423TiP

The First-in-Human, Dose-Finding PROCLAIM-CX-072 Trial to Assess the Antitumor Activity and Tolerability of the Probody™ Therapeutic CX-072 as Monotherapy and in Combination With Ipilimumab or Vemurafenib in Solid Advanced Tumors and Lymphomas

Valentina Boni,[1] Javier Garcia-Corbacho,[2] Jaime Feliu,[3] Jerzy Wydmanski,[4] Zsolt Horvath,[5] Igor Bondarenko,[6] Bryan Irving,[7*] Matthias Will,[7] Elisabeth de Vries,[8] Fiona Thistlethwaite[9]

[1]START Madrid-CIOCC, Madrid, Spain; [2]Hospital Clinic Barcelona, Barcelona, Spain; [3]Hospital Universitario La Paz, Madrid, Spain; [4]NZOZ Vegamed, Katowice, Poland; [5]Debreceni Egyetem Klinikai Kozpont, Debrecen, Hungary; [6]Dnipropetrovsk State Medical Academy, Dnepropetrovsk, Ukraine; [7]CytomX Therapeutics, South San Francisco, CA, USA; [8]University Medical Center Groningen, Groningen, Netherlands; [9]The Christie NHS Foundation Trust and University of Manchester, Manchester, UK

*Affiliation at the time of study design

BACKGROUND

- Tumors can evade host immunity by expression of programmed cell death ligand 1 (PD-L1), a ligand that negatively regulates programmed cell death 1 (PD-1), an inhibitory receptor expressed on activated T cells[1]
- Antibodies targeting PD-L1 have shown activity against a variety of cancers and are being tested in combination with other immunotherapies to improve response rates and durability of response[2]
- However, significant life-threatening immune-related adverse events (irAEs) are known toxicities of PD-1/PD-L1–axis blocking antibodies, especially when used in a wide variety of combinations such as with ipilimumab,[3,4] vemurafenib, pazopanib, or osimertinib[5,6]
- Probody therapeutics are fully recombinant antibody prodrugs that remain relatively inactive systemically and in healthy tissue, thereby avoiding binding to target antigen in healthy tissue. Once a Probody therapeutic reaches the tumor, it is activated by proteases associated with that microenvironment and can freely bind target antigen in the tumor (**Figure 1A**).[7,8] In preclinical studies, Probody therapeutics have demonstrated efficacy similar to that of parental antibodies and safety profiles better than those of parental antibodies (**Figure 2**)[9]
- CX-072 is a Probody therapeutic directed against PD-L1 for the treatment of patients with cancer
 - Given that CX-072 is activated by tumor-associated proteases, it is expected to be relatively inactive in peripheral tissue, thereby potentially reducing systemic irAEs compared with other PD-1/PD-L1 inhibitors
 - In situ studies of tumor samples from 200 patients with a variety of malignancies confirm Probody therapeutic activation in >90% of the samples, which corroborates the presence of tumor microenvironment proteases in the overwhelming majority of tumors necessary to ensure activation of the Probody therapeutic
 - Preclinical studies using a surrogate for CX-072 demonstrated that protease activation restores binding of CX-072 to PD-L1 in vitro (**Figure 1B**); in mouse models, the surrogate demonstrated equivalent antitumor efficacy, reduced induction of autoimmune diabetes, and reduced peripheral binding to circulating T cells compared with the parental antibody[9]

Figure 1. Probody therapeutics are protease-activatable antibody prodrugs. (A) Schematic representation of Probody therapeutic activation in the tumor microenvironment. (B) CX-072 exhibits protease-dependent binding to PD-L1 in vitro based on ELISA.

