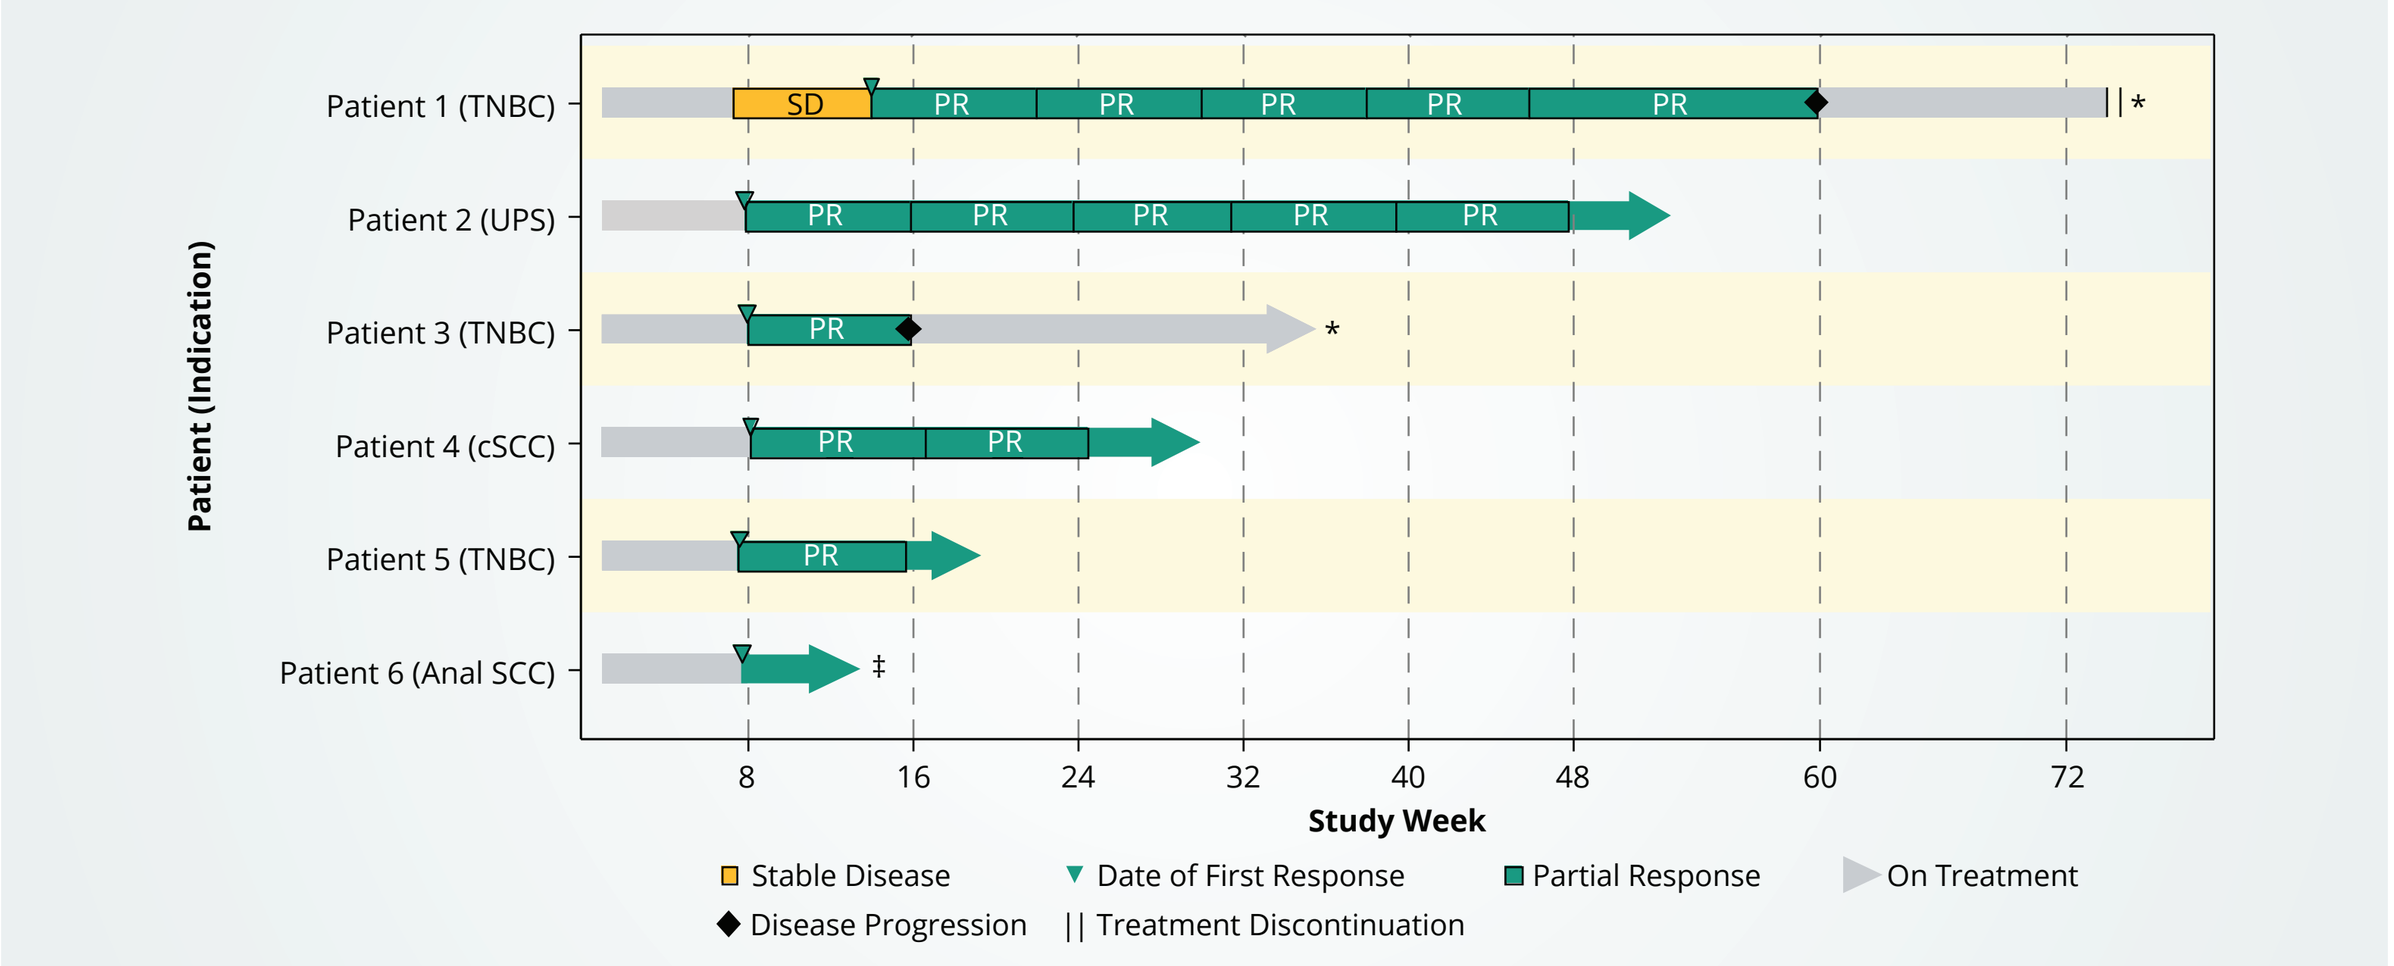
Patient (Indication)
Patient 1 (TNBC)
Patient 2 (UPS)
Patient 3 (TNBC)
Patient 4 (cSCC)
Patient 5 (TNBC)
Patient 6 (Anal SCC)
SD
PR
Study Week
Stable Disease
Date of First Response
Partial Response
On Treatment
Disease Progression
Treatment Discontinuation