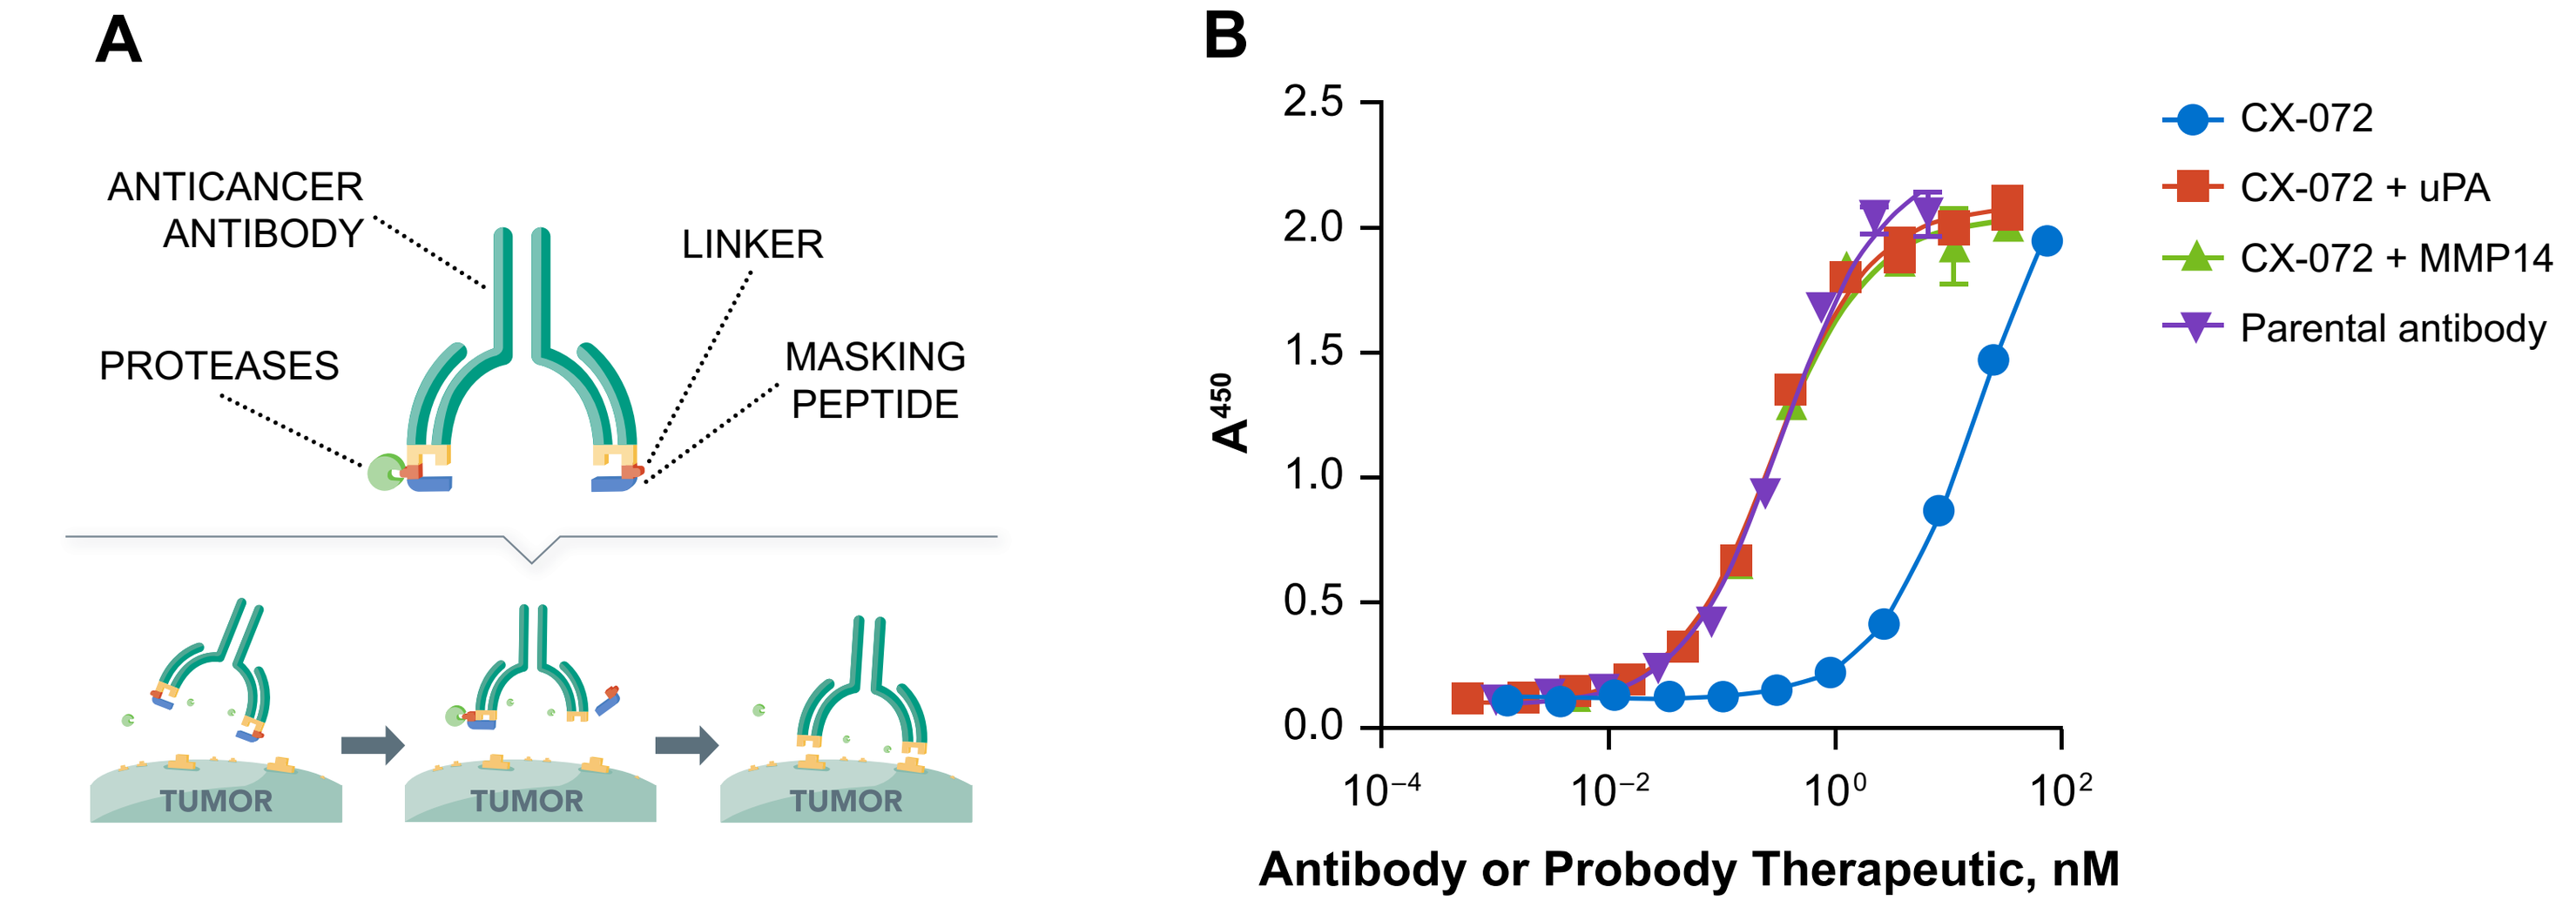

A
ANTICANCER ANTIBODY
LINKER
PROTEASES
MASKING PEPTIDE
TUMOR
TUMOR
TUMOR
B
CX-072
CX-072 + uPA
CX-072 + MMP14
Parental antibody
A_{450}
2.5
2.0
1.5
1.0
0.5
0.0
10^{-4}
10^{-2}
10^{0}
10^{2}
Antibody or Probody Therapeutic, nM

ELISA, enzyme-linked immunosorbent assay; MMP14, matrix metalloproteinase 14; PD-L1, programmed cell death ligand 1; uPA, urokinase-type plasminogen activator. CX-072 (blue) shows reduced binding to recombinant PD-L1, whereas proteolytic activation of CX-072 with uPA or MMP14 proteases (red, green) restores binding to levels comparable to those of the parental antibody (purple).

Figure 2. In preclinical studies, Probody therapeutics display efficacy similar to that of parental antibodies (comparable reduction in tumor growth) with improved safety (reduced autoimmunity).