TNBC=triple-negative breast cancer with skin lesions; SCC=squamous cell carcinoma; cSCC=cutaneous squamous cell carcinoma; UPS=undifferentiated pleomorphic sarcoma; SD=stable disease; PR=partial response.
*Patients with partial response according to RECIST v1.1 could continue beyond initial disease progression. Patient 1 (TNBC) was enrolled in the dose-escalation cohort. Patient 3 (TNBC) had a drug interruption which resulted in a progressive disease response assessment at the second postbaseline visit. The best overall response for the subject is stable disease. Subsequent postbaseline visits showed reductions >30% relative to baseline.
‡At data cutoff, the patient had an unconfirmed partial response that was subsequently confirmed.
Grey bars denote time on study drug. All patients are from dose expansion except for Patient 1 (TNBC) who met the tumor indications and was dosed at 10 mg/kg in the dose-escalation part.

Safety

- CX-072 10 mg/kg monotherapy resulted in rates of grade ≥3 TRAEs of 6%, grade ≥3 irAEs of 3%, and treatment-related serious AEs of 3%, with no grade ≥3 infusion-related reactions and no TRAEs leading to treatment discontinuation or death (**Table 3**)

Table 3. Overview of Treatment-Emergent Adverse Events

n (%)	TNBC (n=15)	Anal SCC (n=15)	cSCC (n=8)	UPS (n=20)	SBA (n=14)	Total* (n=72)
Any TEAE	13 (87)	12 (80)	6 (75)	19 (95)	13 (93)	63 (88)
CX-072-related	9 (60)	6 (40)	3 (38)	4 (20)	5 (36)	27 (38)
TEAE grade ≥3	8 (53)	8 (53)	1 (13)	8 (40)	10 (71)	35 (49)
CX-072-related	2 (13)	1 (7)	0	0	1 (7)	4 (6)
Infusion-related reactions	1 (7)	2 (13)	1 (13)	0	0	4 (6)
Grade ≥3	0	0	0	0	0	0
TEAE leading to discontinuation	0	1 (7)	0	0	1 (7)	2 (3)
CX-072-related	0	0	0	0	0	0
Serious TEAE	4 (27)	5 (33)	1 (13)	6 (30)	6 (43)	22 (31)
CX-072-related	1 (7)	1 (7)	0	0	0	2 (3)
TEAE leading to death	1 (7)	0	0	0	0	1 (1)
CX-072-related	0	0	0	0	0	0
irAEs	4 (27)	2 (13)	1 (13)	2 (10)	1 (7)	10 (14)
Grade ≥3	2 (13)	0	0	0	0	2 (3)

TNBC=triple-negative breast cancer with skin lesions; SCC=squamous cell carcinoma; cSCC=cutaneous squamous cell carcinoma; UPS=undifferentiated pleomorphic sarcoma; SBA=small bowel adenocarcinoma; TEAE=treatment-emergent adverse event; irAE=immune-related adverse event.
*Total refers to the total of the 5 selected indications. All patients are from dose expansion, except for 2 patients who met the tumor indications and were dosed at 10 mg/kg in the dose-escalation part (TNBC [n=1] and anal SCC [n=1]).

- **Table 4** summarizes treatment-emergent adverse events (TEAEs) observed in ≥10% of patients, TRAEs observed in ≥5% of patients, and serious AEs observed in at least 2 patients
- The most common TEAEs included fatigue (25%), diarrhea (22%), increased aspartate aminotransferase (21%), and nausea (21%) (**Table 4**)
- The most common TRAEs included increased aspartate aminotransferase (13%), increased alanine aminotransferase (8%), and fatigue (6%) (**Table 4**)

Table 4. Treatment-Emergent Adverse Events (≥10% of Patients), Treatment-Related Adverse Events (≥5% of Patients), and Serious Adverse Events (≥2 Patients)

n (%)	TNBC (n=15)	Anal SCC (n=15)	cSCC (n=8)	UPS (n=20)	SBA (n=14)	Total* (n=72)
TEAEs (≥10% of patients)						
Fatigue	3 (20)	4 (27)	0	4 (20)	7 (50)	18 (25)
Diarrhea	3 (20)	1 (7)	2 (25)	6 (30)	4 (29)	16 (22)
AST increased	4 (27)	3 (20)	1 (13)	3 (15)	4 (29)	15 (21)
Nausea	6 (40)	2 (13)	3 (38)	1 (5)	3 (21)	15 (21)
Anemia	0	4 (27)	0	8 (40)	2 (14)	14 (19)
Dyspnea	2 (13)	3 (20)	0	5 (25)	2 (14)	12 (17)
Decreased appetite	1 (7)	1 (7)	1 (13)	4 (20)	4 (29)	11 (15)
ALT increased	1 (7)	1 (7)	1 (13)	3 (15)	3 (21)	9 (13)
Vomiting	2 (13)	1 (7)	2 (25)	2 (10)	2 (14)	9 (13)
Headache	3 (20)	1 (7)	2 (25)	2 (10)	0	8 (11)
TRAEs (≥5% of patients)						
AST increased	2 (13)	2 (13)	1 (13)	2 (10)	2 (14)	9 (13)
ALT increased	1 (7)	0	1 (13)	2 (10)	2 (14)	6 (8)
Fatigue	1 (7)	1 (7)	0	0	2 (14)	4 (6)
SAEs (≥2 patients)						
Arthralgia	1 (7)	0	1 (13)	1 (5)	0	3 (4)
Fatigue	0	0	0	1 (5)	1 (7)	2 (3)
Intestinal obstruction	0	0	0	0	2 (14)	2 (3)
Pericardial effusion	2 (13)	0	0	0	0	2 (3)
Pleural effusion	1 (7)	0	0	1 (5)	0	2 (3)
Pneumonia	1 (7)	0	0	1 (5)	0	2 (3)
Pyrexia	0	1 (7)	0	0	1 (7)	2 (3)
Tumor hemorrhage	0	1 (7)	0	1 (5)	0	2 (3)

TNBC=triple-negative breast cancer with skin lesions; SCC=squamous cell carcinoma; cSCC=cutaneous squamous cell carcinoma; UPS=undifferentiated pleomorphic sarcoma; SBA=small bowel adenocarcinoma; TEAE=treatment-emergent adverse event; AST=aspartate aminotransferase; ALT=alanine aminotransferase; TRAE=treatment-related adverse event; SAE= serious adverse event.
*Total refers to the total of the 5 selected indications. All patients are from dose expansion, except for 2 patients who met the tumor indications and were dosed at 10 mg/kg in the dose-escalation part (TNBC [n=1] and anal SCC [n=1]).

- The incidence of grade 3/4 irAEs was 3%; 2 patients with TNBC experienced increased gamma-glutamyl transferase (n=1) and maculopapular rash (n=1; **Table 5**)

Table 5. Grade 1/2 Versus Grade 3/4 Immune-Related Adverse Events

	TNBC (n=15)		Anal SCC (n=15)		cSCC (n=8)		UPS (n=20)		SBA (n=14)		Total* (n=72)	
n (%)	Gr 1/2	Gr 3/4	Gr 1/2	Gr 3/4	Gr 1/2	Gr 3/4	Gr 1/2	Gr 3/4	Gr 1/2	Gr 3/4	Gr 1/2	Gr 3/4
Any irAE	2 (13)	2 (13)	2 (13)	0	1 (13)	0	2 (10)	0	1 (7)	0	8 (11)	2 (3)
AST increased	1 (7)	0	0	0	1 (13)	0	0	0	1 (7)	0	3 (4)	0
ALT increased	0	0	0	0	1 (13)	0	0	0	1 (7)	0	2 (3)	0
GGT increased	0	1 (7)	0	0	0	0	0	0	0	0	0	1 (1)
Hypothyroidism	0	0	1 (7)	0	0	0	1 (5)	0	1 (7)	0	3 (4)	0
Thyroiditis acute	0	0	0	0	0	0	1 (5)	0	0	0	1 (1)	0
Maculopapular rash	1 (7)	1 (7)	0	0	0	0	0	0	0	0	1 (1)	1 (1)
Dyspnea	0	0	1 (7)	0	0	0	0	0	0	0	1 (1)	0
Arthralgia	1 (7)	0	0	0	0	0	0	0	0	0	1 (1)	0