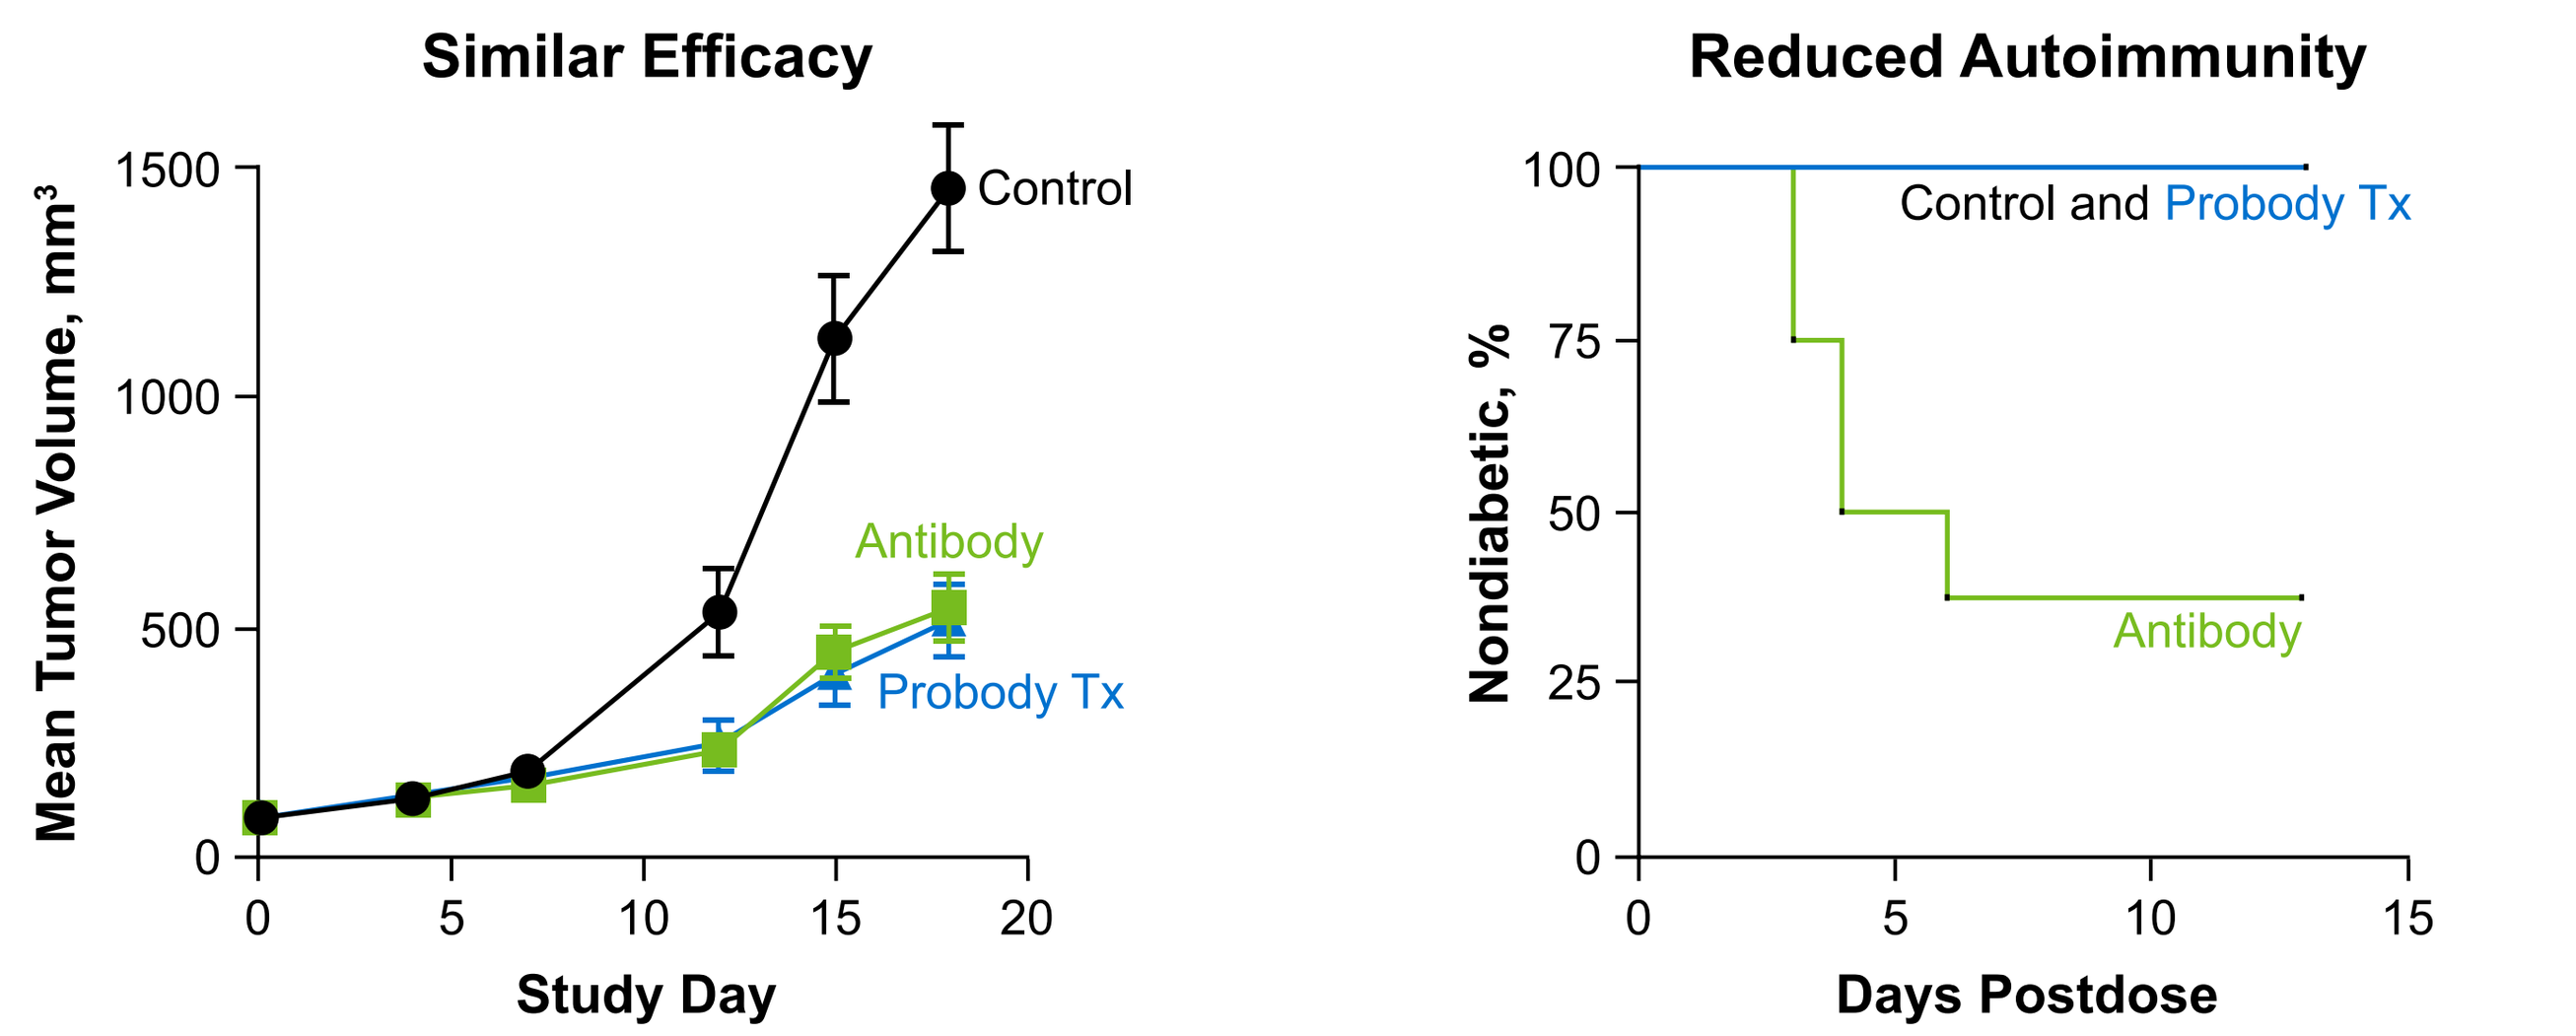

Similar Efficacy
Mean Tumor Volume, mm³
1500
1000
500
0
Control
Antibody
Probody Tx
0
5
10
15
20
Study Day
Reduced Autoimmunity
Nondiabetic, %
100
75
50
25
0
Control and Probody Tx
Antibody
0
5
10
15
Days Postdose

Tx, therapeutic.

OBJECTIVE

- The PROCLAIM-CX-072 (**PRO**body **CL**inical **A**ssessment **I**n **M**an) study is evaluating tolerability and preliminary antitumor activity of multiple doses of CX-072 as monotherapy or as combination therapy with ipilimumab or vemurafenib in patients with advanced, unresectable solid tumors or lymphoma

STUDY DESIGN

- This first-in-human, open-label, multicenter, dose-escalation, phase 1/2 study of CX-072 includes 4 dose-escalation groups (monotherapy, Part A; 2 ipilimumab combination schedules, Parts B1 and B2; vemurafenib combination, Part C), a stage testing biomarkers and efficacy in PD-L1+ tumors (Part A2), and a dose-expansion phase (Part D) (**Figure 3**)
- Treatment will continue for up to 2 years or until disease progression is confirmed or toxicity becomes unacceptable

Figure 3. PROCLAIM-CX-072 phase 1/2 study design.

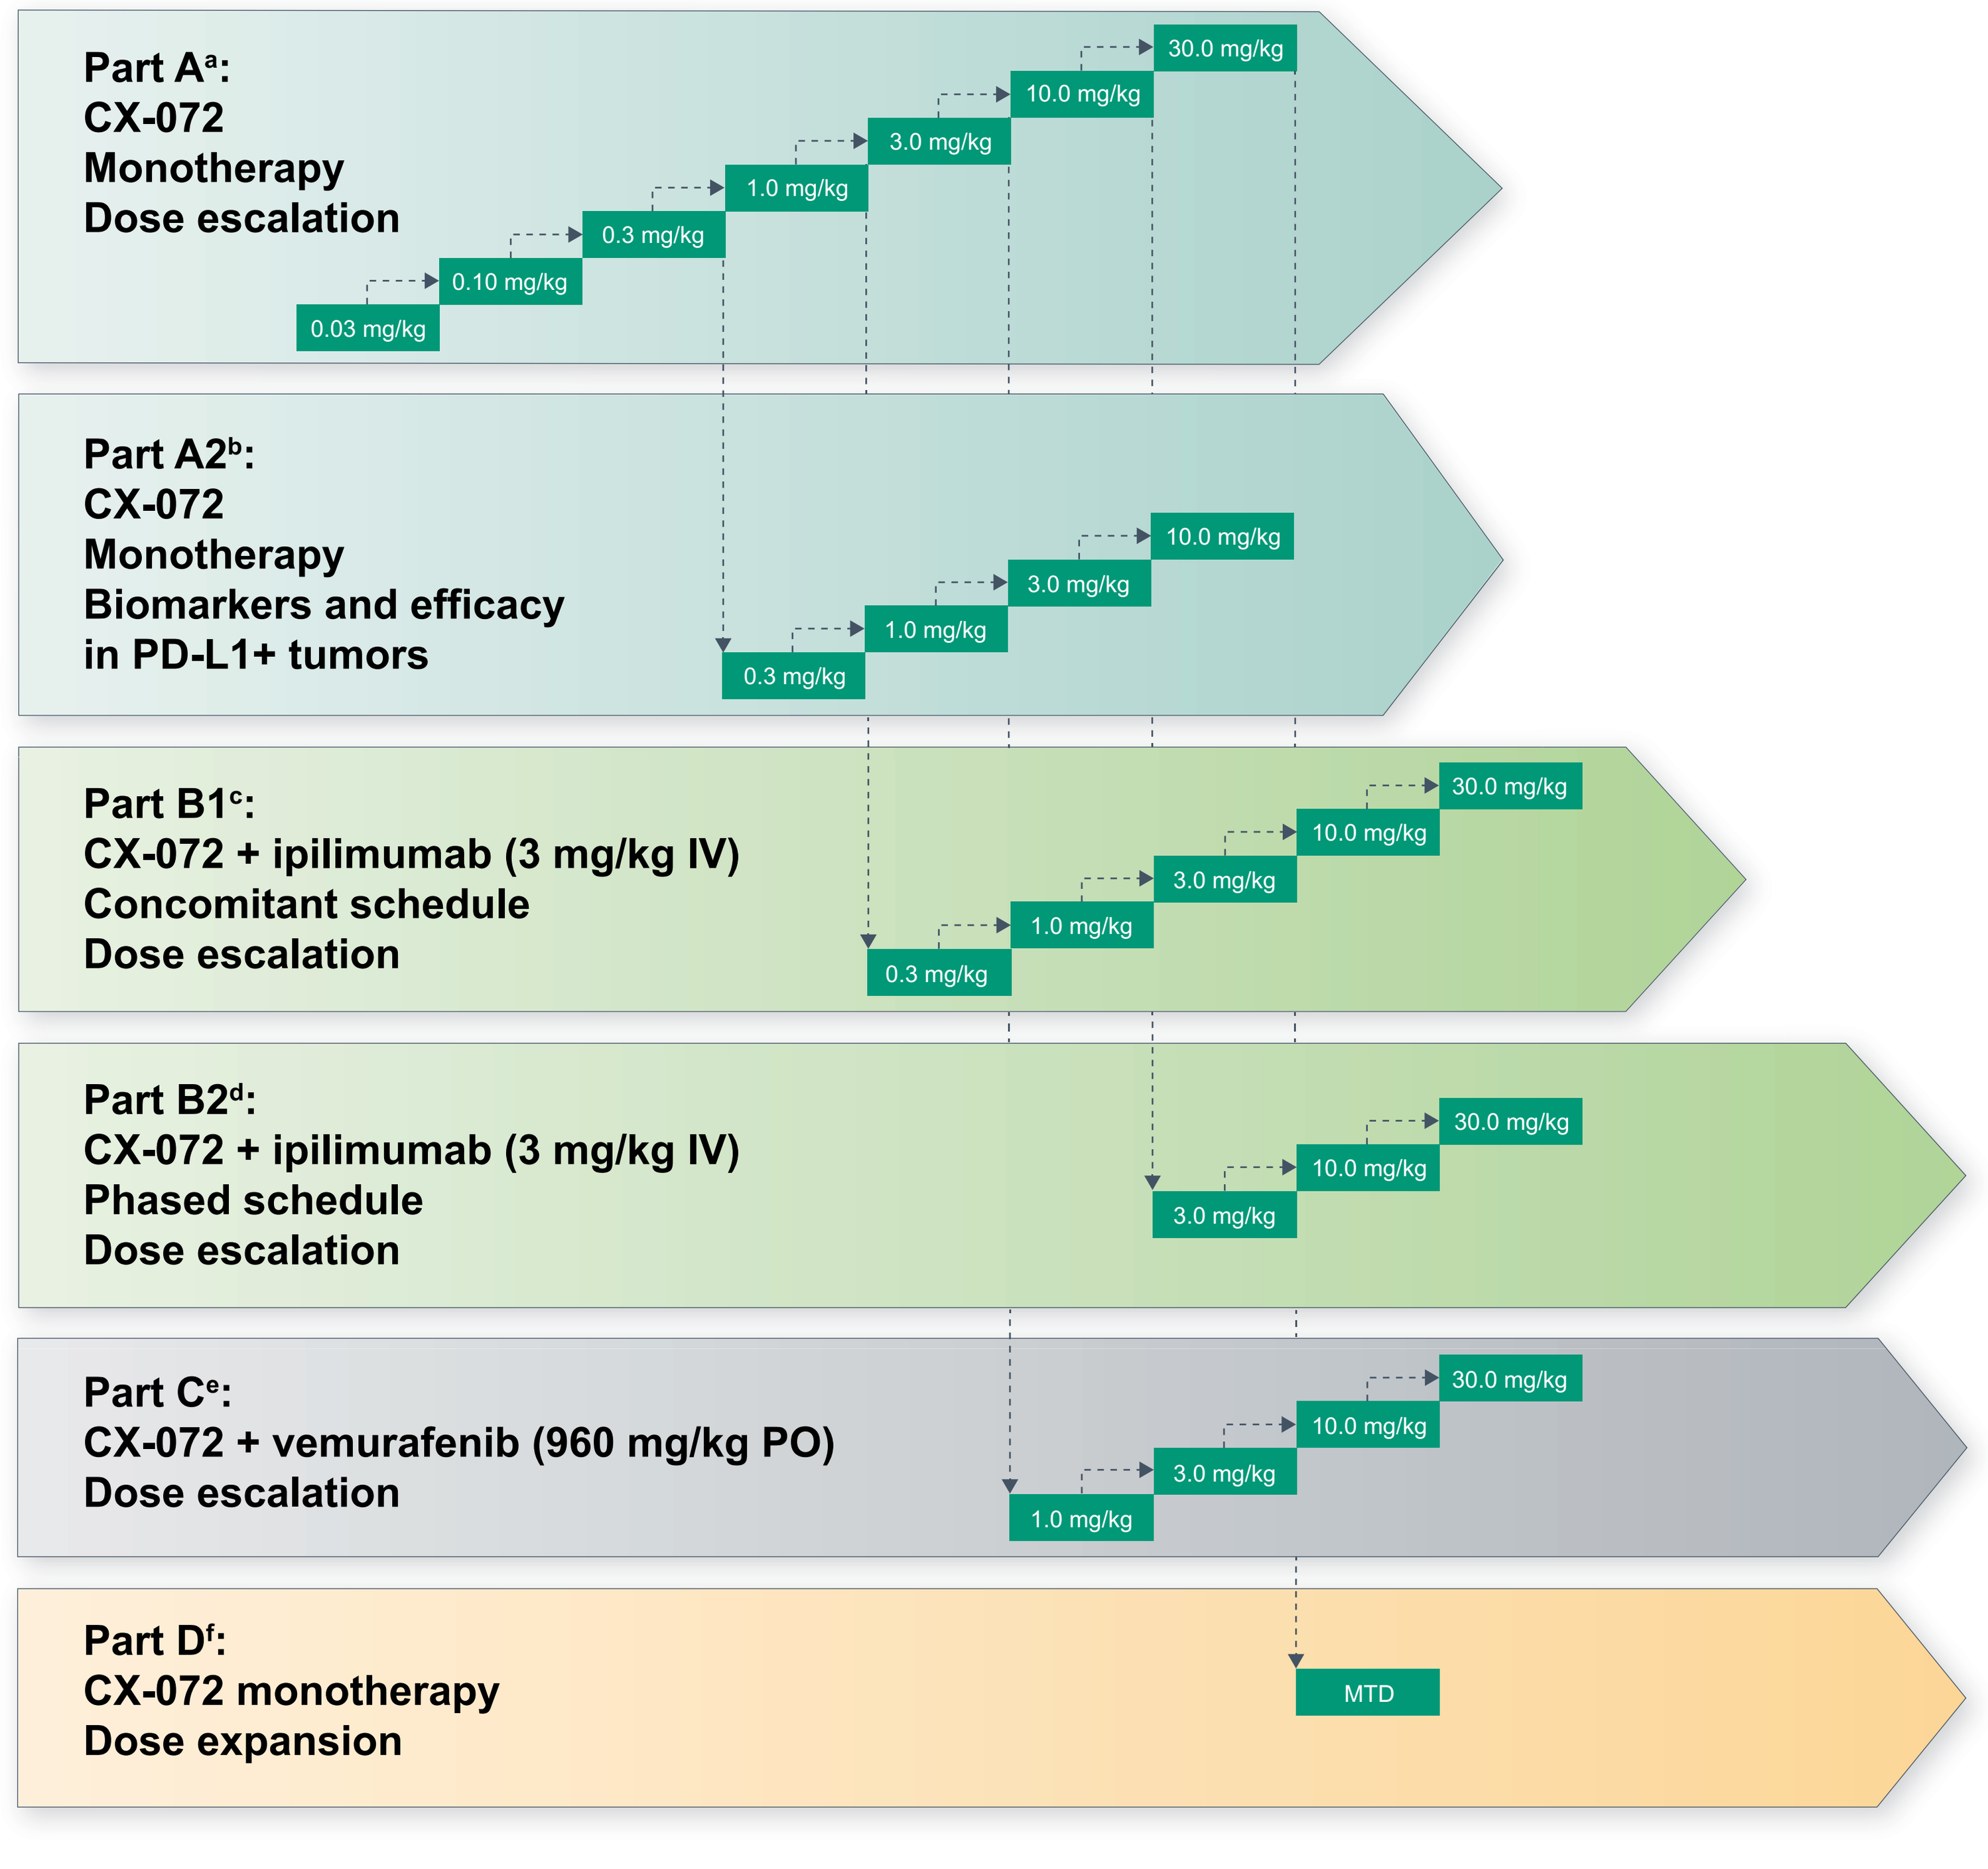

Part A[a]: CX-072 Monotherapy Dose escalation
0.03 mg/kg
0.10 mg/kg
0.3 mg/kg
1.0 mg/kg
3.0 mg/kg
10.0 mg/kg
30.0 mg/kg
Part A2[b]: CX-072 Monotherapy Biomarkers and efficacy in PD-L1+ tumors
0.3 mg/kg
1.0 mg/kg
3.0 mg/kg
10.0 mg/kg
Part B1[c]: CX-072 + ipilimumab (3 mg/kg IV) Concomitant schedule Dose escalation
0.3 mg/kg
1.0 mg/kg
3.0 mg/kg
10.0 mg/kg
30.0 mg/kg
Part B2[d]: CX-072 + ipilimumab (3 mg/kg IV) Phased schedule Dose escalation
3.0 mg/kg
10.0 mg/kg
30.0 mg/kg
Part C[e]: CX-072 + vemurafenib (960 mg/kg PO) Dose escalation
1.0 mg/kg
3.0 mg/kg
10.0 mg/kg
30.0 mg/kg
Part D[f]: CX-072 monotherapy Dose expansion
MTD

IV, intravenously; MAD, maximum achieved dose; MTD, maximum tolerated dose; PD-L1, programmed cell death ligand 1; PO, by mouth.
[a]CX-072 monotherapy will be administered IV every 14 days. One patient each was enrolled in the 0.03-, 0.1-, and 0.3-mg/kg dosing cohorts, and subsequent dose levels follow a 3+3 design, which is also used for all other dose-escalation groups.
[b]After successful completion of the monotherapy dose level in Part A, Part A2 will enroll an additional 6 patients with PD-L1+ cancer at each indicated dose to refine the MTD/MAD and to evaluate the relationship between dose/exposure and safety, efficacy, and pharmacodynamic biomarkers.
[c]Part B1 begins after successful completion of the subsequent monotherapy dose level tested in Part A. CX-072 plus ipilimumab will be administered IV every 21 days × 4 doses, followed by CX-072 monotherapy administered IV every 14 days.
[d]Part B2 begins after successful completion of the subsequent monotherapy dose level tested in Part A. During dose escalation, CX-072 monotherapy will be administered IV every 14 days × 4 doses. If tolerated, CX-072 plus ipilimumab will be administered IV every 21 days × 4 doses, followed by CX-072 monotherapy administered IV every 14 days.