TNBC=triple-negative breast cancer with skin lesions; SCC=squamous cell carcinoma; cSCC=cutaneous squamous cell carcinoma; UPS=undifferentiated pleomorphic sarcoma; SBA=small bowel adenocarcinoma; irAE=immune-related adverse event; AST=aspartate aminotransferase; ALT=alanine aminotransferase; GGT=gamma-glutamyl transferase.
*Total refers to the total of the 5 selected indications. All patients are from dose expansion, except for 2 patients who met the tumor indications and were dosed at 10 mg/kg in the dose-escalation part (TNBC [n=1] and anal SCC [n=1]).
Note: If a patient experienced more than one adverse event within a preferred term, the patient was counted once for that preferred term at the highest severity grade.

Conclusions

- **Preliminary results of PROCLAIM-CX-072 in select tumor types suggest that the CX-072 Probody platform is working as designed:**
 - Durable clinical activity as monotherapy was observed in heavily pretreated patients, with a pattern of efficacy consistent with that of the PD-pathway inhibitor class in the cancer types treated
 - Acceptable safety profile
 - **Low rates of grade ≥3 TRAEs**
 - **No discontinuation for TRAEs**
 - **Low incidence of grade ≥3 irAEs**
 - **Favorable in context of historical data with PD pathway inhibitors in a variety of cancer types, which showed rates of grade ≥3 TRAEs of approximately 16% and discontinuations due to TRAEs of up to 8%[6,12]**
- **Further clinical evaluations of CX-072 both as monotherapy and in combination with other anticancer agents, such as ipilimumab, are ongoing**

References

1. Iwai Y, et al. *J Biomed Sci.* 2017;24:26.
2. Dirix LY, et al. *Breast Cancer Res Treat.* 2018;167:671-86.
3. Emens LA, et al. *JAMA Oncol.* 2019;5:74-82.
4. D'Angelo SP, et al. *Lancet Oncol.* 2018;19:416-26.
5. Ott PA, et al. *Ann Oncol.* 2017;28:1036-41.
6. Larkin J, et al. *N Engl J Med.* 2015;373:23-34.
7. Desnoyers LR, et al. *Sci Transl Med.* 2013;5:207ra144.
8. Polu KR, Lowman HB. *Expert Opin Biol Ther.* 2014;14:1049-53.
9. Lyman S, et al. Presented at: Annual Meeting of the Society for Immunotherapy of Cancer November 7-11, 2018; Washington, D.C.
10. Boni V, et al. Presented at: Annual Meeting of the European Society for Medical Oncology; October 19-23, 2018; Munich, Germany.
11. Plummer R, et al. Presented at: Annual Meeting of the European Society for Medical Oncology; October 19-23, 2018; Munich, Germany.
12. Janjigian YY, et al. *J Clin Oncol.* 2018;36:2836-44.

Acknowledgments

Medical writing assistance was provided by Echelon Brand Communications, an OPEN Health company, Parsippany, NJ, and was funded by CytomX Therapeutics, Inc.

PROBODY is a trademark of CytomX Therapeutics, Inc. All other brands and trademarks referenced herein are the property of their respective owners.

Source of Funding: CytomX Therapeutics, Inc.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

Presented at the 55th Annual Meeting of the American Society of Clinical Oncology, May 31–June 4, 2019, Chicago, IL

© 2019 CytomX Therapeutics, Inc.