[e]Part C begins after successful completion of the subsequent monotherapy dose level tested in Part A. CX-072 will be administered IV every 14 days plus vemurafenib twice daily.
[f]Part D will begin after dose escalation for Part A is complete and the MTD has been determined. CX-072 monotherapy will be administered IV at the MTD (determined from Part A) every 14 days.

Patients

- Up to 175 patients who meet defined criteria (**Table 1**) will be enrolled in the study

Table 1. Key Eligibility Criteria

All parts	• Age ≥18 years • ECOG performance status 0-1
Part A	• Advanced, unresectable solid tumor or lymphoma with no further standard of care available • Immunotherapy naive (including PD-1/PD-L1 and CTLA-4 inhibitor therapy) • Immunotherapy unavailable for patient's disease
Part A2	• Same requirements as for Part A and must be PD-L1+ (tumor proportion score ≥1% membranous staining) • Must agree to participate in biomarker analysis and have a tumor site that is safe to biopsy
Part B1	• Advanced, unresectable solid tumor or lymphoma with no further standard of care available • Immunotherapy naive • Immunotherapy unavailable for patient's disease
Part B2	• Advanced, unresectable solid tumor or lymphoma with no further standard of care available • Previously treated with a PD-1/PD-L1 inhibitor (discontinued for reasons other than toxicity) • CTLA-4 inhibitor naive
Part C	• Advanced, unresectable melanoma • *BRAF*^{V600E} mutation positive • BRAF inhibitor naive • Immunotherapy naive • Immunotherapy unavailable for patient's disease
Part D	• Advanced, unresectable PD-L1–responsive tumor types • Measurable disease • PD-L1+ or status unknown (not known to be PD-L1–) • Immunotherapy naive • Immunotherapy unavailable for patient's disease

CTLA-4, cytotoxic T-lymphocyte–associated antigen 4; ECOG, Eastern Cooperative Oncology Group; PD-1, programmed cell death 1; PD-L1, programmed cell death ligand 1.

END POINTS

Primary End Points

- Safety and tolerability of CX-072 alone or in combination with ipilimumab or vemurafenib
- MTD and dose-limiting toxicities of CX-072 alone or in combination with ipilimumab or vemurafenib

Secondary End Points

- Objective response according to Response Evaluation Criteria in Solid Tumors version 1.1 (RECIST v1.1), immune-related RECIST v1.1, or modified Cheson/Lugano classification for lymphomas
- Time to response
- Duration of response
- Progression-free survival
- Incidence of antidrug antibodies
- Single- and multiple-dose pharmacokinetic profile of CX-072 alone and of CX-072 in combination with ipilimumab or vemurafenib
- Overall survival

Exploratory Objectives

- Protease activity and degree of CX-072 cleavage in tumor and peripheral blood
- Immunomodulatory activity of CX-072 in on-treatment biopsy samples
- Potential predictive markers of CX-072 activity

SPECIFIC ASSESSMENTS

- Imaging for tumor response assessment will be performed every 8 weeks for the first 12 months, then every 12 weeks thereafter
- After the last dose of study medication, patients will be evaluated every 3 months for disease progression and overall survival until study withdrawal or death
- Archival tissue or fresh biopsy samples must be provided at baseline. Patients in Part A2 and Part B2 must be willing to undergo ≥1 on-treatment tumor biopsy, and all patients can consent to biopsy to aid in translational analyses

Translational Analyses

- Several translational strategies will be used to investigate Probody therapeutic activation, PD-L1 inhibition, and immune response pattern in the tumor (**Table 2, Figure 4**)

Table 2. Translational Analyses Included in PROCLAIM-CX-072

Goal	Sample(s)	Assay	Description
Probody therapeutic activation	Biopsy, plasma	WES™ assay	Capillary electrophoresis with immunodetection to identify masked and activated CX-072
	Biopsy	QZ™ assay	Protease activity detection
PD biomarkers	Biopsy	Nanostring™	Gene expression panel
	Biopsy	IHC	Immune cell infiltration
	Plasma	Luminex®	Cytokine panel
PD-L1 expression	Biopsy	IHC	PD-L1 expression

IHC, immunohistochemistry; PD, pharmacodynamics; PD-L1, programmed cell death ligand 1.

Figure 4. Preclinical measurement of cleaved (green) and intact (blue) Probody therapeutic by WES assay in tumor and plasma.

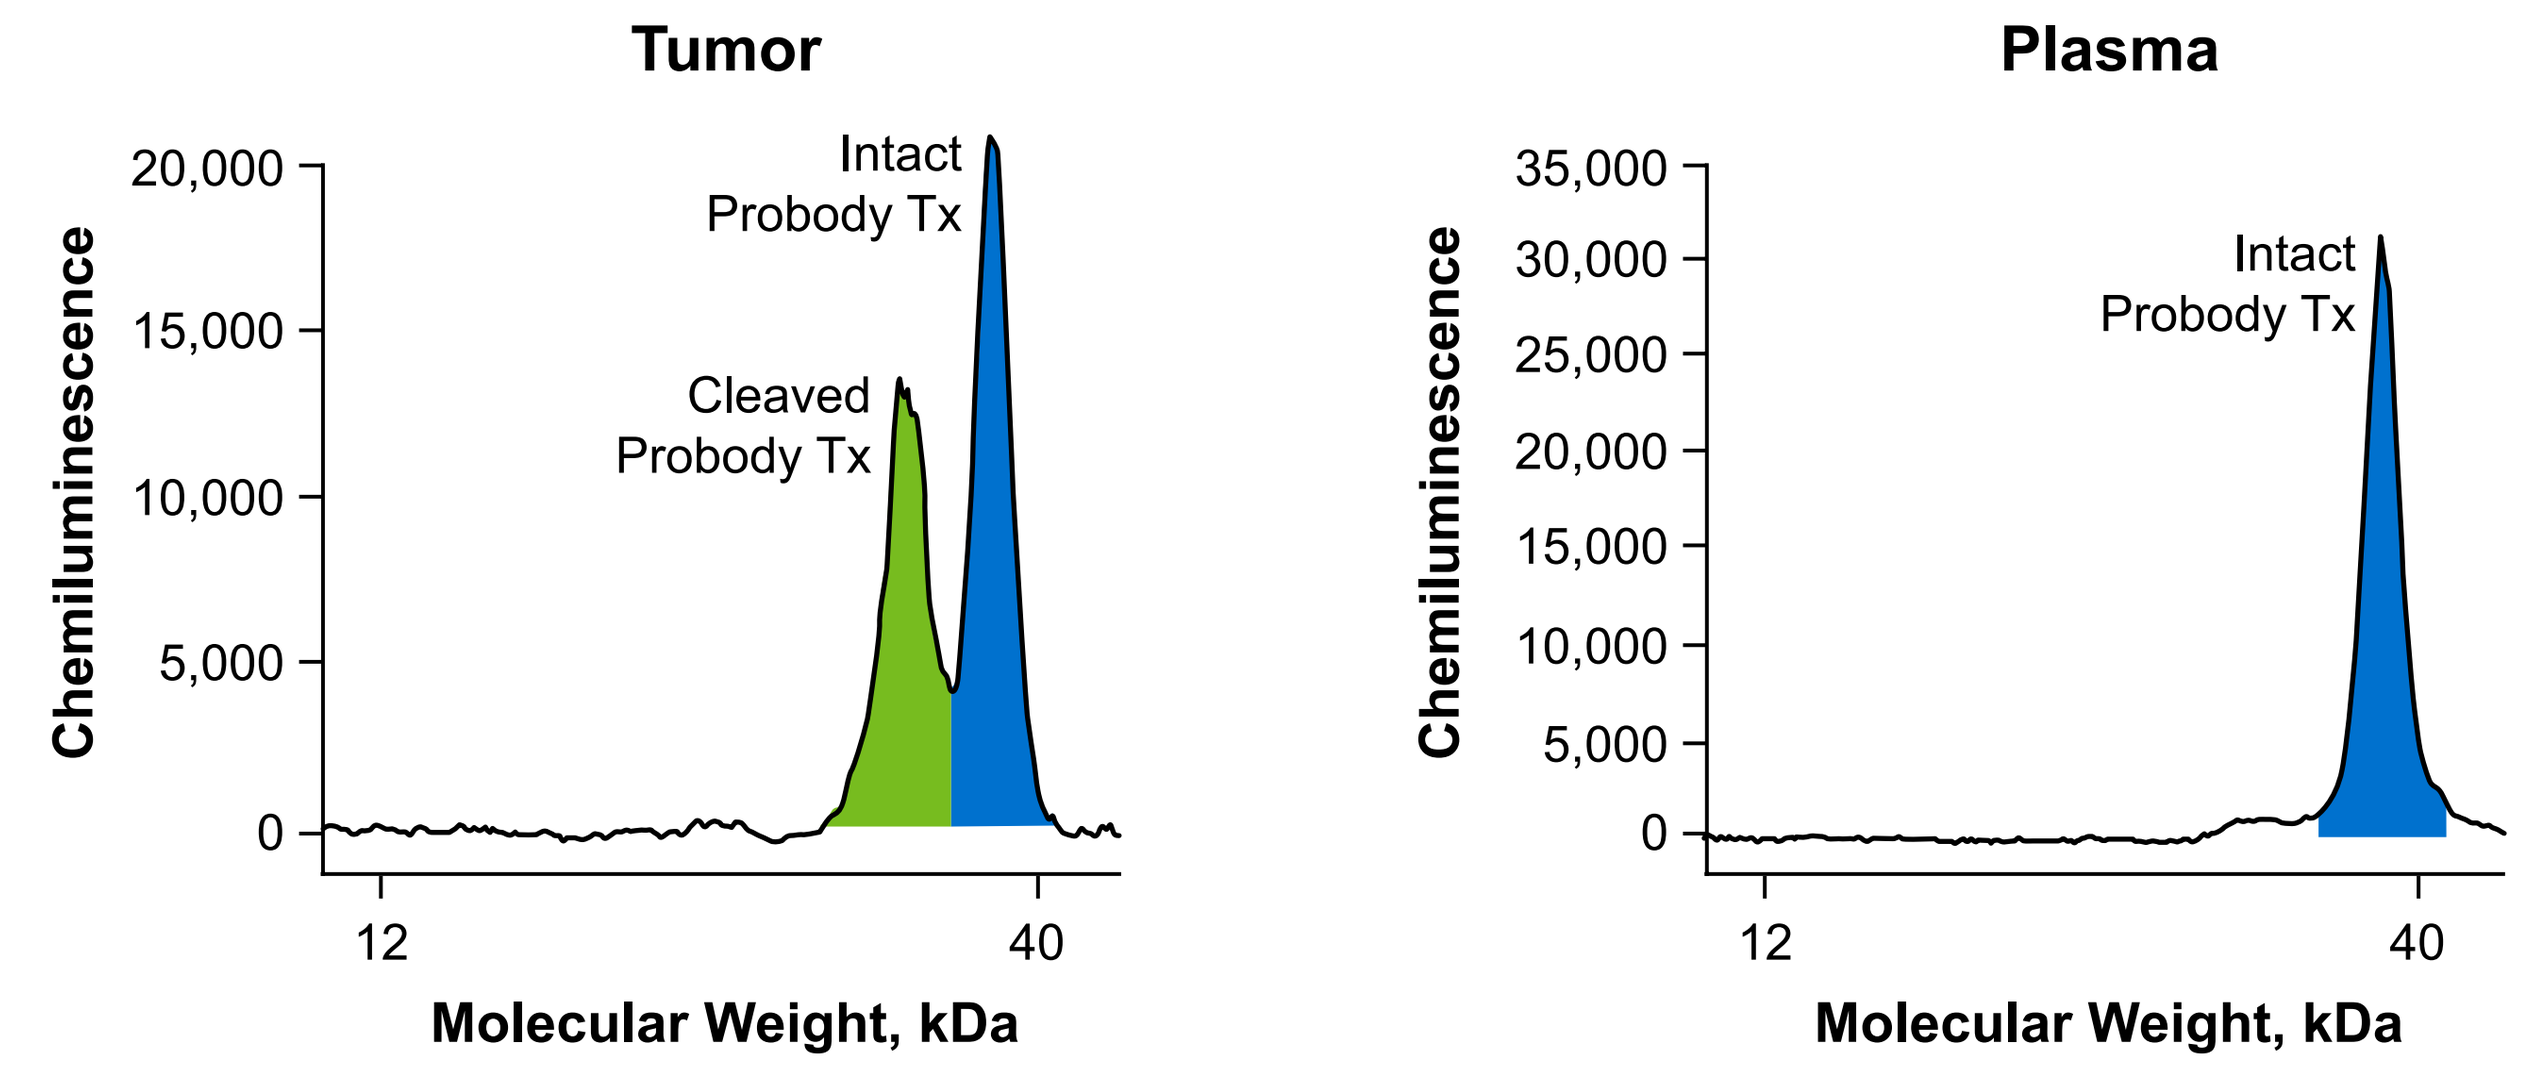

Tumor
Chemiluminescence
20,000
15,000
10,000
5,000
0
Intact Probody Tx
Cleaved Probody Tx
12
40
Molecular Weight, kDa
Plasma
Chemiluminescence
35,000
30,000
25,000
20,000
15,000
10,000
5,000
0
Intact Probody Tx
12
40
Molecular Weight, kDa

kDa, kilodalton; Tx, therapeutic.

STUDY PROGRESS

- The study started in January 2017. Sites are open for Part A in the United States and in Europe, and enrollment through the 3-mg/kg dose is complete. Part A is expected to be completed by the end of 2017. Parts B1 and C are also open for enrollment
- This study is registered with ClinicalTrials.gov, number NCT03013491
- For more information, please visit https://clinicaltrials.gov/ct2/show/NCT03013491 or contact clinicaltrials@cytomx.com

REFERENCES

1. Herbst RS et al. *Nature.* 2014;515:563-567.
2. Iwai Y et al. *J Biomed Sci.* 2017;24:26.
3. Wolchok JD et al. *N Engl J Med.* 2013;369:122-133.
4. Larkin J et al. *N Engl J Med.* 2015;373:23-34.
5. Ahn MJ et al. *Expert Opin Drug Saf.* 2017;16:465-469.
6. Hwu P et al. *Ann Oncol.* 2016;27:379-400.
7. Desnoyers LR et al. *Sci Transl Med.* 2013;5:207ra144.
8. Polu KR et al. *Expert Opin Biol Ther.* 2014;14:1049-1053.
9. Wong C et al. Presented at: CRI-CIMT-EATI-AACR; September 16-19, 2015; New York, NY.

ACKNOWLEDGMENTS

The authors thank the participating patients and their families and all staff at the participating sites. This study is sponsored by CytomX Therapeutics, Inc. Medical writing assistance was provided by ApotheCom (San Diego, CA, USA) and was funded by CytomX Therapeutics, Inc.

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors.

PROBODY and QZ are trademarks of CytomX Therapeutics, Inc. All other brands and trademarks referenced herein are the property of their respective owners.

http://bit.ly/2vy64Zn

View Other Publications

Presented at the ESMO (European Society of Medical Oncology) 2017 Congress; September 8–12, 2017; Madrid, Spain

© 2017 CytomX Therapeutics, Inc.